ECCMID 2022 | Poster Number: 567 1

Activity of Meropenem-Vaborbactam and Comparators Against European Carbapenem-Resistant Enterobacterales Isolates Producing KPC Carbapenemase (2018-2020)

Dee Shortridge, Lalitagauri M. Deshpande, Jennifer M. Streit, Mariana Castanheira

JMI Laboratories, North Liberty, Iowa

Objective

Meropenem-vaborbactam (MVB) is a combination of a carbapenem, and a carbapenemase inhibitor developed to inhibit Class A and C beta-lactamases, including the common *Klebsiella pneumoniae* carbapenemase (KPC).

MVB was approved by the European Medicines Agency for the treatment of complicated urinary tract infections (cUTI), including acute pyelonephritis, complicated intra-abdominal infections (cIAI), hospital-acquired bacterial pneumonia, ventilator-associated pneumonia, and bacteremia (BSI) associated with any of the infections listed above.

The objective of this study was to analyse the activity of MVB and comparators against KPC-producing carbapenem-resistant Enterobacterales (CRE) collected in European hospitals from 2018 to 2020 as part of the SENTRY Antimicrobial Surveillance Program.

Methods

- A total of 16,866 Enterobacterales clinical isolates were consecutively collected from hospitalized patients in 39 sites from 19 European countries.
- Susceptibility testing was performed using the CLSI broth microdilution method. EUCAST (2022) interpretive criteria were used.
- All infection types were included (Figure 1).
- 679 CRE were identified as having an MIC >2 mg/L to meropenem and/or imipenem. Whole genome sequencing was performed on each CRE isolate; 227 isolates contained bla_{KPC}.
 - 226/227 isolates with bla_{KPC} also contained 1-6 additional beta-lactamase enzymes including various bla_{TEM}, bla_{SHV} and $bla_{CTX\text{-}M}$. 6 isolates also contained a metallo-beta-lactamase, $bla_{VIM\text{-}1}$ or $bla_{NDM\text{-}1}$.
 - Species containing bla_{KPC}: *Citrobacter freundii* species complex (1), *Enterobacter cloacae* species complex (2), *Escherichia coli* (6), and *Klebsiella pneumoniae* (218).

Results

Figure 1. KPC-Producing Isolates by Infection Type

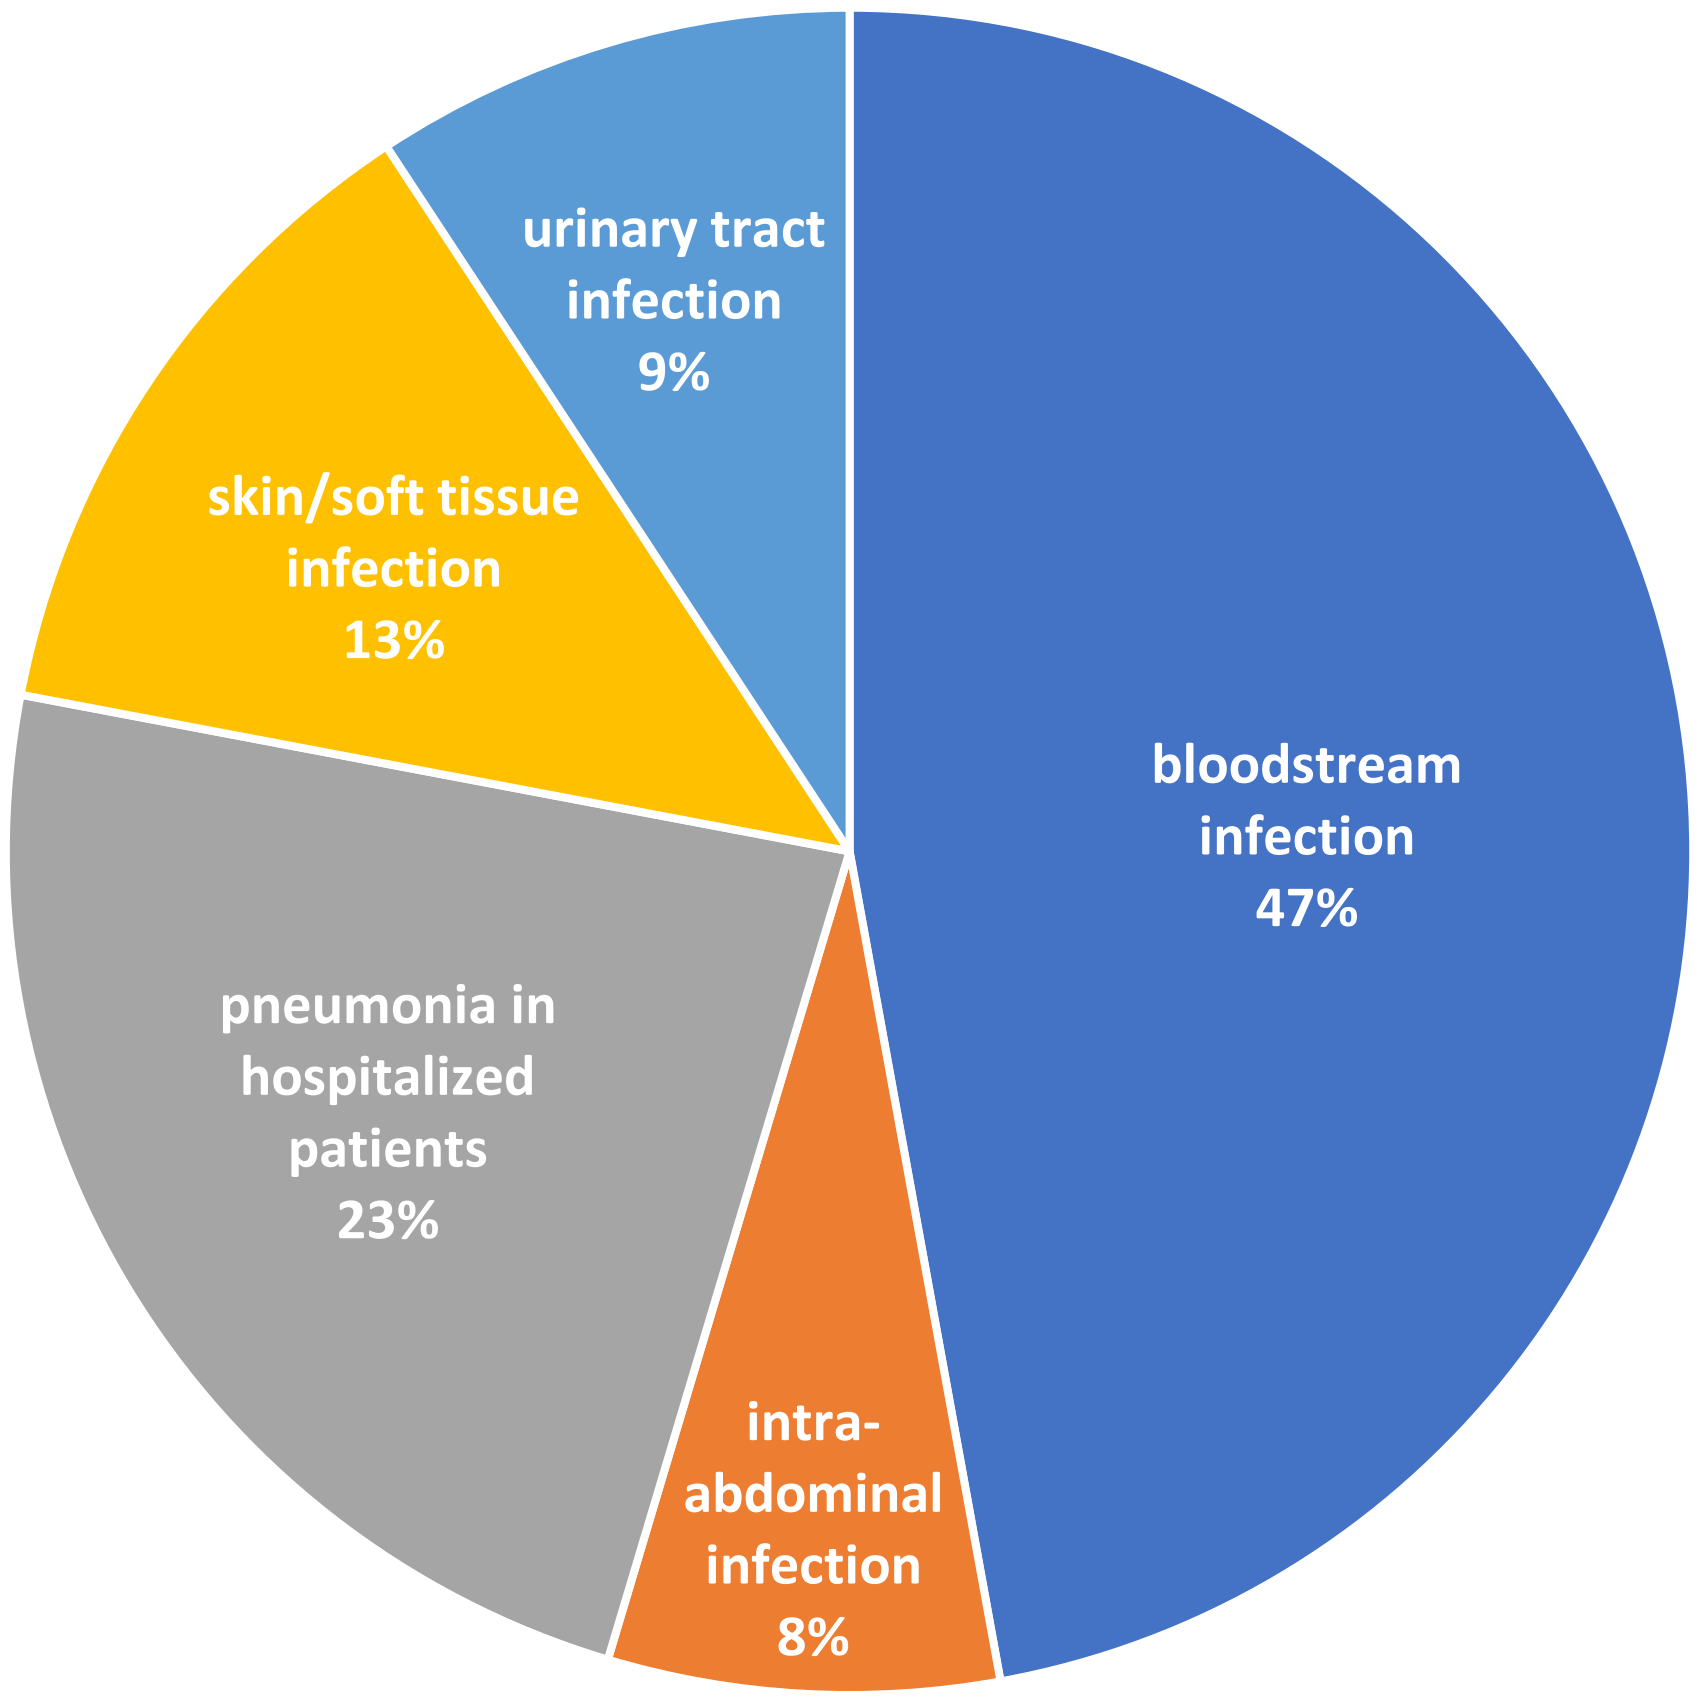
urinary tract infection 9%
skin/soft tissue infection 13%
bloodstream infection 47%
pneumonia in hospitalized patients 23%
intra-abdominal infection 8%

Figure 2. KPC-Producing Isolates by Country and Year

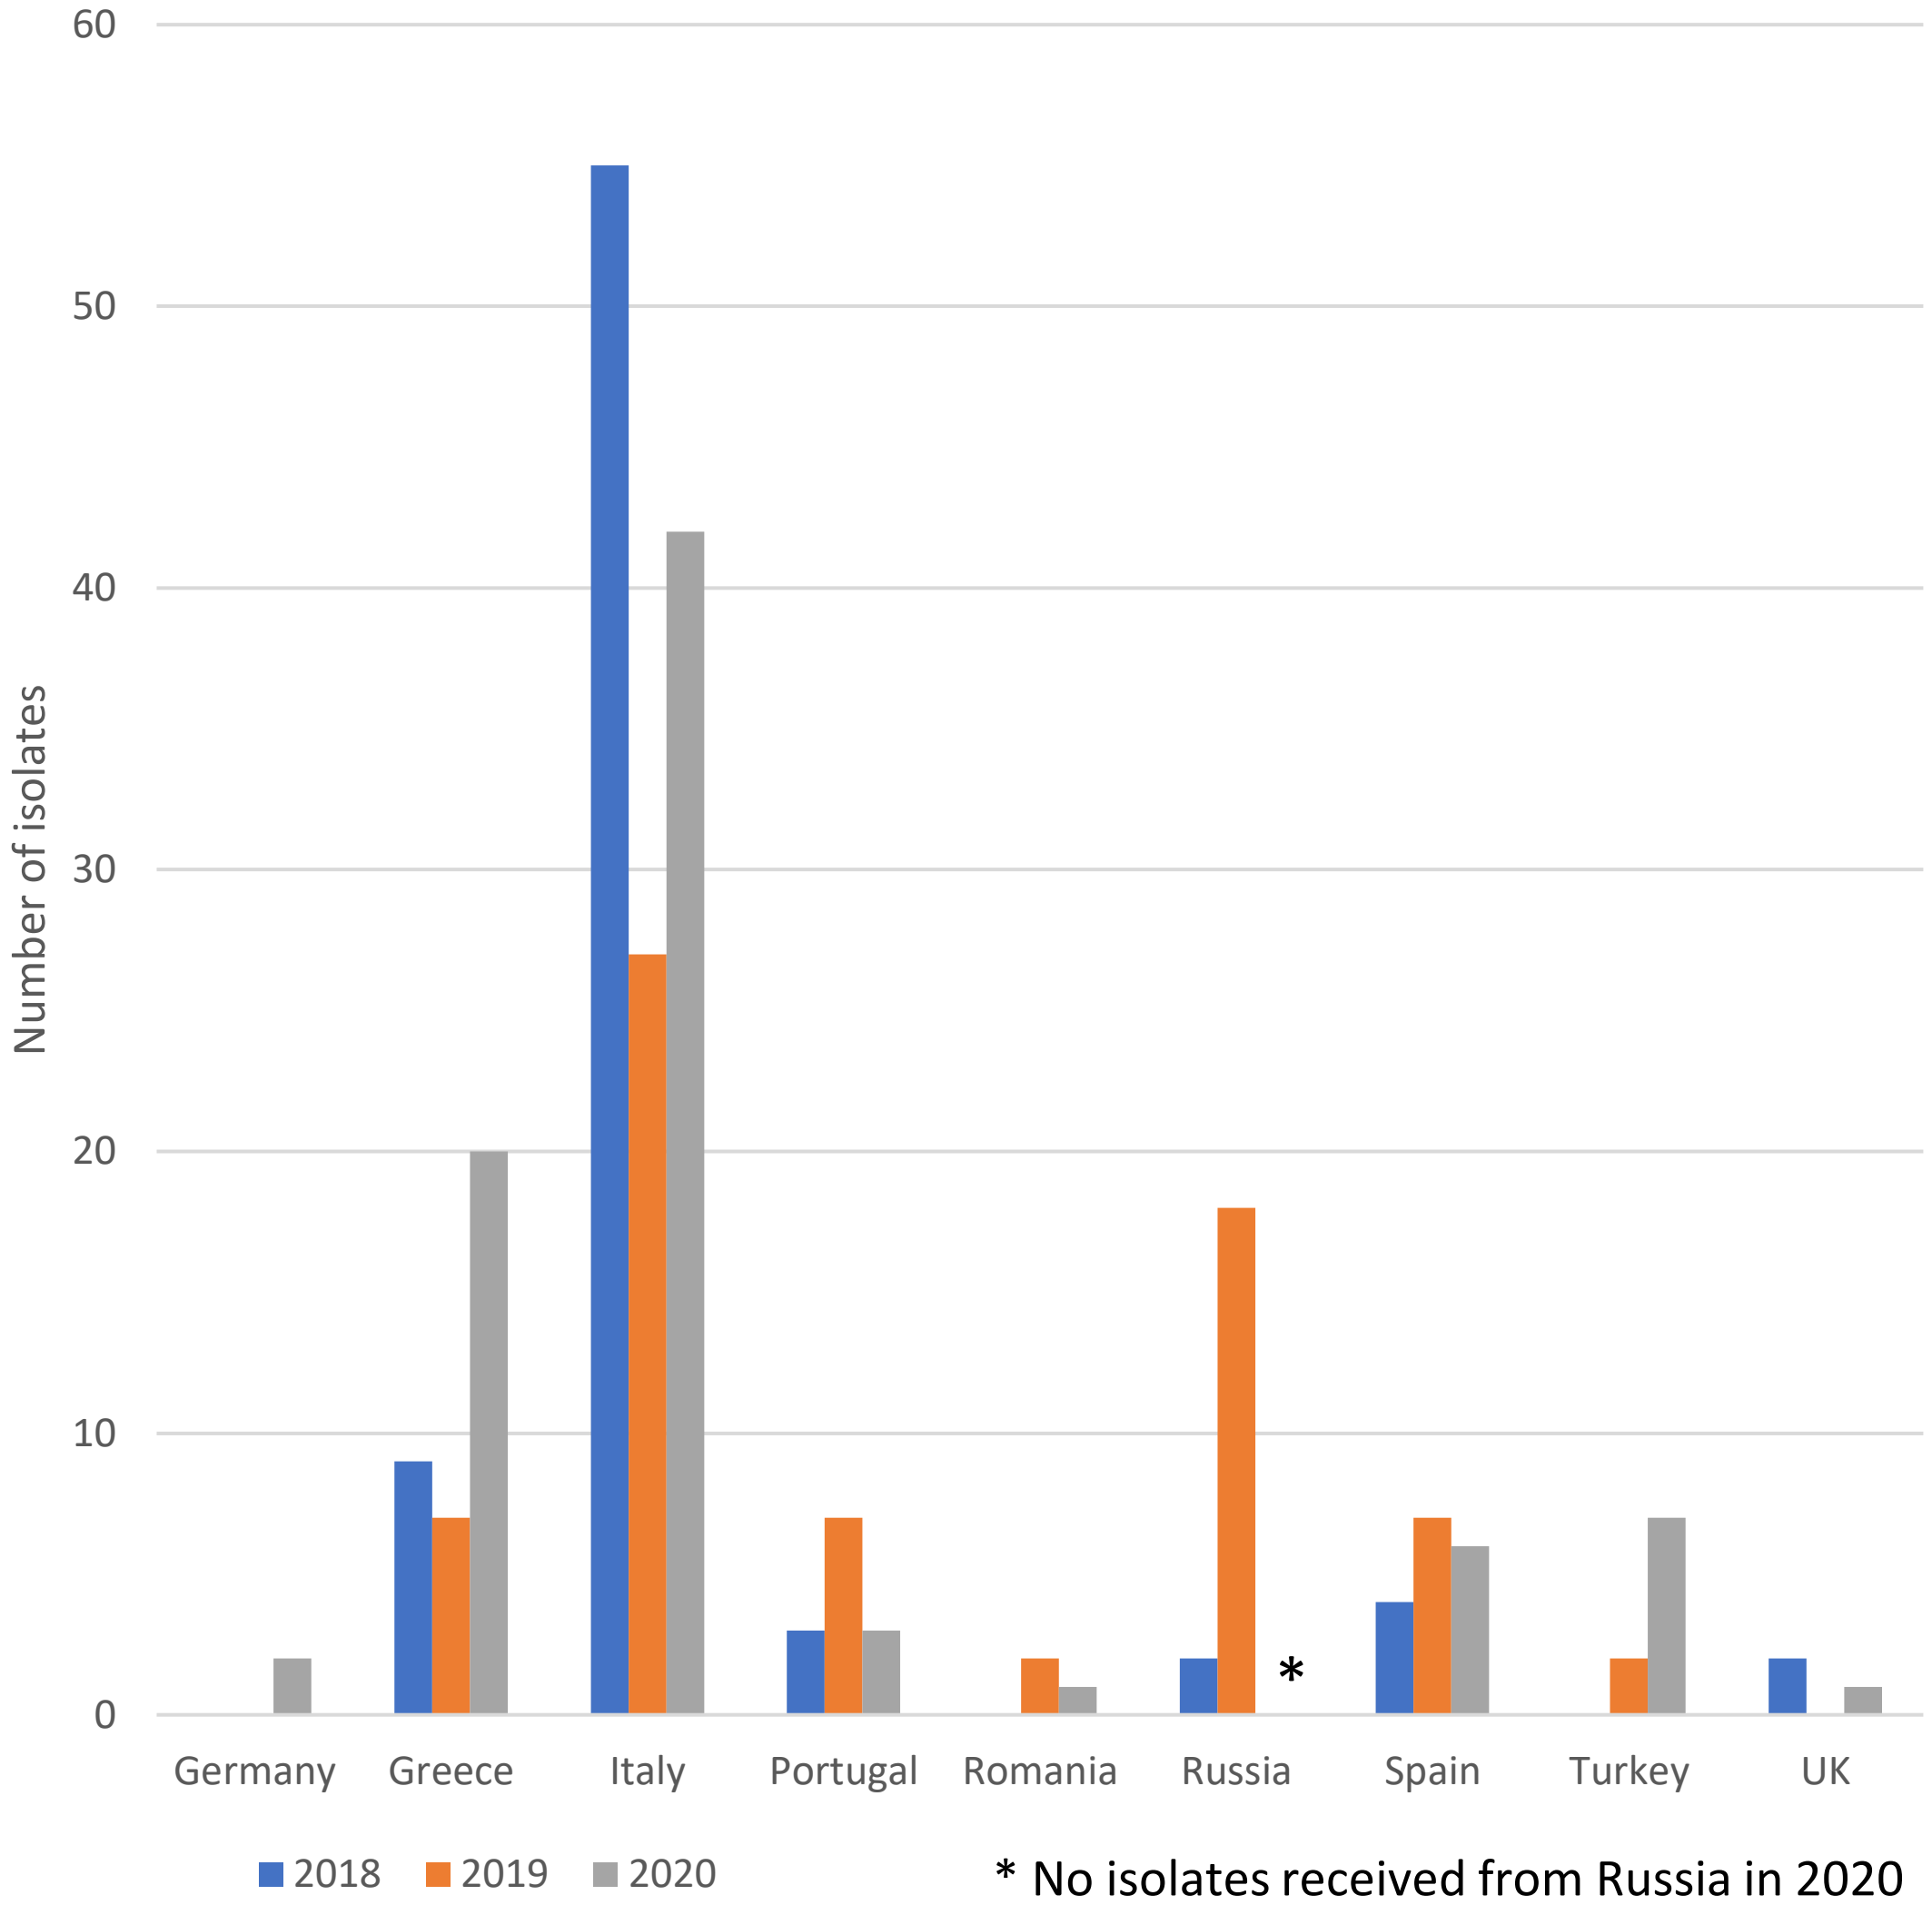
Number of isolates
0
10
20
30
40
50
60
Germany
Greece
Italy
Portugal
Romania
Russia
Spain
Turkey
UK
*
2018
2019
2020

* No isolates received from Russia in 2020

2

Results

Table 1. Activity of Meropenem-vaborbactam and Comparator Antimicrobial Agents Tested Against 227 KPC-producing European Isolates (2018-2020)

Antimicrobial agent	mg/L		EUCAST[a]		
	MIC_{50}	MIC_{90}	%S	%I	%R
Meropenem-vaborbactam	0.25	2	97.4		2.6
Meropenem	>32	>32	3.1 [b] 3.1 [c]	 14.1	96.9 82.8
Imipenem	>8	>8	0.0	3.5	96.5
Amikacin	8	>32	50.2 [d]		49.8
Aztreonam	>16	>16	0.0	0.0	100.0
Cefepime	>32	>32	0.4	0.4	99.1
Ceftazidime	>32	>32	0.0	0.4	99.6
Colistin	0.25	>8	83.7		16.3
Gentamicin	2	>16	53.7 [d]		46.3
Levofloxacin	>32	>32	6.2	0.4	93.4
Piperacillin-tazobactam	>128	>128	0.0		100.0

[a] Criteria as published by EUCAST (2021).
[b] Using meningitis breakpoints.
[c] Using non-meningitis breakpoints.
[d] For infections originating from the urinary tract. For systemic infections, aminoglycosides must be used in combination with other active therapy.
Organisms include *Citrobacter freundii* species complex (1), *Enterobacter cloacae* species complex (2), *Escherichia coli* (6), and *Klebsiella pneumoniae* (218).

Figure 3 MIC distributions of KPC 2 & 3 Producing Isolates

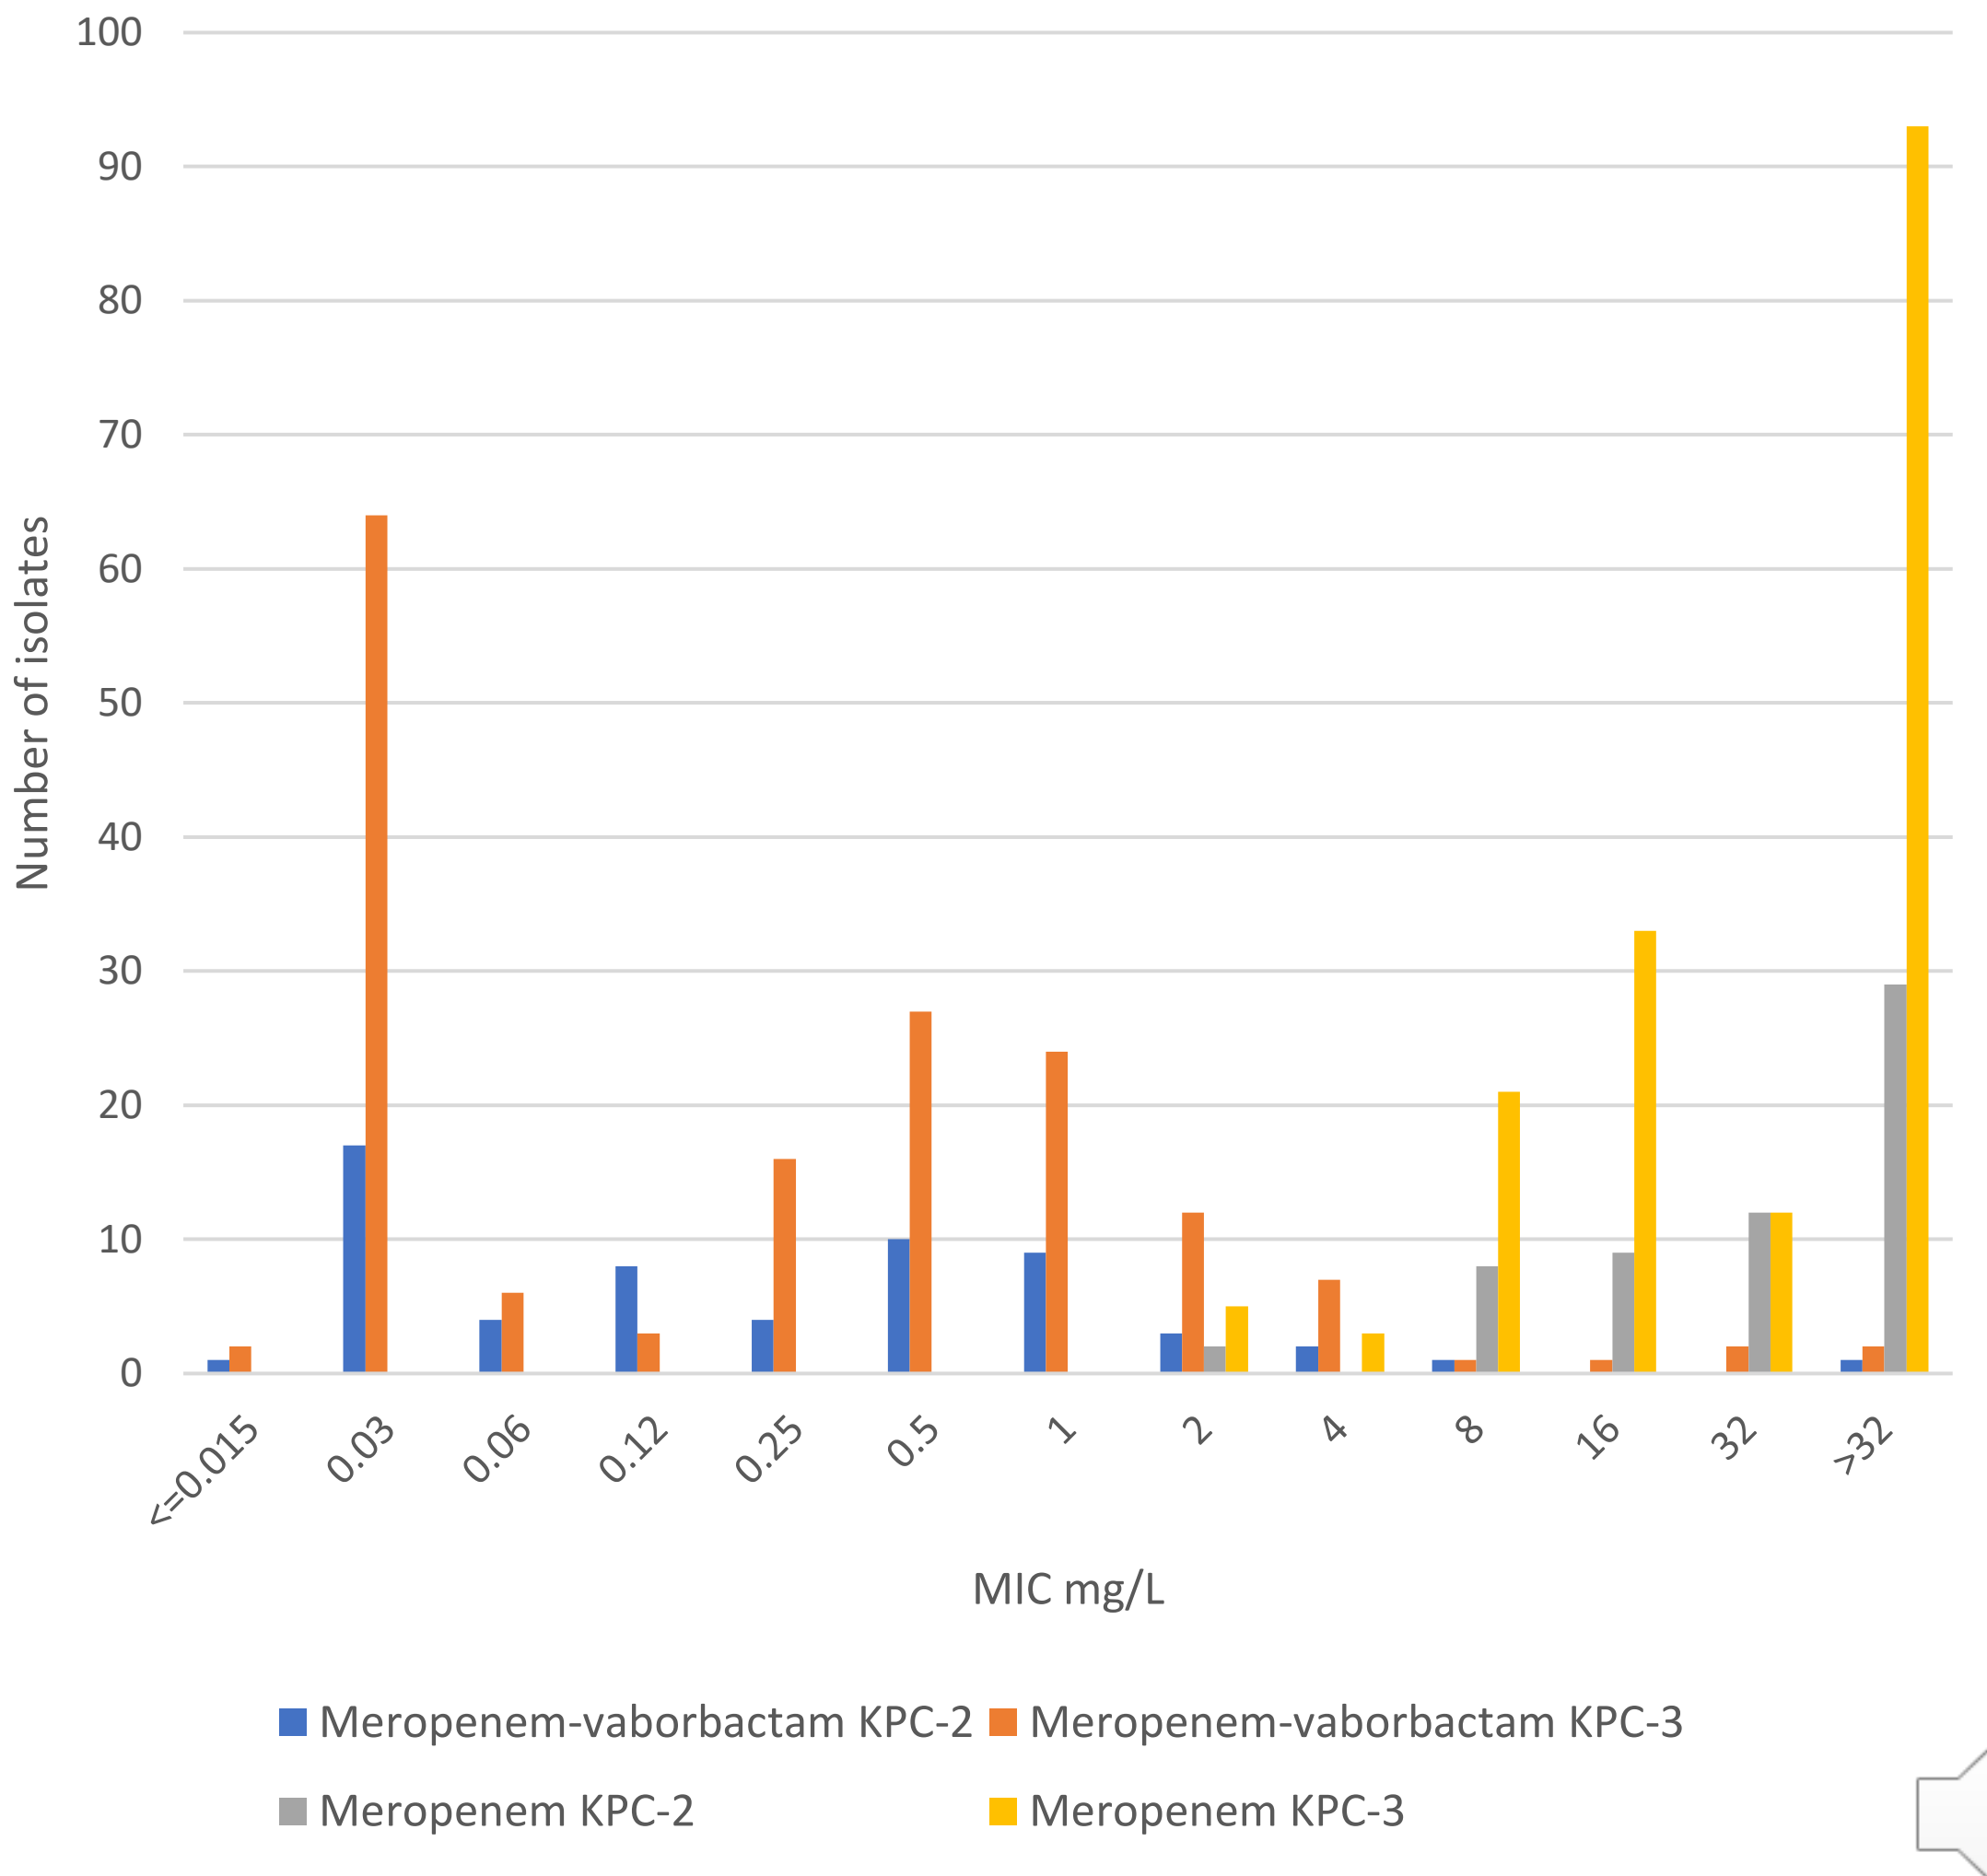

100
90
80
70
60
50
40
30
20
10
0
Number of isolates
<=0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
>32
MIC mg/L
Meropenem-vaborbactam KPC-2
Meropenem-vaborbactam KPC-3
Meropenem KPC-2
Meropenem KPC-3

3

Results

- The most common KPC-producing species were *Klebsiella pneumoniae* (KPN; *n* = 218) and *Escherichia coli* (*n* = 6; Table 1).
 - Bloodstream infections had the most KPC-producing isolates (Figure 1).
- The number of KPC-producing isolates varied by country and year (Figure 2).
- Of the 227 KPC-producing isolates, 167 produced KPC-3 and 60 had KPC-2. MVB activity was similar against both groups (Figure 3).
- The susceptibility to MVB was 97.4%, the highest of the agents tested; meropenem susceptibility was 3.1% (Table 1).
 - 6 MVB-resistant isolates contained both KPC and a metallo-beta lactamase, either VIM-1 or NDM-1.
 - All but 1 isolate also contained other beta-lactamase enzymes, CTX-M-15 (*n* = 71) was the most common extended spectrum beta-lactamase (ESBL).

Conclusions

- KPC prevalence varied by country.
 - Italy had the highest number of KPCs.
 - The number of KPCs declined in Italy but increased in Greece and Turkey from 2018 to 2020.
- MVB had potent *in vitro* activity against CRE isolates producing KPC and had the highest susceptibility of tested agents.
- These in vitro data suggest that MVB is a useful treatment for infections caused by CRE that produce KPC and ESBLs.

Acknowledgements

This poster has been funded by A. Menarini Industrie Farmaceutiche Riunite SRL.

Contact

Dee Shortridge, PhD, JMI Labs
Email: dee-Shortridge@jmilabs.com

4