CHEST 2021

Evaluation of the *In Vitro* Activity of Ceftaroline and Comparators against *Streptococcus pneumoniae* Isolates from the United States: Results from 10 Years of the AWARE Surveillance Program (2011–2020)

Helio S. Sader, Cecilia G. Carvalhaes, Jennifer M. Streit, Dee Shortridge, Rodrigo E. Mendes, Mariana Castanheira

JMI Laboratories, North Liberty, Iowa, USA

CONCLUSIONS

Ceftaroline was very active against *S. pneumoniae* from US medical centers, including MDR and XDR isolates and isolates nonsusceptible to ceftriaxone and other antimicrobial agents commonly used to treat CABP.

Ceftaroline has consistently retained potency since its US clinical introduction.

The results of this investigation also indicate that antimicrobial susceptibility of *S. pneumoniae* improved in the 2011–2015 period and then remained stable in the 2015–2020 period in US medical centers participating in the AWARE program.

Increases in susceptibility rates could be related to the anti-pneumococcal vaccine PVC-13 introduced in 2010.

Continued surveillance is crucial since resistance clones not covered by the vaccine may expand.

INTRODUCTION

- The epidemiology of *S. pneumoniae* in the US is constantly changing, requiring continuous monitoring of the antimicrobial resistance profile of this organism.
- The superior activity of ceftaroline against *S. pneumoniae* isolates with elevated MICs to other β-lactams has been attributed to ceftaroline's higher affinity for altered penicillin binding protein targets, including PBP-1A, -2B, and -2X, which are associated with β-lactam resistance in MRSA.
- Ceftaroline fosamil is approved by the US FDA and the European Medicines Agency (EMA) for treating community-acquired bacterial pneumonia (CABP) and acute bacterial skin and skin structure infection (ABSSSI) in adults and children 2 months of age and older, including ABSSSI caused by MRSA.
- The Assessing Worldwide Antimicrobial Resistance Evaluation (AWARE) Program has monitored ceftaroline activity against bacterial organisms in US medical centers since 2008.
- The aim of this investigation is to describe the *in vitro* activity of ceftaroline and comparator agents against *S. pneumoniae,* including multidrug-resistant (MDR) and extensively drug-resistant (XDR) isolates, collected from US medical centers participating in the AWARE Program from 2011 through 2020.

MATERIALS AND METHODS

Bacterial isolates

- 7,901 *S. pneumoniae* isolates were collected from patients in 28 medical centers that participated in the AWARE Program during the entire investigation (2011–2020; 20 centers) or at least 8 of the 10 years (8 centers).
 - Only isolates deemed clinically relevant by the submitting laboratory were included (1 isolate per patient infection episode).
 - The isolates were from respiratory tract infections (88.5%), bloodstream infections (8.4%), and other infection types (3.1%).
 - Isolates were submitted to the central monitoring laboratory (JMI Laboratories, North Liberty, Iowa, USA) for identification, susceptibility testing, and molecular characterization, as needed.

Antimicrobial susceptibility testing

- Broth microdilution tests were conducted at the central reference laboratory according to CLSI methods to determine the susceptibility to ceftaroline and comparator antimicrobial agents.
 - Validated MIC panels were manufactured at JMI Laboratories (2015–2020) or by ThermoFisher Scientific (2011–2014) (Cleveland, Ohio, USA).
 - *S. pneumoniae* isolates were tested in cation-adjusted Mueller-Hinton broth supplemented with 2.5–5% lysed horse blood according to CLSI M07 (2018).
 - The quality control (QC) strain *S. pneumoniae* ATCC 49619 was tested concurrently with clinical isolates and all results were within CLSI QC limits.
 - The ceftaroline susceptibility breakpoint applied in this study was ≤0.5 mg/L as published by CLSI, USFDA, and EUCAST.
- MDR status was determined based on nonsusceptibility to ≥3 classes of the following antimicrobial agents: penicillin (MIC, ≥4 mg/L), ceftriaxone (MIC, ≥2 mg/L), erythromycin (MIC, ≥0.5 mg/L), clindamycin (MIC, ≥0.5 mg/L), levofloxacin (MIC, ≥4 mg/L), tetracycline (MIC, ≥2 mg/L), and trimethoprim-sulfamethoxazole (TMP-SMX; MIC, ≥1 mg/L).
- XDR status was determined based on nonsusceptibility to ≥5 antimicrobial classes.
- Further susceptibility analyses were performed for *S. pneumoniae* isolates that tested as nonsusceptible to ceftriaxone, penicillin, amoxicillin-clavulanate, erythromycin, clindamycin, tetracycline, and/or levofloxacin.

RESULTS

- Ceftaroline inhibited 99.99% of *S. pneumoniae* isolates at ≤0.5 mg/L; only 1 isolate had a ceftaroline MIC >0.5 mg/L (at 1 mg/L; Table 1).
- Ceftaroline retained potent activity against all resistant subgroups, including amoxicillin-clavulanate-nonsusceptible (NS; 8.9% at ≥4 mg/L), penicillin-NS (6.2% at ≥4 mg/L), ceftriaxone-NS (5.1% at ≥2 mg/L), erythromycin-NS (45.0% at ≥0.5 mg/L), clindamycin-NS (16.2% at ≥0.5 mg/L), tetracycline-NS (21.6% at ≥2 mg/L), and levofloxacin-NS (0.7% at ≥4 mg/L; Figure 1).
- Ceftaroline activity remained stable against all resistant subgroups each year (data not shown).
- Susceptibility to penicillin, ceftriaxone, amoxicillin-clavulanate, clindamycin, trimethoprim-sulfamethoxazole, and tetracycline increased from 2011 to 2015 and remained relatively stable from 2016 to 2020 (Figure 2).
- MDR and XDR rates decreased from 2011 to 2015 and remained stable from 2016 to 2020 (Figure 3).
- Susceptibility to erythromycin ranged from 52.2% (2014) to 56.4% (2011) and levofloxacin was very active during the study period (98.9-100.0% susceptible [S]; Figure 2).
- MDR rates varied from 23.7% in 2011 to 16.2% in 2020 (19.7% overall) and XDR rates varied from 13.3% in 2011 to 1.8% in 2019 (6.1% overall; Figure 3).
- Against MDR isolates (n=1,555), ceftaroline ($MIC_{50/90}$, 0.06/0.25 mg/L; 99.9%S), levofloxacin ($MIC_{50/90}$, 1/1 mg/L; 98.1%S), linezolid ($MIC_{50/90}$, 1/1 mg/L; 100.0%S), and vancomycin ($MIC_{50/90}$, 0.25/0.5 mg/L; 100.0%S) were the most active agents (Table 1).
- All XDR isolates were susceptible to ceftaroline ($MIC_{50/90}$, 0.12/0.25 mg/L), linezolid ($MIC_{50/90}$, 0.5/1 mg/L), and vancomycin ($MIC_{50/90}$, 0.25/0.5 mg/L) and 97.5% were susceptible to tigecycline ($MIC_{50/90}$, ≤0.03/0.06 mg/L; Table 1).
- The ceftaroline-NS isolate was isolated in 2014 and had multiple substitutions in the penicillin binding proteins (PBP), mainly PBP2x, when compared with reference sequences. It also showed 31 amino acid alterations in MurM (data not shown).

Table 1. Activity of ceftaroline and comparator antimicrobial agents when tested against *S. pneumoniae* isolates from US medical centers (2011–2020)

Organism group/ antimicrobial agent	mg/L		CLSI[a]		
	MIC_{50}	MIC_{90}	%S	%I	%R
All isolates (7,901)					
Ceftaroline	≤0.015	0.12	>99.9		
Ceftriaxone	≤0.06	1	83.9 [b] 94.9 [c]	10.9 4.4	5.1 0.7
AMX-CLA (2:1 ratio)	≤1	2	91.1	3.2	5.6
Penicillin	≤0.06	2	61.2 [d] 61.2 [e] 93.8 [f]	25.2 5.6	13.6 38.8 0.6
Erythromycin	≤0.12	>2	55.0	0.7	44.2
Clindamycin	≤0.25	>1	83.8	0.6	15.5
Levofloxacin	1	1	99.3	0.1	0.6
Linezolid	1	1	100.0		
Tetracycline	≤0.5	>4	78.4	0.3	21.3
TMP-SMX	≤0.5	>4	70.1	11.7	18.1
Tigecycline	≤0.03	0.06	97.7 [g]		
Vancomycin	0.25	0.5	100.0		
MDR (1,555)					
Ceftaroline	0.06	0.25	99.9		
Ceftriaxone	0.5	2	55.9 [b] 74.8 [c]	18.8 21.8	25.2 3.4
AMX-CLA (2:1 ratio)	≤1	>4	63.6	8.3	28.1
Penicillin	0.5	4	11.6 [d] 11.6 [e] 68.7 [f]	46.6 28.4	41.8 88.4 3.0
Erythromycin	>2	>2	0.0	0.5	99.5
Clindamycin	>1	>1	23.1	1.7	75.2
Levofloxacin	1	1	98.1	0.3	1.6
Linezolid	1	1	100.0		
Tetracycline	>4	>4	5.5	0.8	93.8
TMP-SMX	4	>4	24.8	24.4	50.7
Tigecycline	≤0.03	0.06	97.4 [g]		
Vancomycin	0.25	0.5	100.0		
XDR (480)					
Ceftaroline	0.12	0.25	100.0		
Ceftriaxone	2	2	1.9 [b] 26.5 [c]	24.6 64.6	73.5 9.0
AMX-CLA (2:1 ratio)	>4	>4	4.6	12.7	82.7
Penicillin	4	4	0.2 [d] 0.2 [e] 6.0 [f]	1.3 85.2	98.5 99.8 8.8
Erythromycin	>2	>2	0.0	0.0	100.0
Clindamycin	>1	>1	4.6	0.2	95.2
Levofloxacin	1	1	96.7	0.4	2.9
Linezolid	0.5	1	100.0		
Tetracycline	>4	>4	2.5	0.2	97.3
TMP-SMX	>4	>4	0.6	1.2	98.1
Tigecycline	≤0.03	0.06	97.5 [g]		
Vancomycin	0.25	0.5	100.0		

[a] Criteria as published by CLSI (2021).
[b] Using meningitis breakpoints.
[c] Using non-meningitis breakpoints.
[d] Using oral breakpoints.
[e] Using parenteral, meningitis breakpoints.
[f] Using parenteral, non-meningitis breakpoints.
[g] US FDA breakpoints were applied.
Abbreviations: AMX-CLA, amoxicillin-clavulanate; TMP-SMX, trimethoprim-sulfamethoxazole; MDR, multidrug-resistant; XDR, extensively drug-resistant.

Figure 1. Ceftaroline activity (MIC distributions) against resistant subsets of *S. pneumoniae* (2011–2020)

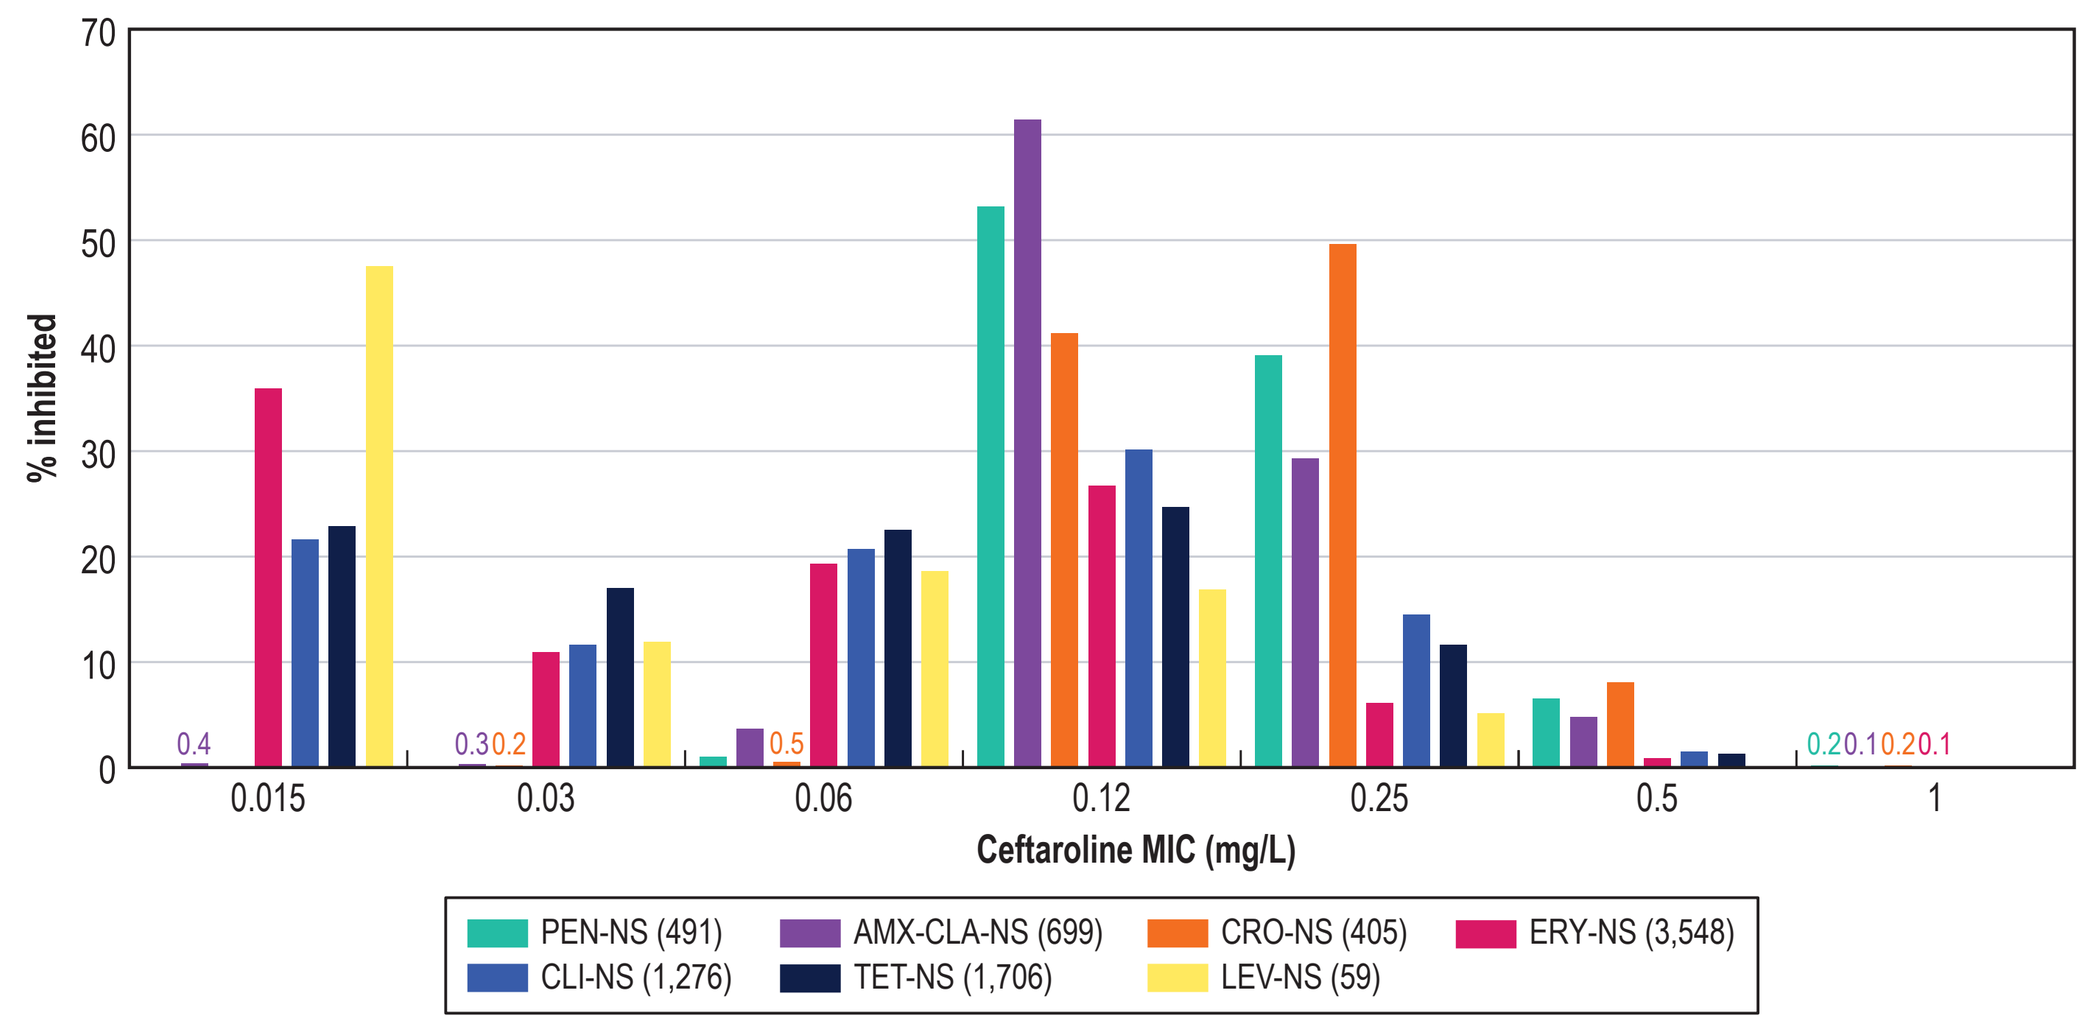
% inhibited
Ceftaroline MIC (mg/L)
0.015
0.03
0.06
0.12
0.25
0.5
1
PEN-NS (491)
AMX-CLA-NS (699)
CRO-NS (405)
ERY-NS (3,548)
CLI-NS (1,276)
TET-NS (1,706)
LEV-NS (59)

Abbreviations: PEN, penicillin (MIC ≥4 mg/L); NS, nonsusceptible; AMX-CLA, amoxicillin-clavulanate; CRO, ceftriaxone (MIC ≥2 mg/L); ERY, erythromycin; CLI, clindamycin; TET, tetracycline; LEV, levofloxacin.

Figure 2. Activity of ceftaroline and comparator antimicrobial agents when tested against *S. pneumoniae* isolates from US medical centers (2011–2020)

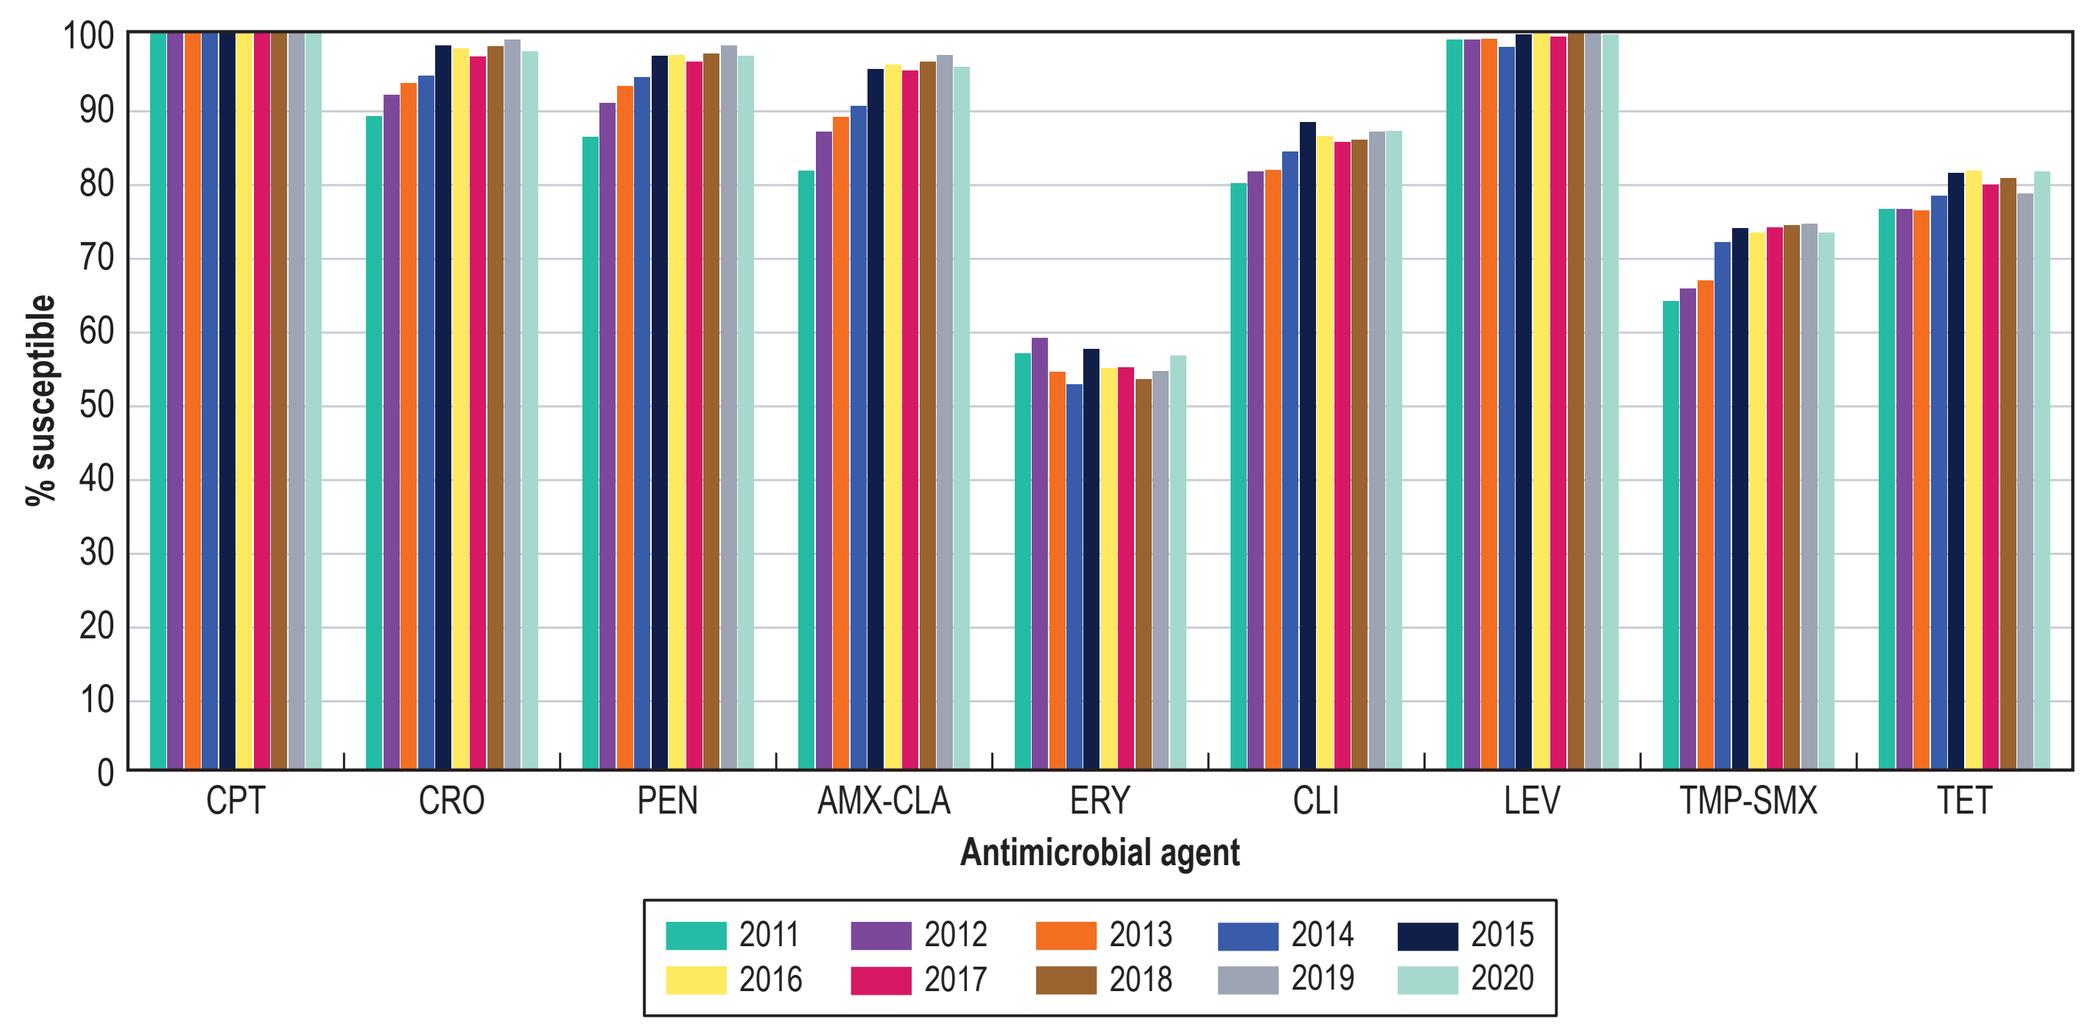
% susceptible
Antimicrobial agent
CPT
CRO
PEN
AMX-CLA
ERY
CLI
LEV
TMP-SMX
TET
2011
2012
2013
2014
2015
2016
2017
2018
2019
2020

Abbreviations: CPT, ceftaroline; CRO, ceftriaxone; PEN, penicillin; AMX-CLA, amoxicillin-clavulanate; ERY, erythromycin; CLI, clindamycin; LEV, levofloxacin; TMP-SMX, trimethoprim-sulfamethoxazole; TET, tetracycline.

Figure 3. Yearly frequency (%) of MDR and XDR phenotypes among *S. pneumoniae* from the United States (2011–2020)

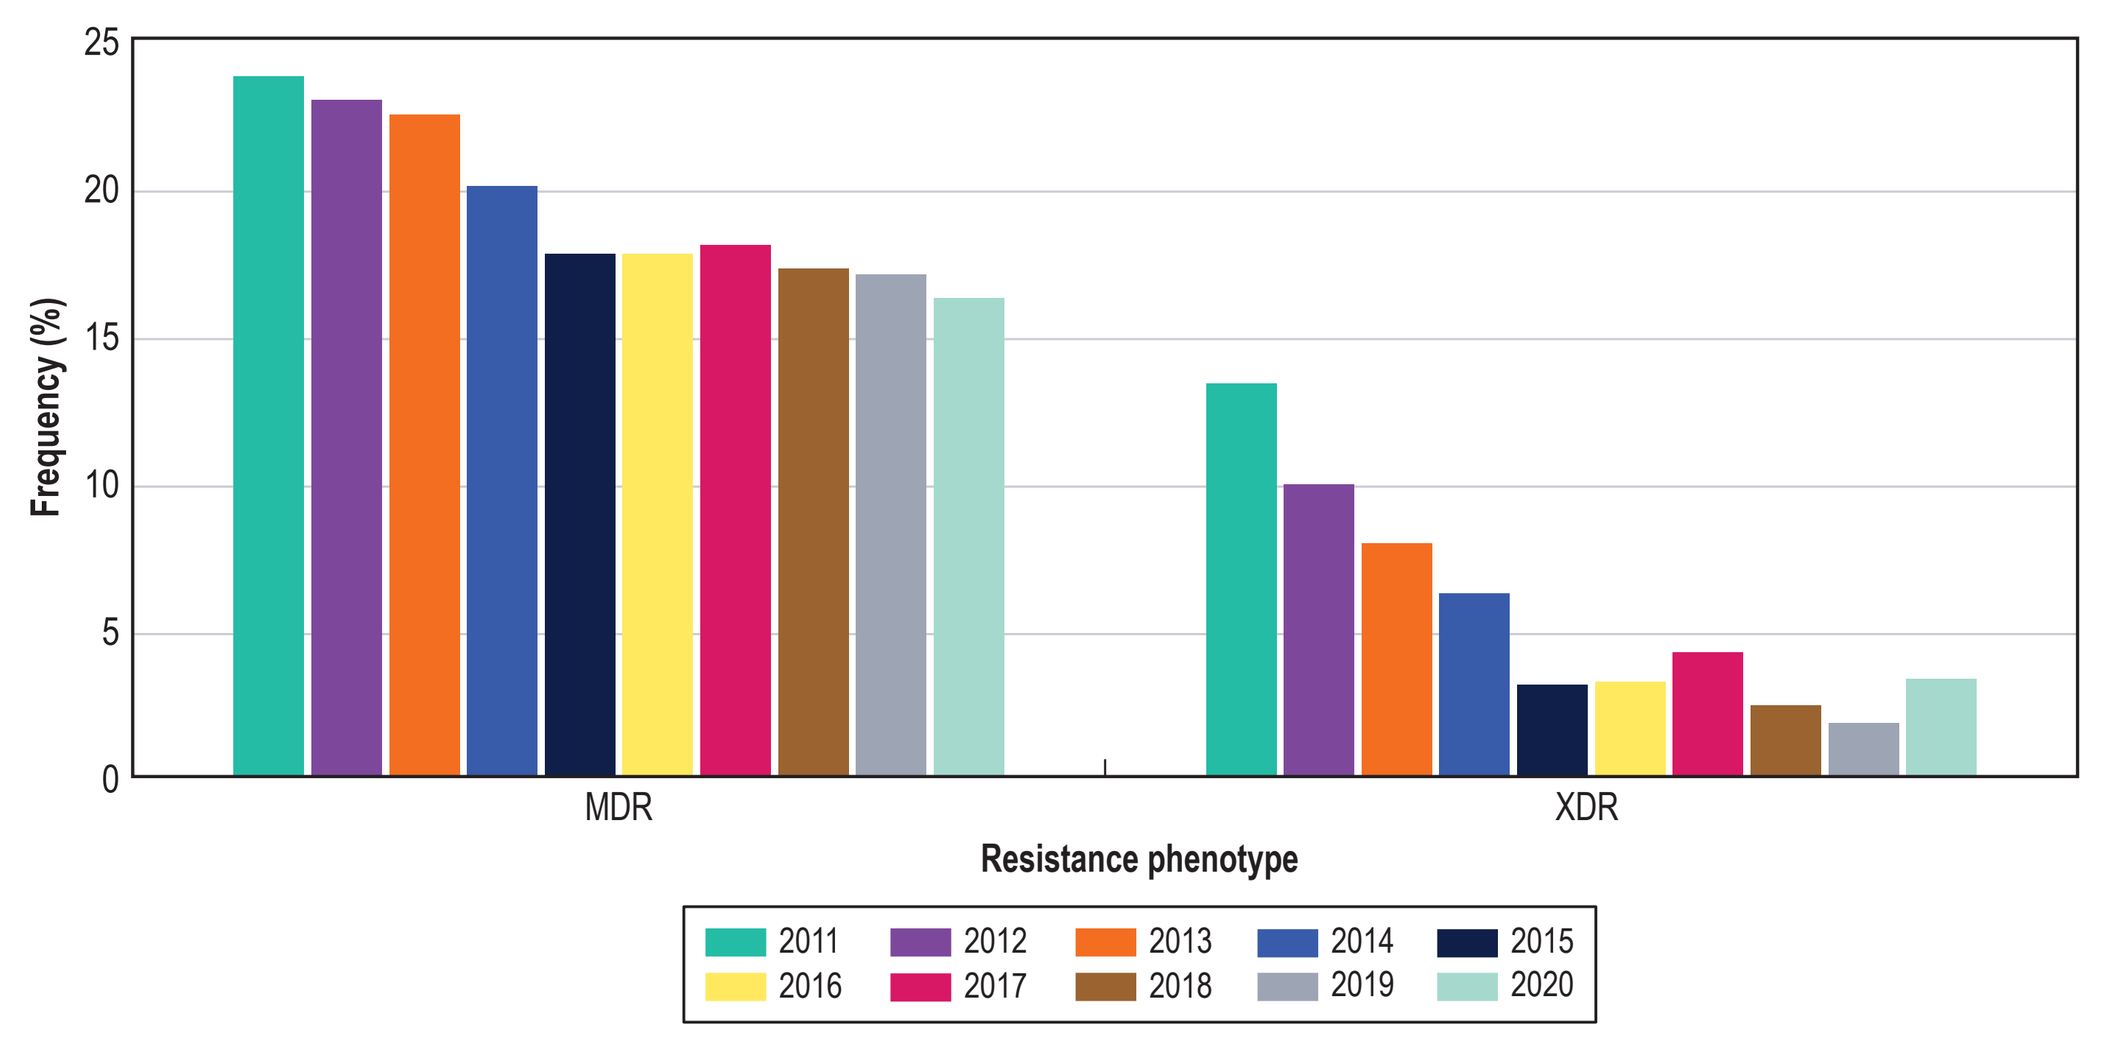
Frequency (%)
Resistance phenotype
MDR
XDR
2011
2012
2013
2014
2015
2016
2017
2018
2019
2020

Contact Information

Helio S. Sader, MD, PhD, FIDSA
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie CHEST 2021 scientific presentations:

https://abbvie1.outsystemsenterprise.com/GMAEventPublications/Assets.aspx?ConferenceId=261

QR code expiration: October 17, 2022

To submit a medical question, please visit www.abbviemedinfo.com

Acknowledgements

The authors would like to thank all participants of the Assessing Worldwide Antimicrobial Resistance Evaluation (AWARE) Program for providing bacterial isolates.
This study was supported by AbbVie. AbbVie was involved in the design and decision to present these results, and JMI Laboratories received compensation for services in relation to preparing this presentation. AbbVie had no involvement in the collection, analysis, and interpretation of data.

References

1. Clinical and Laboratory Standards Institute (2021). *M100Ed31E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition.* Wayne, PA: CLSI.
3. Golden AR, Rosenthal M, Fultz B, et al. (2015). Characterization of MDR and XDR *Streptococcus pneumoniae* in Canada, 2007–13. *J Antimicrob Chemother 70:2199–202.*
4. Pani A, Colombo F, Agnelli F, et al. (2019) Off-label use of ceftaroline fosamil: A systematic review. *Int J Antimicrob Agents* 54:562-71.
5. Teflaro® Package Insert (2021). Available at http://www.allergan.com/assets/pdf/teflaro_pi.

abbvie