ECCMID 2021 | Poster #722

Antimicrobial Activities of Aztreonam-Avibactam and Comparator Agents Tested against *Enterobacterales* from European Hospitals Analysed by Infection Type (2019–2020)

Helio S. Sader, Rodrigo E. Mendes, Cecilia G. Carvalhaes, S. J. Ryan Arends, Mariana Castanheira
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Aztreonam is a monobactam stable to hydrolysis by metallo-β-lactamases (MBL) and avibactam is a non-β-lactam β-lactamase inhibitor that inhibits Ambler class A, C, and some class D enzymes (e.g., ESBL, KPC, and AmpC).
- Because *Enterobacterales* isolates that produce MBLs usually coproduce serine β-lactamases, aztreonam was combined with avibactam. This novel β-lactamase-inhibitor combination is under clinical development for the treatment of gram-negative infections.
- We assessed the *in vitro* activity of aztreonam-avibactam against a large collection of contemporary (2019–2020) clinical isolates recovered from patients hospitalised with pneumonia in European medical centres.

Materials and Methods

Bacterial isolates

- A total of 11,253 isolates were consecutively collected from 38 medical centres: 25 from Western Europe (W-EU; n=8,511; 10 countries) and 13 from Eastern Europe (E-EU; n=2,742; 10 countries).
- These isolates were collected from patients with bloodstream infections (BSIs; 3,352 isolates; 29.8%), pneumonia (2,233; 19.8%), skin and soft tissue infections (SSTIs; 1,820; 16.2%), complicated urinary tract infections (cUTIs; 2,741; 24.4%), and other infection types (1,107; 9.8%).
- Only isolates determined to be significant by local criteria as the reported probable cause of infection were included in the program.

Resistant subsets

- Carbapenem-resistant *Enterobacterales* (CRE) isolates were defined as displaying imipenem and/or meropenem MIC values at ≥4 mg/L (CLSI, 2021).
 - Imipenem was not applied to *Proteus mirabilis* and indole-positive *Proteeae* due to their intrinsically elevated MIC values.
- MDR and extensively drug-resistant (XDR) *Enterobacterales* isolates were classified according to their recommended guidelines (Magiorakos et al., 2012).
- Classifications were based on the following recommended parameters:
 - MDR = nonsusceptible (NS; CLSI breakpoints) to at least 3 antimicrobial classes.
 - XDR = susceptible (S) to 2 or fewer antimicrobial classes.

Susceptibility testing

- The broth microdilution test method was conducted according to CLSI specifications.
- Aztreonam-avibactam was tested with avibactam at a fixed concentration of 4 mg/L.
- EUCAST susceptibility interpretive criteria were applied for comparator agents and a tentative susceptible breakpoint of ≤8 mg/L was applied for ATM-AVI for comparison purposes.

Results

- Aztreonam-avibactam was active against 99.9-100.0% of W-EU isolates and 99.6-99.9% of E-EU isolates at ≤8 mg/L (Table 1), with $MIC_{50/90}$ values of ≤0.03/0.12 mg/L in W-EU and 0.06/0.25 mg/L in E-EU.
- Aztreonam-avibactam retained potent activity against CRE, inhibiting 100.0% of W-EU isolates and 98.9-100.0% of E-EU isolates at ≤8 mg/L (Figure 1).
- Susceptibility to comparator agents were consistently lower among isolates from E-EU compared to W-EU for all evaluated infection types (Table 1).
- MDR rates among *Enterobacterales* varied from 8.7% (UTI) to 12.8% (BSI) in W-EU and from 28.1% (UTI) to 39.0% (pneumonia) in E-EU (Figure 2).
- XDR rates among *Enterobacterales* varied from 0.6% (SSTI) to 1.4% (BSI) in W-EU and from 6.6% (UTI) to 17.0% (pneumonia) in E-EU.
- Carbapenem-resistance (CRE) rates varied from 0.7% (UTI and SSTI) to 1.7% (BSI) in W-EU and from 5.9% (UTI) to 17.0% (pneumonia) in E-EU (Figure 2).
- In W-EU, susceptibility to meropenem, levofloxacin, and gentamicin were lowest among isolates from BSI, and susceptibility to ceftriaxone, ceftolozane-tazobactam (C-T), and piperacillin-tazobactam (PIP-TAZ) were lowest among isolates from pneumonia (Table 1).
- In E-EU, susceptibility rates were generally lowest among isolates from pneumonia and highest among isolates from UTI (Table 1).
- The most active compounds against MDR *Enterobacterales* were aztreonam-avibactam (100.0% and 99.1%-99.5% inhibited at ≤8 mg/L in W-EU and E-EU, respectively), meropenem (89.0-93.5%S in W-EU and 61.0-80.4%S in E-EU), amikacin (76.0-87.1%S in W-EU and 58.7-68.9%S in E-EU), and colistin (67.4-79.5%S in W-EU and 68.9-83.2%S in E-EU; Table 1).
- The most active compounds against XDR *Enterobacterales* were aztreonam-avibactam (100.0% and 98.9%-100.0% inhibited at ≤8 mg/L in W-EU and E-EU, respectively), colistin (76.5-100.0%S in W-EU and 48.4-66.8%S in E-EU), and amikacin (41.2-85.7%S in W-EU and 16.4-29.4%S in E-EU; Table 1).

Figure 1. Cumulative MIC distributions of aztreonam-avibactam tested against carbapenem-resistant *Enterobacterales* (CRE) isolates from Western Europe (A) and Eastern Europe (B) stratified by infection type

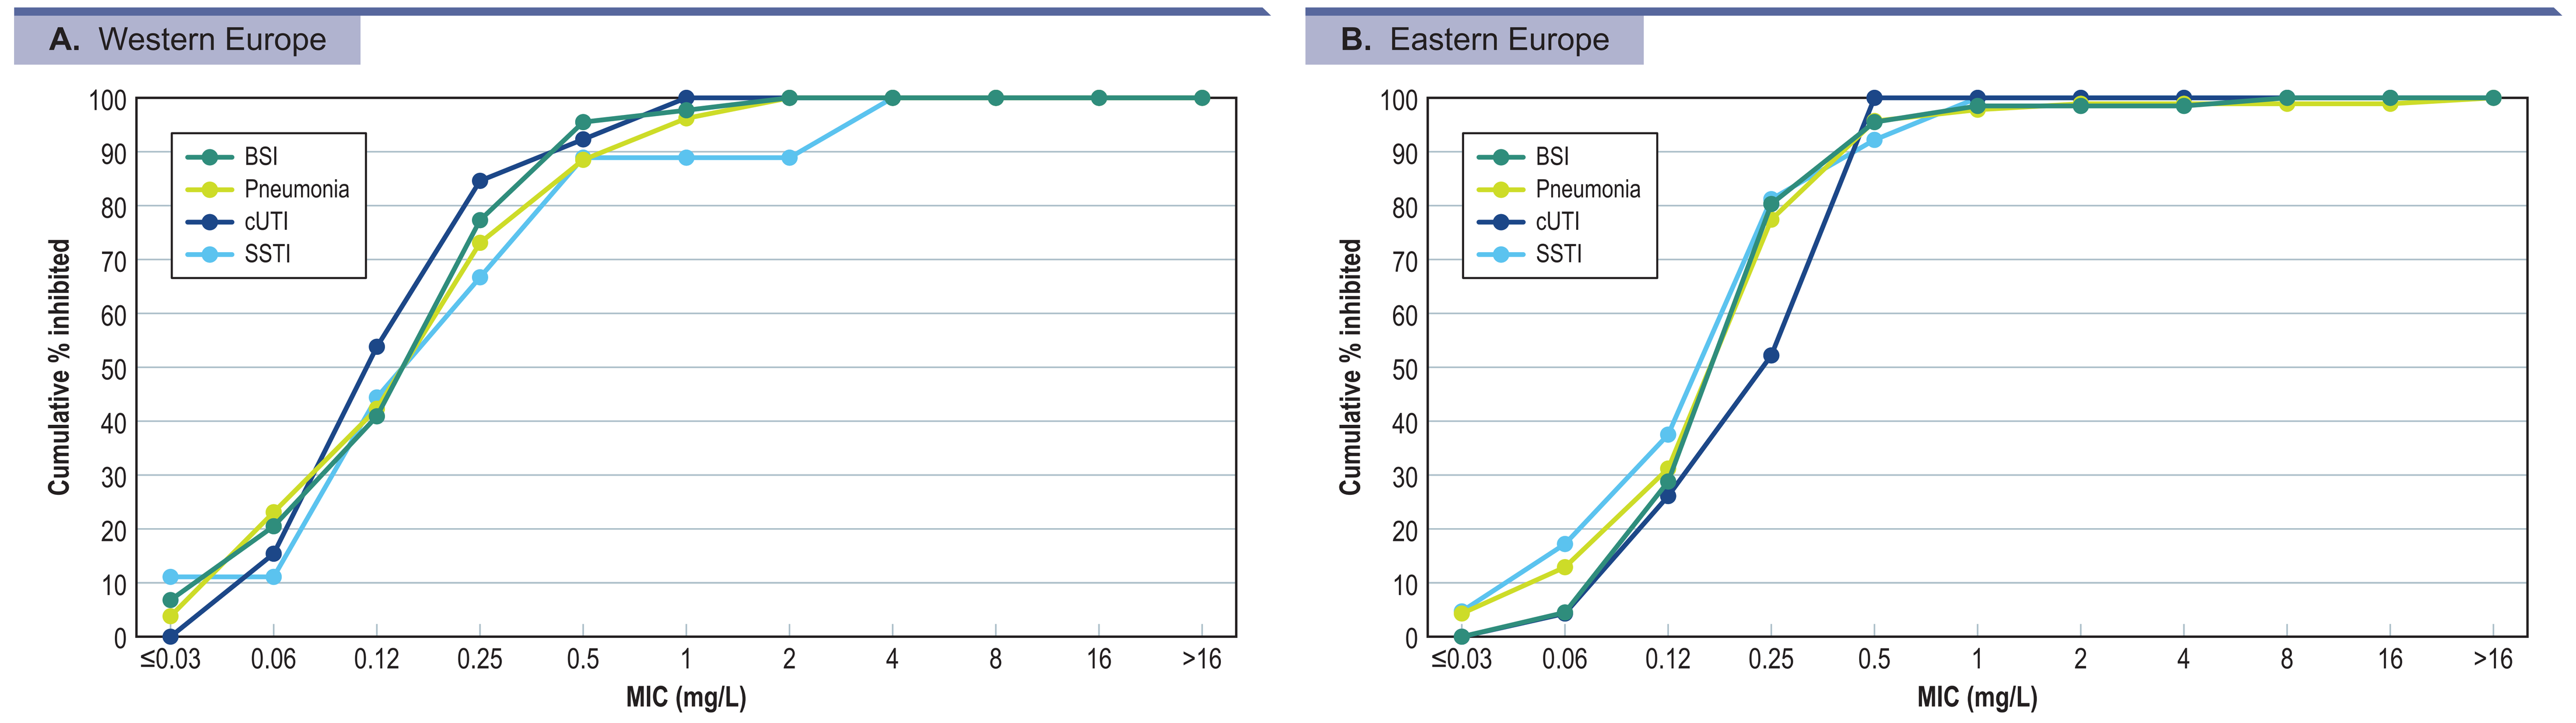
A. Western Europe
B. Eastern Europe
BSI
Pneumonia
cUTI
SSTI
Cumulative % inhibited
MIC (mg/L)
≤0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
>16

Abbreviations: BSI, bloodstream infection; cUTI, complicated urinary tract infection; SSTI, skin and soft tissue infection.

Figure 2. Frequency of occurrence of multidrug-resistant (MDR), extensively drug-resistant (XDR), and carbapenem-resistant (CRE) *Enterobacterales* isolates from Western Europe (A) and Eastern Europe (B) stratified by infection type

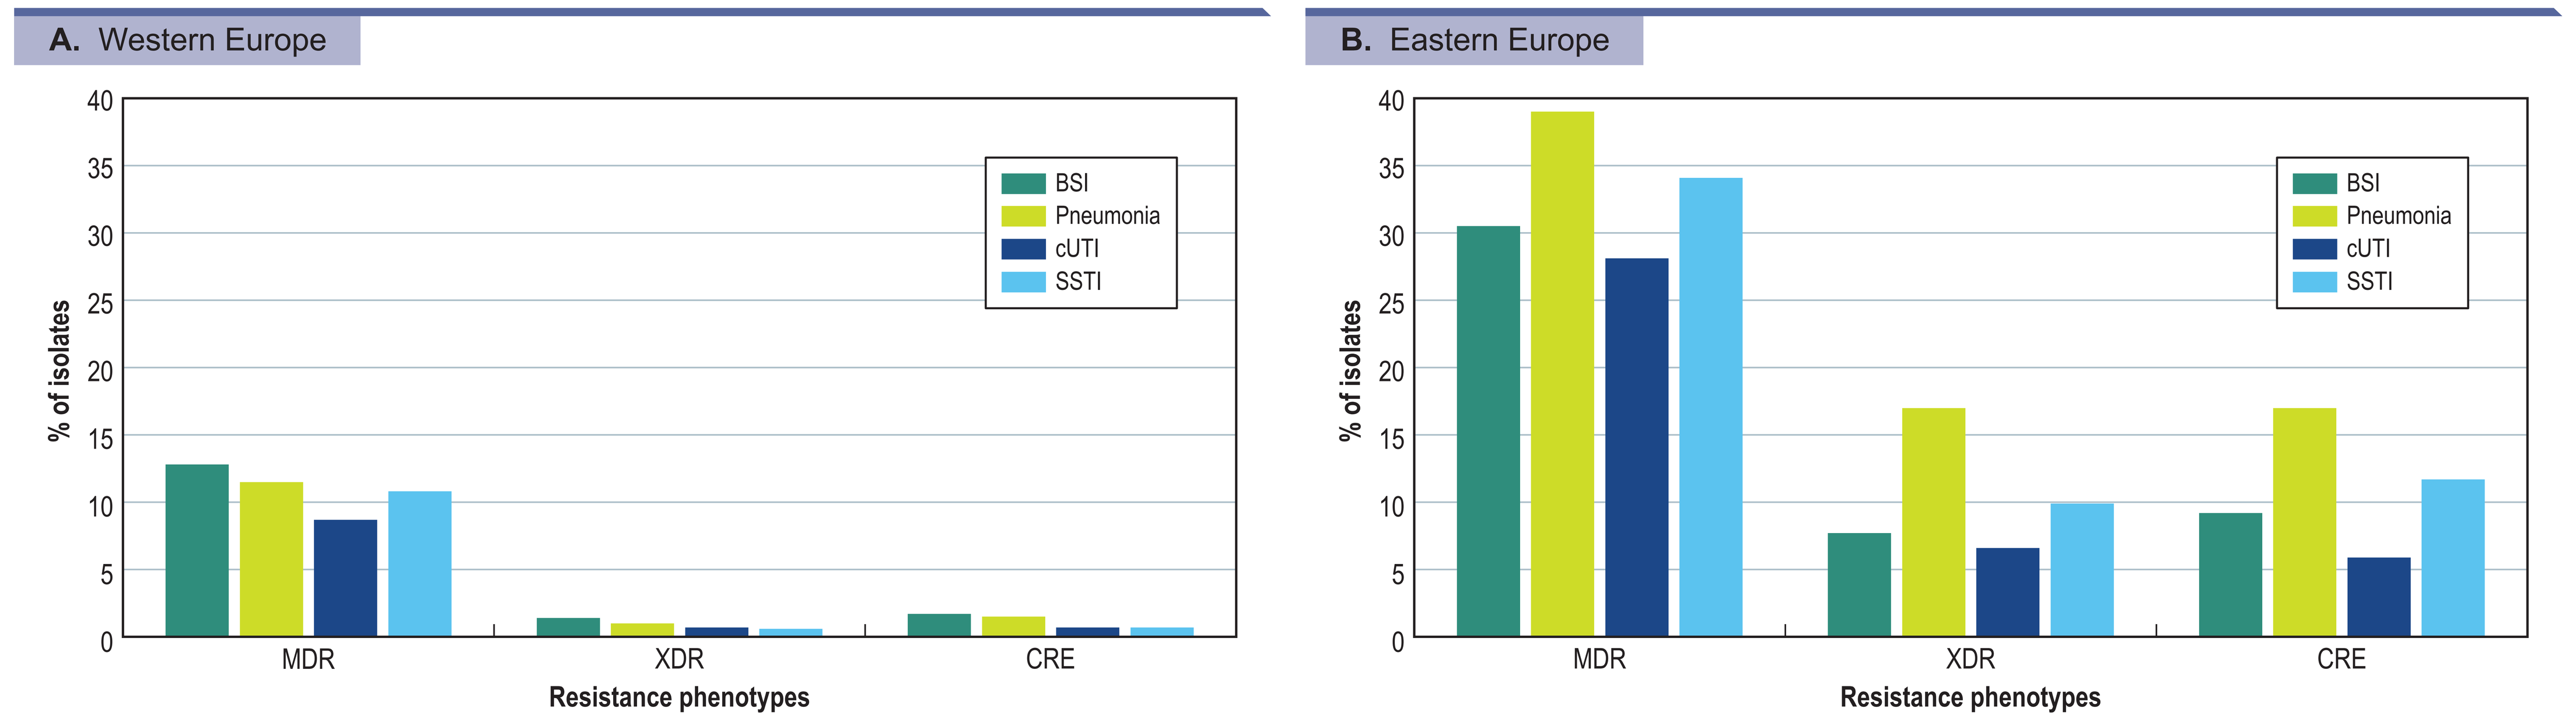
A. Western Europe
B. Eastern Europe
BSI
Pneumonia
cUTI
SSTI
% of isolates
MDR
XDR
CRE
Resistance phenotypes

Abbreviations: MDR, multidrug-resistant; XDR, extensively drug-resistant; CRE, carbapenem-resistant *Enterobacterales*; BSI, bloodstream infection; UTI, urinary tract infection; SSTI, skin and soft tissue infection.

Table 1. Antimicrobial activity of aztreonam-avibactam (AZT-AVI) and comparator agents tested against *Enterobacterales* isolates from European hospitals stratified by geographic region and infection type

Organism/ Antimicrobial agent	% Susceptible per EUCAST criteria (no. tested)							
	Western Europe				Eastern Europe			
	BSI	Pneumonia	UTI	SSTI	BSI	Pneumonia	UTI	SSTI
Enterobacterales	**(2,635)**	**(1,687)**	**(1,963)**	**(1,272)**	**(717)**	**(546)**	**(778)**	**(548)**
AZT-AVI	[100.0][a]	[100.0][a]	[99.9][a]	[100.0][a]	[99.9][a]	[99.6][a]	[99.9][a]	[99.8][a]
Meropenem	98.2	98.8	99.4	99.3	91.9	84.8	94.3	89.4
Ceftriaxone	78.1	77.2	84.5	80.3	59.0	52.4	62.5	55.8
Ceftolozane-tazobactam	94.3	92.0	96.2	93.9	81.9	70.6	85.2	77.0
Piperacillin-tazobactam	85.2	80.0	90.5	84.8	70.3	59.3	74.3	64.8
Levofloxacin	78.0	83.0	81.4	81.6	58.4	57.4	56.4	58.2
Gentamicin	88.2	92.6	90.8	91.4	78.2	74.7	79.0	77.4
Amikacin	96.5	98.5	98.2	98.3	88.3	83.7	90.7	86.7
Colistin	88.4	76.5	85.7	73.5	84.9	77.0	84.1	81.8
CRE	**(44)**	**(26)**	**(13)**	**(9)**	**(66)**	**(93)**	**(46)**	**(64)**
AZT-AVI	[100.0][a]	[100.0][a]	[100.0][a]	[100.0][a]	[100.0][a]	[98.9][a]	[100.0][a]	[100.0][a]
Levofloxacin	6.8	23.1	23.1	0.0	15.2	8.7	2.2	4.7
Gentamicin	56.8	57.7	38.5	33.3	50.0	40.9	23.9	40.6
Amikacin	65.9	65.4	53.8	88.9	37.9	37.6	34.8	40.6
Colistin	93.2	88.5	92.3	100.0	75.8	67.7	73.9	82.8
MDR	**(327)**	**(194)**	**(170)**	**(138)**	**(219)**	**(213)**	**(219)**	**(187)**
AZT-AVI	[100.0][a]	[100.0][a]	[100.0][a]	[100.0][a]	[99.5][a]	[99.1][a]	[99.5][a]	[99.5][a]
Meropenem	89.0	89.2	93.5	93.5	74.0	61.0	80.4	69.0
Ceftriaxone	9.2	17.0	20.0	19.6	9.1	3.8	10.0	8.6
Ceftolozane-tazobactam	72.7	66.0	71.8	72.5	47.0	31.1	53.9	42.8
Piperacillin-tazobactam	37.7	25.8	38.2	36.2	20.1	12.2	28.8	15.0
Levofloxacin	14.3	24.7	15.9	22.5	14.6	9.4	5.9	11.2
Gentamicin	31.2	51.5	37.1	44.2	39.7	36.6	36.1	41.2
Amikacin	76.0	87.1	81.2	85.5	64.4	58.7	68.9	61.0
Colistin	79.5	68.0	77.5	67.4	74.8	68.9	70.6	83.2
XDR	**(36)**	**(17)**	**(13)**	**(7)**	**(55)**	**(93)**	**(51)**	**(54)**
AZT-AVI	[100.0][a]	[100.0][a]	[100.0][a]	[100.0][a]	[99.5][a]	[98.9][a]	[100.0][a]	[100.0][a]
Meropenem	22.2	17.6	15.4	0.0	21.8	22.6	25.5	18.5
Ceftriaxone	2.8	0.0	0.0	0.0	0.0	2.2	0.0	1.9
Ceftolozane-tazobactam	2.8	0.0	0.0	0.0	3.6	6.5	3.9	11.1
Piperacillin-tazobactam	0.0	0.0	0.0	0.0	0.0	0.0	0.0	0.0
Levofloxacin	0.0	11.8	15.4	0.0	5.5	2.2	2.0	3.7
Gentamicin	33.3	35.3	30.8	14.3	23.6	24.7	13.7	20.4
Amikacin	52.8	41.2	46.2	85.7	16.4	28.0	29.4	25.9
Colistin	83.3	76.5	84.6	100.0	58.2	48.4	56.9	66.8

[a] Values in brackets indicate percentages inhibited at ≤8 mg/L.
Abbreviations: CRE, carbapenem-resistant *Enterobacterales*; MDR, multidrug-resistant; XDR, extensively-drug resistant.

Conclusions

- Aztreonam-avibactam demonstrated potent and consistent activity against *Enterobacterales* from all infection types in W-EU and E-EU.
- Susceptibility rates for comparators were generally lower among isolates from patients with pneumonia and BSI compared to UTI and SSTI.
- Susceptibility rates for comparators were markedly lower among isolates from E-EU compared to W-EU.
- Aztreonam-avibactam represents a potential valuable option for empiric antimicrobial therapy in European hospitals with elevated rates of CRE, MDR, or XDR *Enterobacterales*.

Acknowledgements

This study at JMI Laboratories was supported by Pfizer Inc. (New York, NY). JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by Pfizer Inc.

References

Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition*. Wayne, PA: CLSI.

Cornely OA, Cisneros JM, Torre-Cisneros J, et al. (2020). Pharmacokinetics and safety of aztreonam/avibactam for the treatment of complicated intra-abdominal infections in hospitalized adults: results from the REJUVENATE study. *Journal of Antimicrobial Chemotherapy* 75:618-627.

Magiorakos AP, Srinivasan A, Carey RB, et al. (2012). Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. *Clin Microbiol Infect* 18: 268-281.

Sader HS, Carvalhaes CG, Arends SJR, Castanheira M, Mendes RE (2021). Aztreonam/avibactam activity against clinical isolates of *Enterobacterales* collected in Europe, Asia and Latin America in 2019. *Journal of Antimicrobial Chemotherapy* 76:659-666.

EUCAST (2021). Breakpoint tables for interpretation of MICs and zone diameters. Version 11.0, January 2021. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_11.0_Breakpoint_Tables.pdf

Contact

Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit http://www.jmilabs.com/data/posters/ECCMID2018-ceftaroline-potency-spectrum.pdf

Charges may apply. No personal information is stored.

ECCMID 2021, July 9–12, 2021