ASM Microbe 2022 | P2363

Ceftibuten-avibactam (fixed 4 µg/mL), Ceftibuten, and Ceftazidime-avibactam (fixed 4 µg/mL) Broth Microdilution Quality Control Ranges Using a CLSI M23 (2018) Multi-Laboratory Study Design

MD Huband[1], HS Sader[1], Y Edah[1], K Fedler[1], GG Stone[2], M Castanheira[1]

[1] JMI Laboratories, North Liberty, IA, USA; [2] Pfizer Inc., New York, NY, USA

Introduction

- Ceftibuten-avibactam (fixed 4 µg/mL) is an orally active antibacterial combination being developed for the treatment of Gram-negative bacterial infections, particularly those responsible for urinary tract infection.
- A Clinical and Laboratory Standards Institute (CLSI) M23 (Tier 2) quality control (QC) study was conducted to establish ceftibuten-avibactam (fixed 4 µg/mL) broth microdilution QC ranges against 5 CLSI QC strains and to provide additional QC ranges for ceftibuten and ceftazidime-avibactam (fixed 4 µg/mL).
- Establishing ceftibuten-avibactam (fixed 4 µg/mL) broth microdilution QC ranges will assist both clinical and reference laboratories in generating reliable, accurate susceptibility testing results during clinical trials and in clinical microbiology practice.

Materials and Methods

- Eight institutions participated in the CLSI M23 (2018) Tier 2 broth microdilution QC study for ceftibuten-avibactam (fixed 4 µg/mL), ceftibuten, and ceftazidime-avibactam (fixed 4 µg/mL).
- Reference broth microdilution susceptibility testing panels were prepared in a certified GMP facility (Trek Diagnostic Systems/Thermo Fisher Scientific, Oakwood Village, Ohio) using 3 lots of Mueller-Hinton broth obtained from at least 2 different manufacturers (*n*=3).
- Mueller-Hinton broth was obtained from Difco (Becton Dickinson, Lot 9156821), BBL (Becton Dickinson, Lot 0252344), and Oxoid (Thermo Fisher Scientific, Lot 3163254).
- Testing was performed over multiple days (≥3) and ≥10 replicate tests/QC strain.

Results

- Three dilution QC ranges were approved for ceftibuten-avibactam (fixed 4 µg/mL) against *Escherichia coli* NCTC 13353 (0.03/4-0.12/4; 100.0%), *Klebsiella pneumoniae* ATCC 700603 (0.06/4-0.25/4; 98.8%), and *K. pneumoniae* ATCC BAA-2814 (0.12/4-0.5/4; 99.6%) (Table 1 and Figures 2-3 and 5).
- Four dilution QC ranges were approved for *E. coli* ATCC 25922 (0.016/4-0.12/4; 100.0%) and *K. pneumoniae* ATCC BAA-1705 (0.03/4-0.25/4; 100.0%) based on the bimodal distribution of MIC values (Table 1 and Figures 1 and 4).
- All (100.0%) of the ceftibuten MIC values against *E. coli* ATCC 25922, *E. coli* NCTC 13353, *K. pneumoniae* ATCC BAA-1705, and *K. pneumoniae* ATCC BAA-2814 were within current CLSI-approved QC ranges listed in the M100 (2022) document providing validated internal controls on each day of susceptibility testing (Table 1).
- All (100.0%) of the ceftazidime-avibactam (fixed 4 µg/mL) MIC values against *E. coli* ATCC 25922 and *K. pneumoniae* ATCC 700603 were within current CLSI-approved QC ranges listed in the M100 (2022) document providing validated internal controls on each day of susceptibility testing (Table 1).
- Comparator agent QC ranges were also approved for ceftibuten against *K. pneumoniae* ATCC 700603 (0.25-1 µg/mL; 99.2%) and ceftazidime-avibactam (fixed 4 µg/mL) against *E. coli* NCTC 13353 (0.12/4-0.5/4; 100.0%), *K. pneumoniae* ATCC BAA-1705 (0.25/4-2/4; 100.0%), and *K. pneumoniae* ATCC BAA-2814 (1/4-4/4; 100.0%) (Table 1).

Table 1. CLSI approved broth microdilution quality control ranges

QC Strain	Approved broth microdilution QC range (µg/mL) (% in range; no. of dilutions): Ceftibuten-avibactam (fixed 4 µg/mL)	Ceftibuten	Ceftazidime-avibactam (fixed 4 µg/mL)
E. coli ATCC 25922	**0.016/4 – 0.12/4 (100.0%; 4)**	0.12 – 0.5[a] (100.0%; 3)	0.06/4 – 0.5/4[a] (100.0%; 4)
E. coli NCTC 13353	**0.03/4 – 0.12/4 (100.0%; 3)**	16 – 64[a] (100.0%; 3)	**0.12/4 – 0.5/4 (100.0%; 3)**
K. pneumoniae ATCC 700603	**0.06/4 – 0.25/4 (98.8%; 3)**	**0.25 – 1 (99.2%; 3)**	0.25/4 – 2/4[a] (100.0%; 4)
K. pneumoniae ATCC BAA-1705	**0.03/4 – 0.25/4 (100.0%; 4)**	4 – 32[a] (100.0%; 4)	**0.25/4 – 2/4 (100.0%; 4)**
K. pneumoniae ATCC BAA-2814	**0.12/4 – 0.5/4 (99.6%; 3)**	8 – 32[a] (100.0%; 3)	**1/4 – 4/4 (100.0%; 3)**

[a] Current CLSI approved QC range.

Figure 1. Ceftibuten-avibactam (fixed 4 µg/mL) MIC distributions by medium lot for *E. coli* ATCC 25922

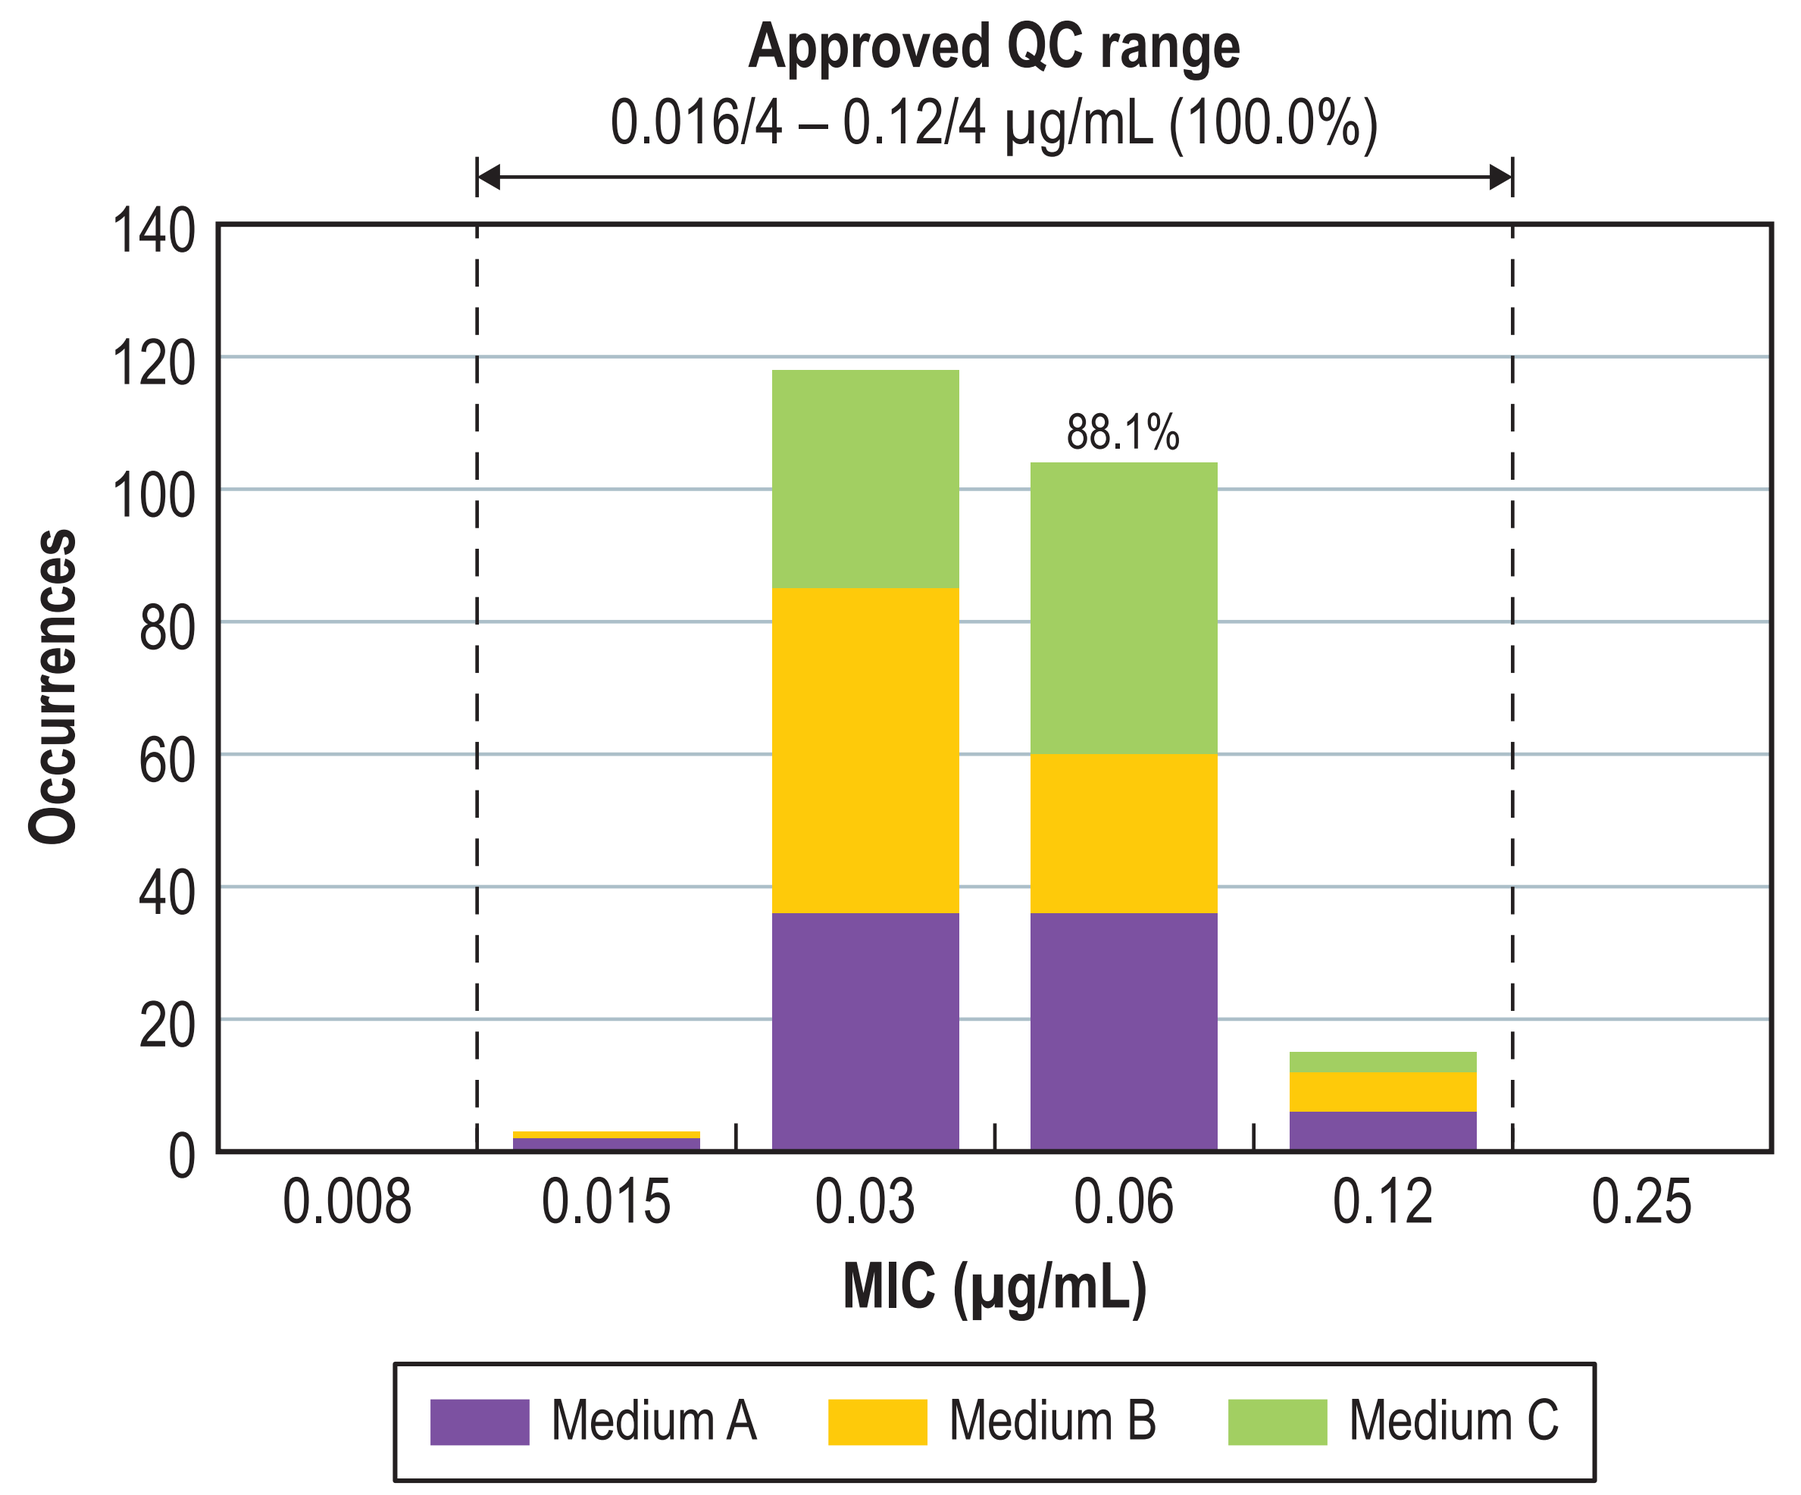
Approved QC range
0.016/4 – 0.12/4 µg/mL (100.0%)
88.1%
Occurrences
MIC (µg/mL)
Medium A
Medium B
Medium C

Figure 2. Ceftibuten-avibactam (fixed 4 µg/mL) MIC distributions by medium lot for *E. coli* NCTC 13353

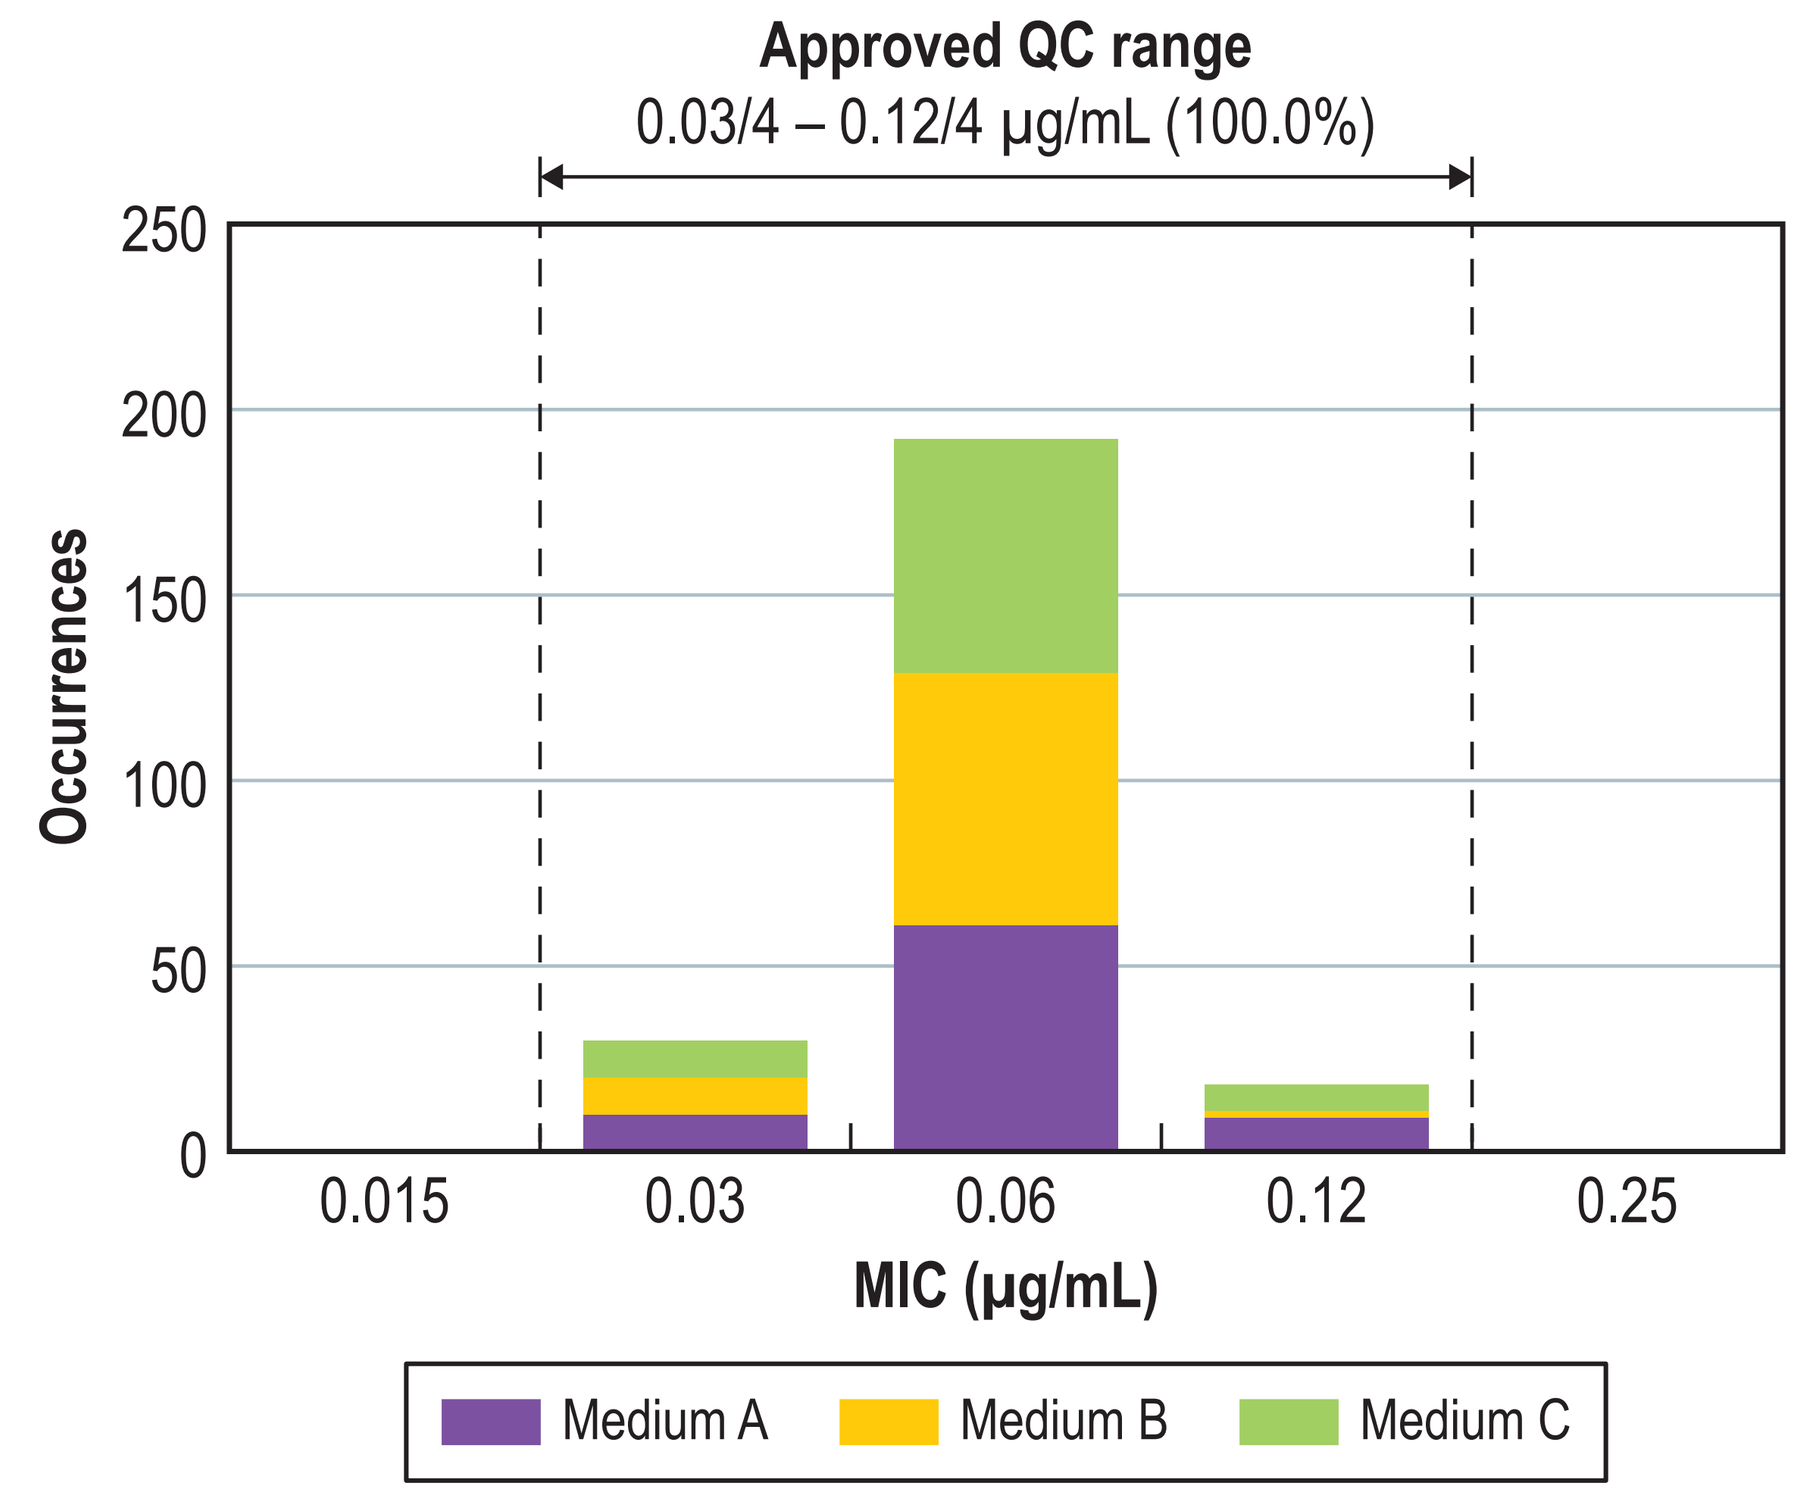
Approved QC range
0.03/4 – 0.12/4 µg/mL (100.0%)
Occurrences
MIC (µg/mL)
Medium A
Medium B
Medium C

Figure 3. Ceftibuten-avibactam (fixed 4 µg/mL) MIC distributions by medium lot for *K. pneumoniae* ATCC 700603

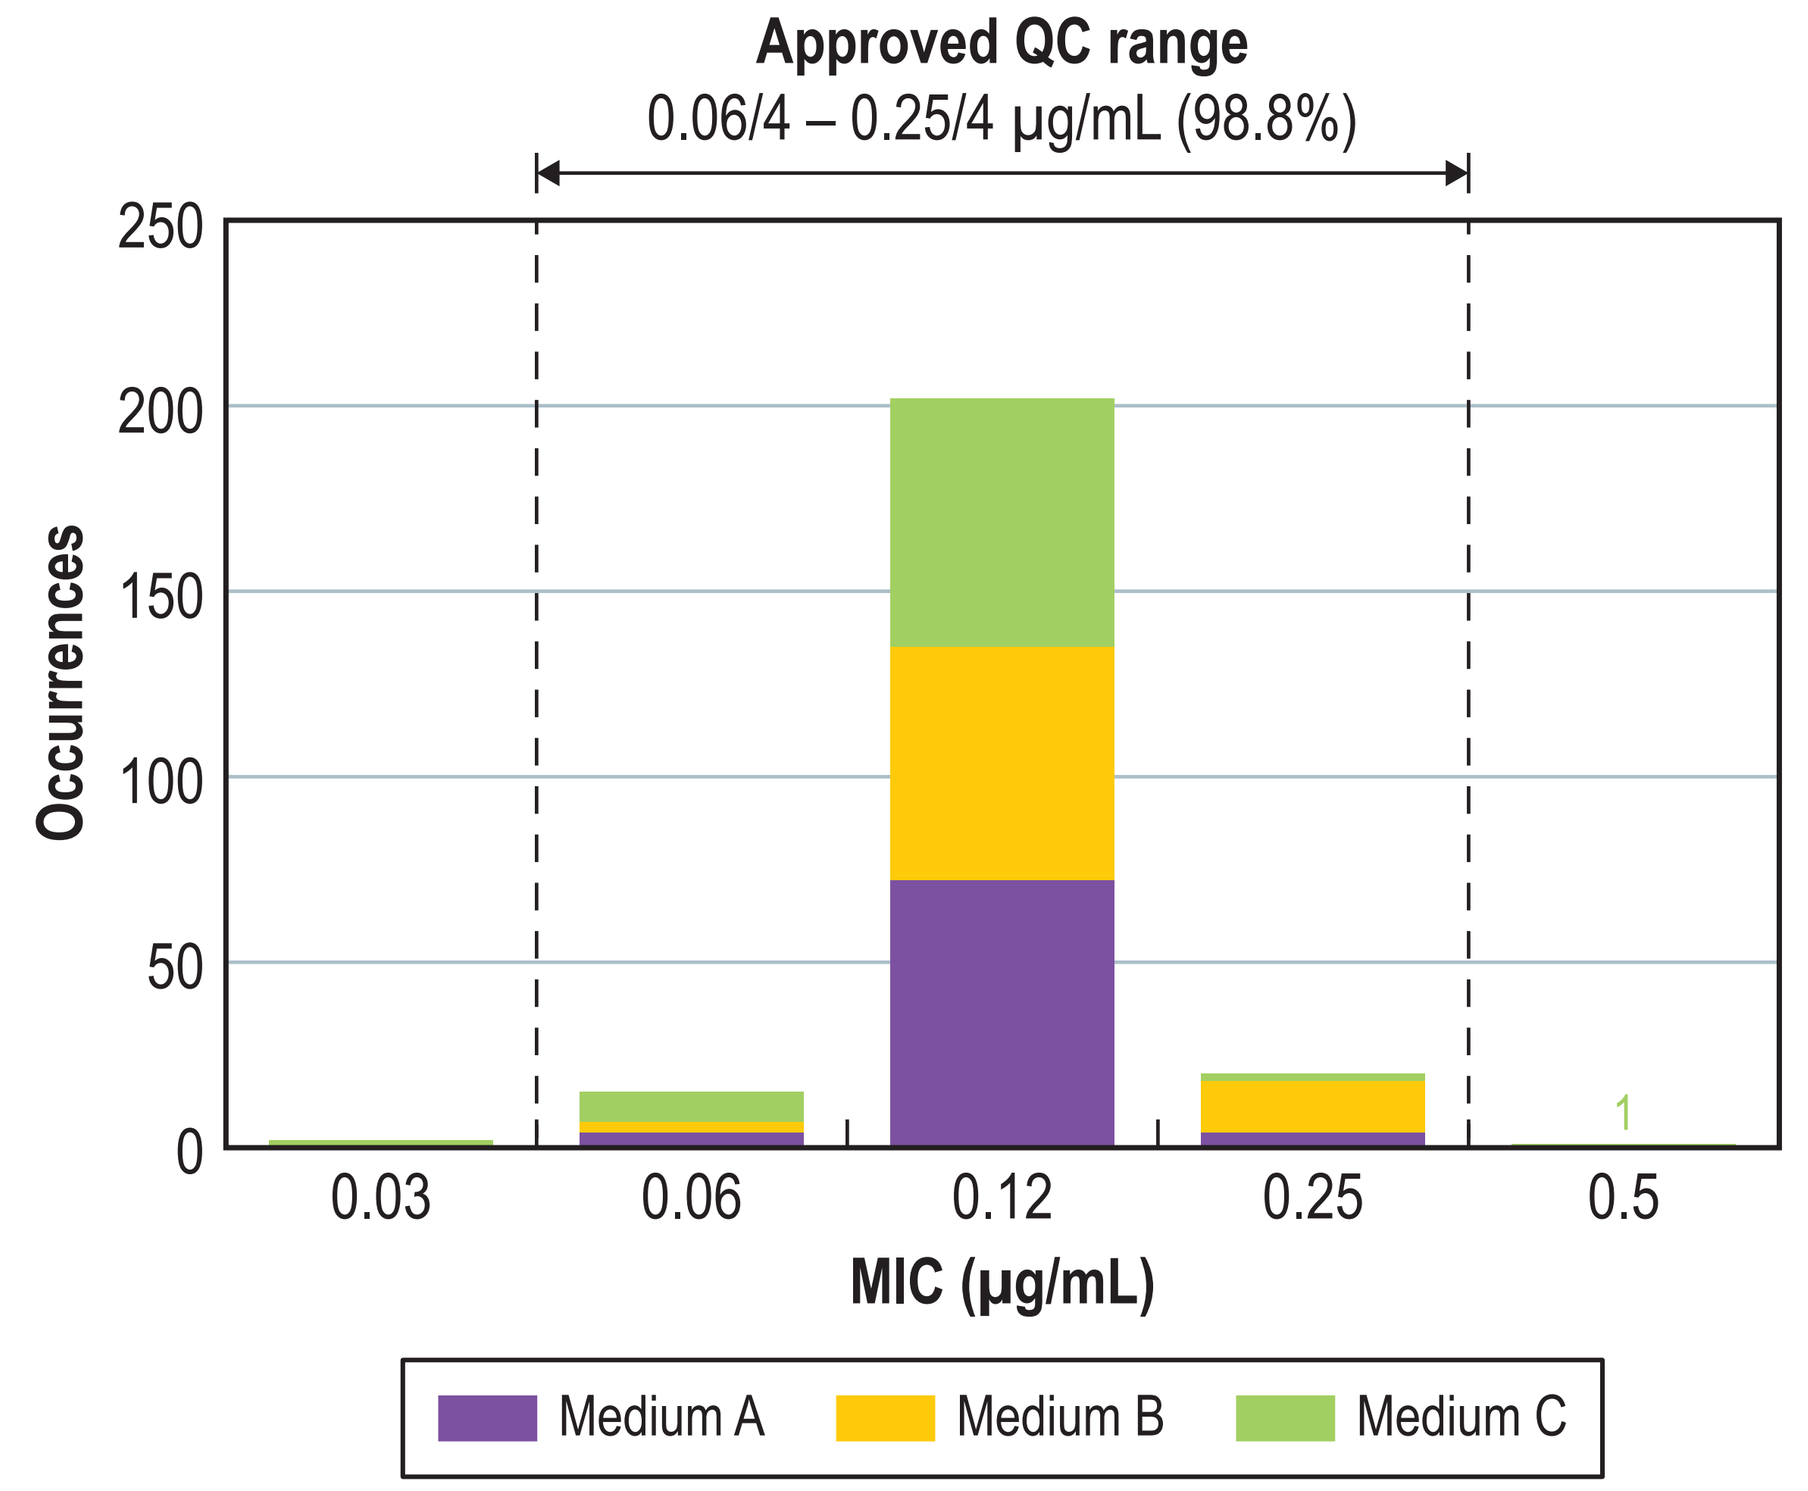
Approved QC range
0.06/4 – 0.25/4 µg/mL (98.8%)
Occurrences
MIC (µg/mL)
Medium A
Medium B
Medium C

Figure 4. Ceftibuten-avibactam (fixed 4 µg/mL) MIC distributions by medium lot for *K. pneumoniae* ATCC BAA-1705

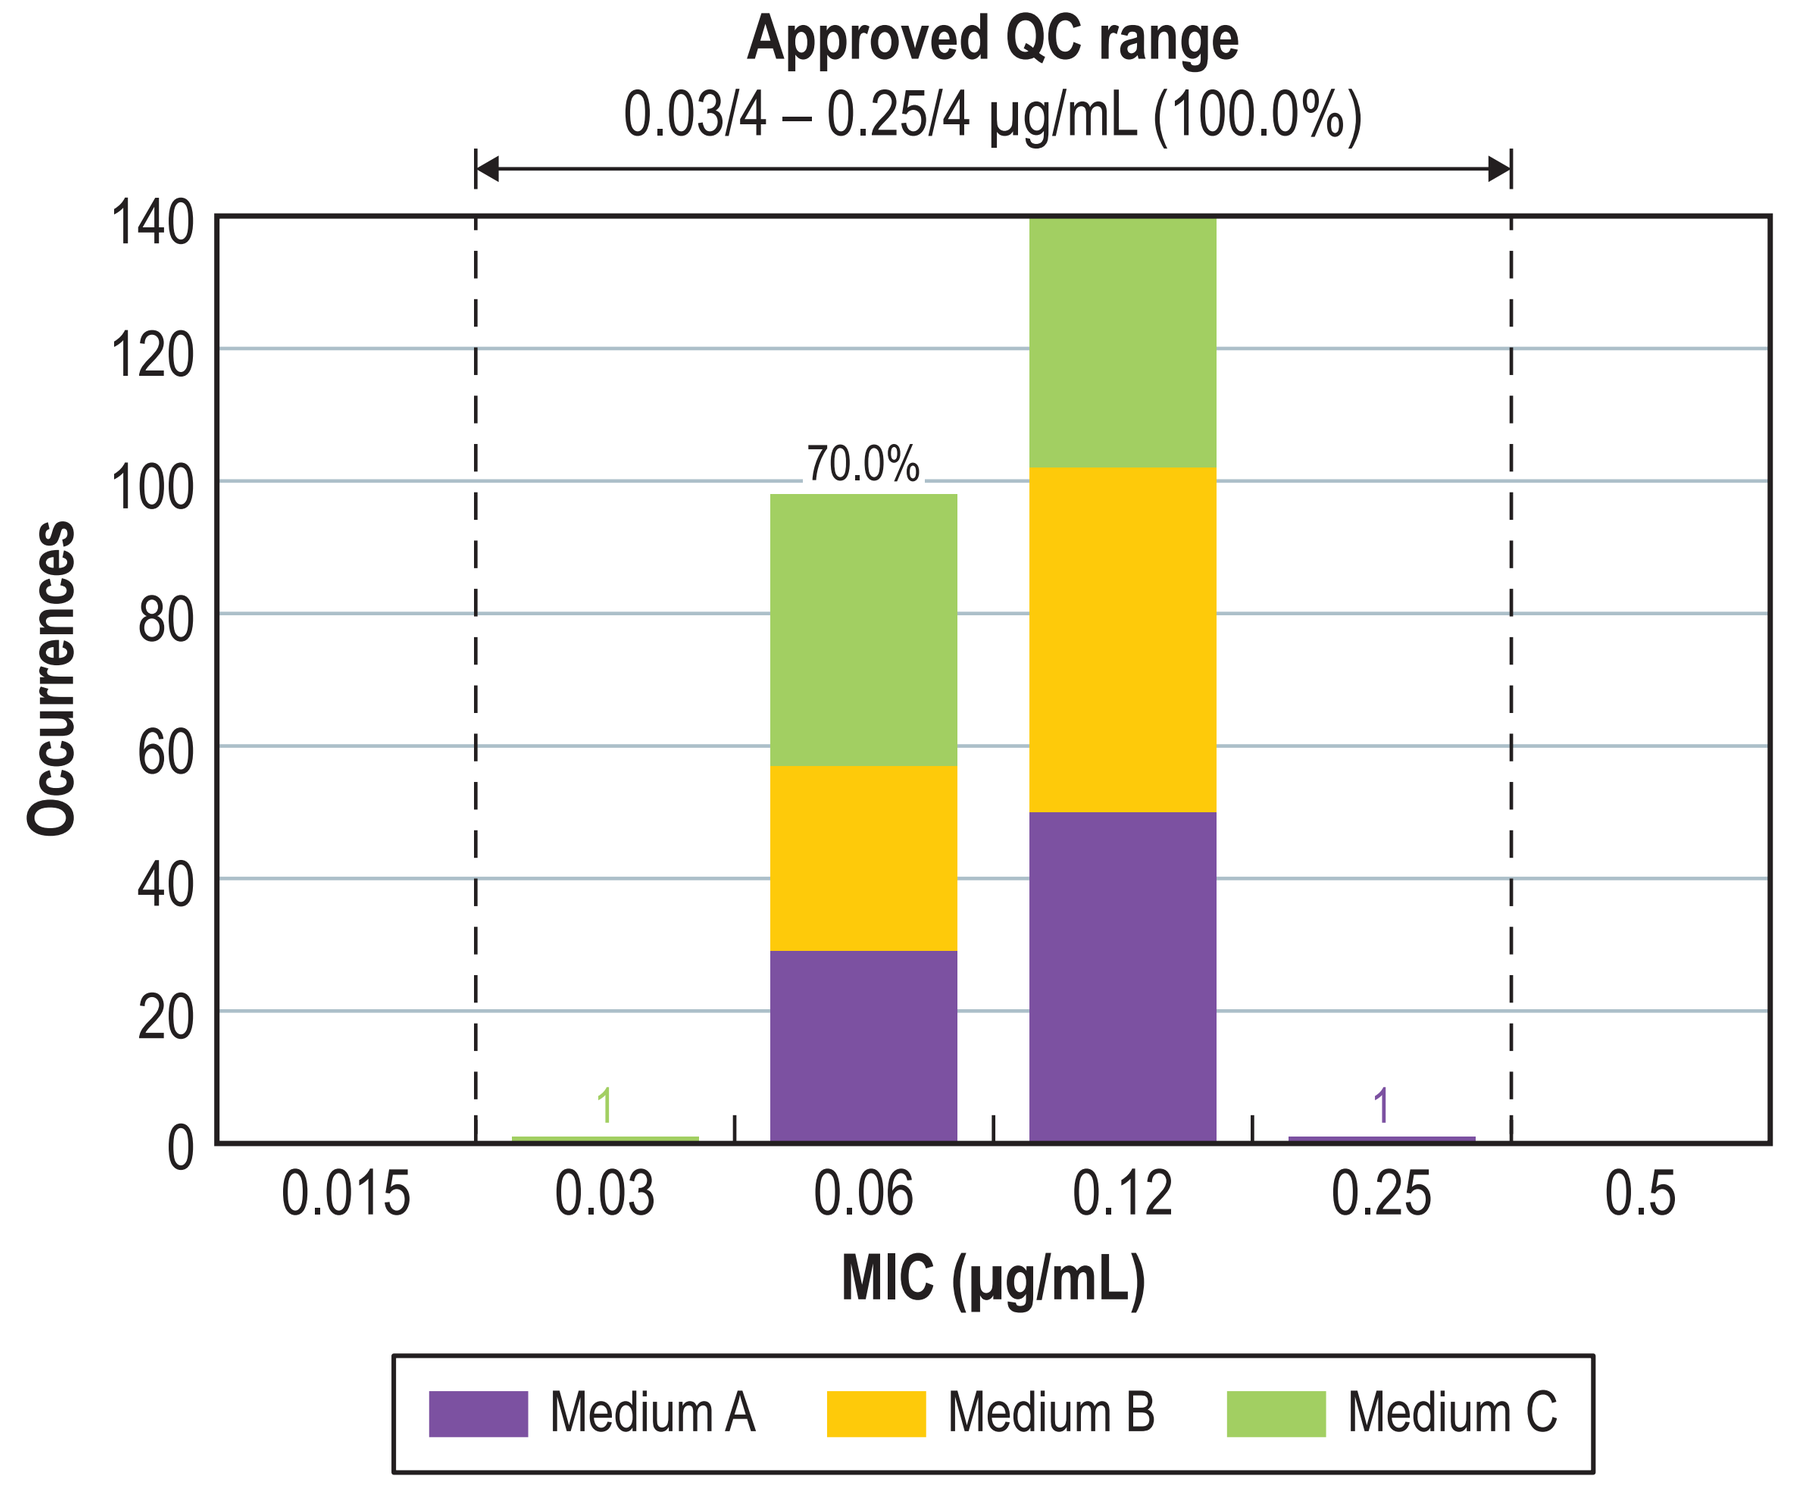
Approved QC range
0.03/4 – 0.25/4 µg/mL (100.0%)
70.0%
Occurrences
MIC (µg/mL)
Medium A
Medium B
Medium C

Figure 5. Ceftibuten-avibactam (fixed 4 µg/mL) MIC distributions by medium lot for *K. pneumoniae* ATCC BAA-2814

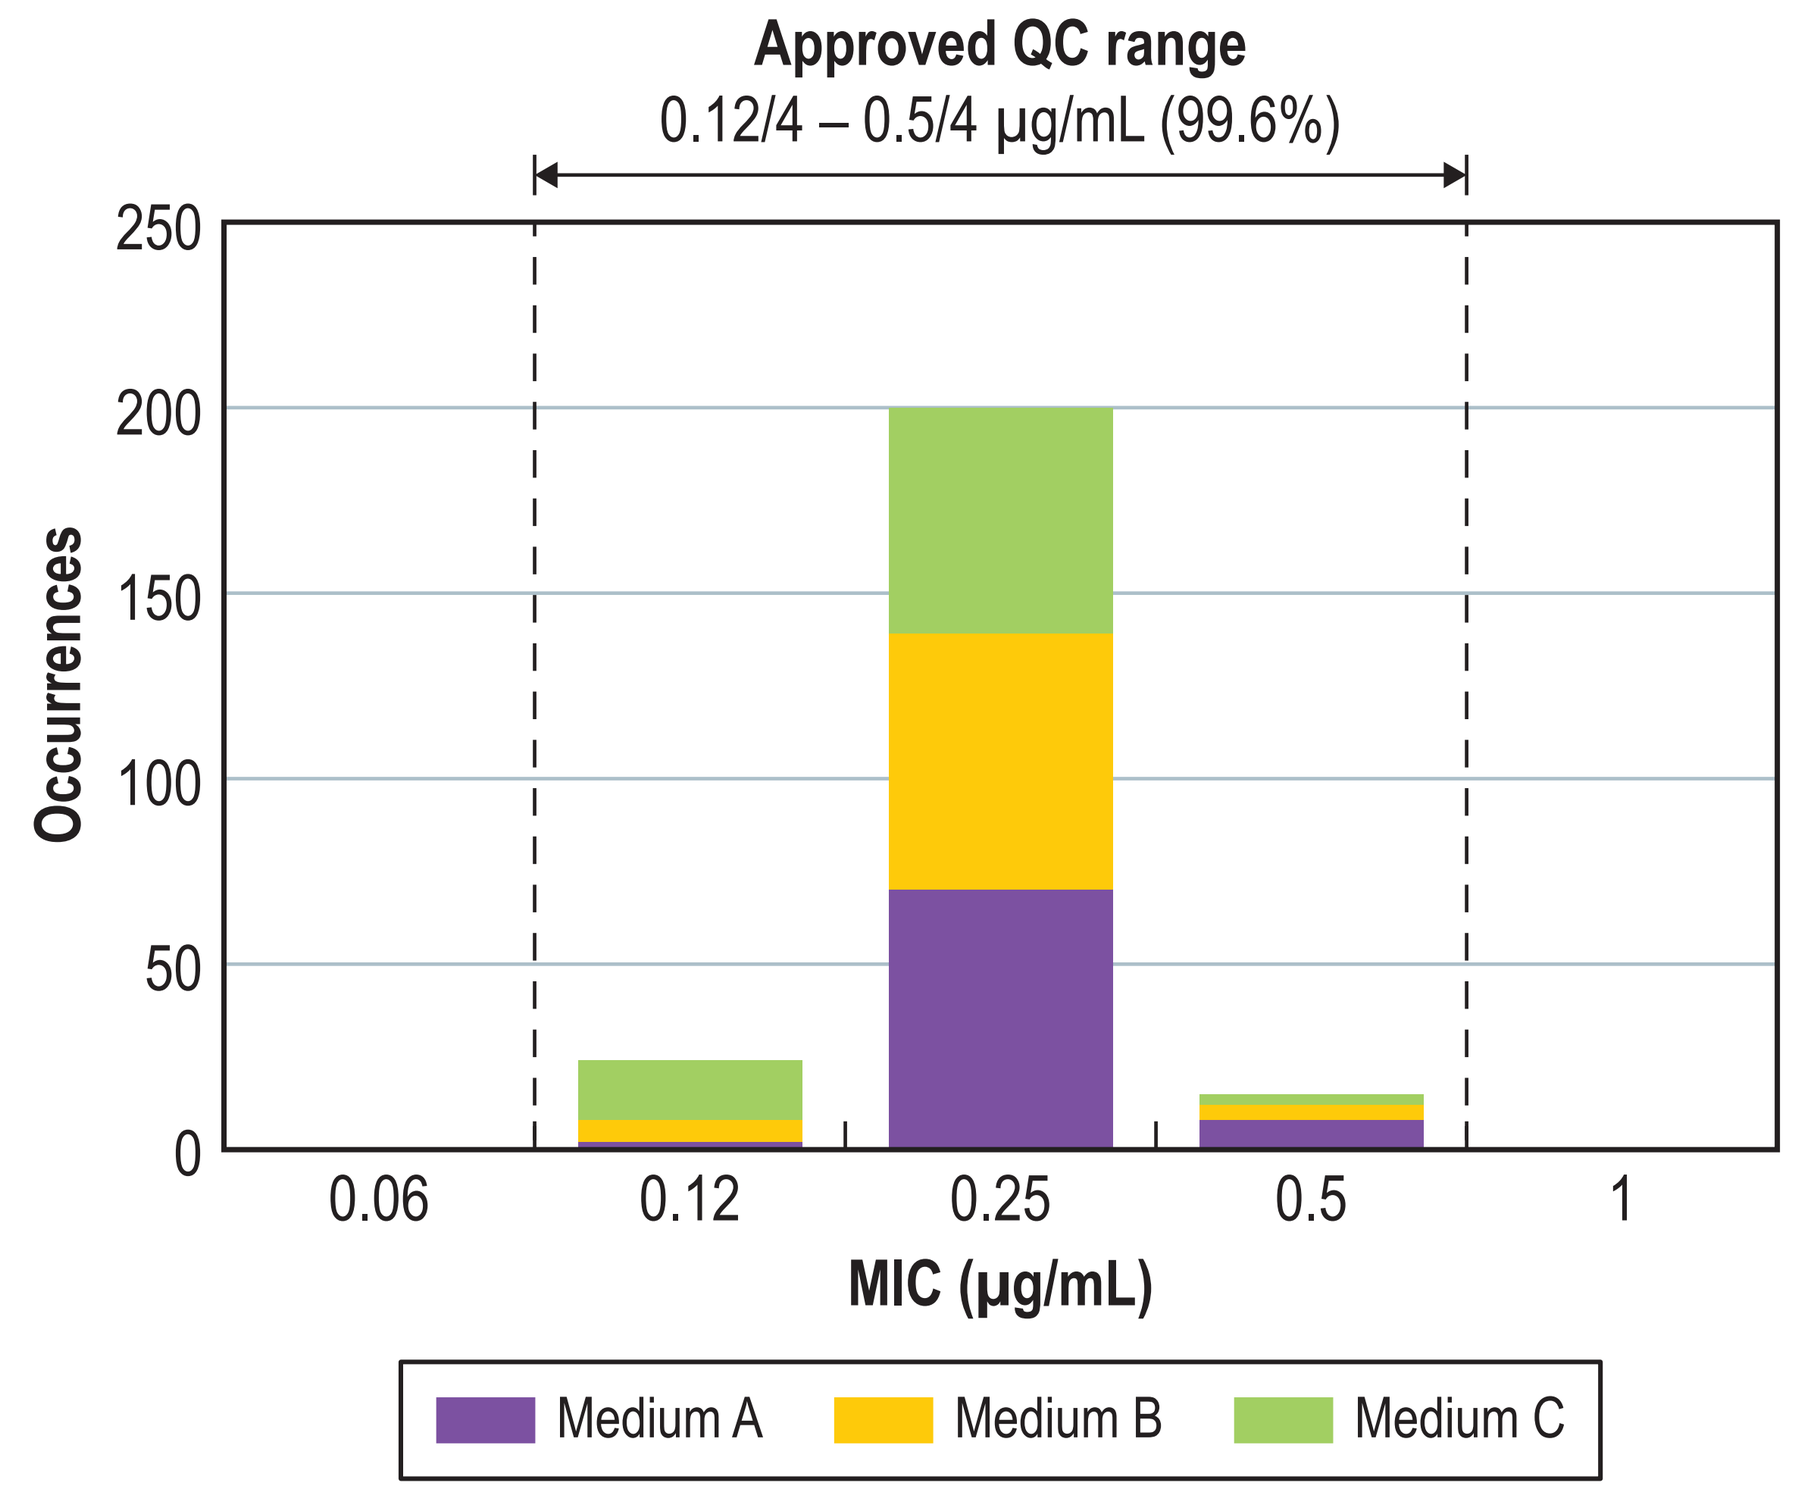
Approved QC range
0.12/4 – 0.5/4 µg/mL (99.6%)
Occurrences
MIC (µg/mL)
Medium A
Medium B
Medium C

Conclusions

- The CLSI committee on Antimicrobial Susceptibility Testing approved ceftibuten-avibactam (fixed 4 µg/mL) broth microdilution QC ranges for *E. coli* ATCC 25922, *E. coli* NCTC 13353, *K. pneumoniae* ATCC 700603, *K. pneumoniae* ATCC BAA-1705, and *K. pneumoniae* ATCC BAA-2814; ceftibuten QC ranges for *K. pneumoniae* ATCC 700603; and ceftazidime-avibactam (fixed 4 µg/mL) QC ranges for *E. coli* NCTC 13353, *K. pneumoniae* ATCC BAA-1705, and *K. pneumoniae* ATCC BAA-2814 at the January 2022 CLSI meeting.
- Approved ceftibuten-avibactam (fixed 4 µg/mL), ceftibuten, and ceftazidime-avibactam (fixed 4 µg/mL) broth microdilution QC ranges will be published in Table 5A-2 of the 2023 CLSI M100 document.
- *E. coli* NCTC 13353, *K. pneumoniae* ATCC BAA-1705, and *K. pneumoniae* ATCC BAA-2814 should be utilized as routine QC strains when evaluating the activity of ceftibuten-avibactam (fixed 4 µg/mL) as they provide QC for both the ceftibuten and avibactam components of the antimicrobial combination.
- The approved broth microdilution QC ranges for ceftibuten-avibactam (fixed 4 µg/mL), ceftibuten, and ceftazidime-avibactam (fixed 4 µg/mL) will assist both clinical laboratories and reference laboratories participating in clinical trials.

Acknowledgements

This study and abstract/poster presentation were funded by a research grant from Pfizer Inc.

References

CLSI. *Development of in vitro susceptibility testing criteria and quality control parameters:* 5th ed. CLSI guideline M23. Wayne, PA: Clinical and Laboratory Standards Institute; 2018.

CLSI. *Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically*. 11th ed. CLSI standard M07. Wayne, PA: Clinical and Laboratory Standards Institute; 2018.

CLSI. *Performance standards for antimicrobial susceptibility testing.* 32nd ed. CLSI supplement M100. Clinical and Laboratory Standards Institute; 2022.

Turnidge J, Bordash G (2007). Statistical methods for establishing quality control ranges for antibacterial agents in Clinical and Laboratory Standards Institute susceptibility testing. *Antimicrob Agents Chemother* 51:2483-2488.

Contact

Michael D. Huband, Associate Director
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: michael-huband@jmilabs.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ASMMicrobe2022_M23_Ceftibuten.pdf

ASM Microbe 2022, June 9–13, 2022, Washington, DC