ECCMID 2023 | Poster #P0240

Comparison of *In Vitro* Activity of Cefiderocol and Comparator Agents Against Molecularly Characterized Carbapenem-resistant Enterobacterales Clinical Isolates Causing Infections in Europe and Surrounding Regions (2020–2021)

D. Shortridge[1], R.E. Mendes[1], J.H. Kimbrough[1], T.B. Doyle[1], M. Castanheira[1]

[1]JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Cefiderocol (CFDC) represents an important addition to the antimicrobial armamentarium due to its broad activity against Gram-negative bacteria.
- CFDC is stable to hydrolysis by most serine β-lactamases (ESBL, KPC, and OXA-type carbapenemases) and metallo-β-lactamases.
- CFDC and comparator activities were analysed against molecularly characterized carbapenem-resistant Enterobacterales isolates (CRE), which were collected as part of the SENTRY Antimicrobial Surveillance Program for Europe and adjacent regions.

Materials and Methods

- A total of 7,739 Enterobacterales were collected from hospitalised patients in 37 medical centres in 16 European countries, Israel, and Turkey during 2020–2021.
- Susceptibility testing was by broth microdilution, comparators were tested using cation-adjusted Mueller Hinton broth (CAMHB). CFDC testing used iron-depleted CAMHB.
- CLSI/EUCAST 2022 breakpoints were used.
- Isolates displaying MIC values ≥4 mg/L for imipenem (excluding *P. mirabilis*, *P. penneri*, and indole-positive *Proteus*) or meropenem (MER) were subjected to whole genome sequencing and analysis for β-lactamase genes.

Results

- A total of 260 (3.4%) CRE were detected, 233 were *Klebsiella pneumoniae*.
- The CRE mostly carried *bla*$_{\text{KPC}}$ (47.7%; 124/260), followed by the MBL genes, *bla*$_{\text{NDM}}$ or *bla*$_{\text{VIM}}$ (21.2%; 55/260), and *bla*$_{\text{OXA-48}}$–like (20.8%; 54/260; Table 1 and Figure 1).
- Twelve isolates (4.6%; 12/260) co-produced an MBL with KPC (n=8), or OXA-48–like (n=4) enzymes.
- CFDC had the highest susceptibility against all CRE (96.5/82.3%, CLSI/EUCAST; $MIC_{50/90}$, 1/4 mg/L; Table 1; Figure 2).
- KPC-producing isolates were susceptible to CFDC, imipenem-relebactam (IMR), meropenem-vaborbactam (MEV), and ceftazidime-avibactam (CZA), with >91.4% susceptible.
- CFDC ($MIC_{50/90}$, 0.5/2 mg/L) and CZA ($MIC_{50/90}$, 1/2 mg/L) were active against isolates carrying *bla*$_{\text{OXA-48}}$–like genes.
- CFDC was the most active agent against isolates carrying MBLs, with or without other carbapenemases, with 32 isolates inhibited at ≤2 mg/L (EUCAST susceptible breakpoint), 49 isolates inhibited at ≤4 mg/L (CLSI susceptible breakpoint), and the remaining six at 8 mg/L.
- CFDC ($MIC_{50/90}$, 1/4 mg/L), IMR ($MIC_{50/90}$, 0.5/2 mg/L), MEV ($MIC_{50/90}$, 2/8 mg/L), and CZA ($MIC_{50/90}$, 2/4 mg/L) showed similar activity against 39 CRE with no detected carbapenemases.

Conclusions

- CFDC showed potent activity against a collection of contemporary CRE isolates, collected from hospitalised patients in 16 European countries, Israel, and Turkey during 2020–2021, which carried a heterogeneous array of carbapenemase genes.
- The activity of CFDC was consistent against various genotypes, where approved β-lactam/β-lactamase inhibitor combinations showed limited activity.
- These data confirm CFDC as an important option for the treatment of infections caused by resistant Enterobacterales, including those producing MBLs.

Acknowledgements

This research and poster presentation were sponsored by Shionogi & Co., LTD.

Table 1. Susceptibilities of CRE isolates with various genotypes to cefiderocol and comparators

Phenotype/genotype (No. isolates)	MIC_{50}/MIC_{90} in mg/L (% susceptible by CLSI/EUCAST criteria)[a]				
	CFDC	IMR	MEV	CZA	MER
CRE[b] (260)	1/4 (96.5/82.3)	0.5/>8 (58.1/62.7)	2/>8 (63.5/67.3)	2/>32 (78.5)	32/>32 (3.1/5.4)
KPC[c] (116)	0.5/2 (99.1/91.4)	0.25/0.5 (99.1/100.0)	0.25/2 (96.6/98.3)	1/4 (100.0)	>32/>32 (0.0/2.6)
OXA-48–like[d] (50)	0.5/2 (100/90.0)	8/>8 (4.0/16.0)	>8/>8 (20.0/22.0)	1/2 (100.0)	32/>32 (8.0/14.0)
MBL[e] (55)	2/8 (89.1/58.2)	>8/>8 (0.0/1.8)	>8/>8 (16.4/21.8)	>32/>32 (0.0)	32/>32 (3.6/3.6)
Non-carbapenemases[f] (39)	1/4 (94.9/79.5)	0.5/2 (87.2/97.4)	2/8 (87.2/97.4)	2/4 (97.4)	8/16 (5.1/5.1)

Abbreviations: CFDC, cefiderocol; IMR, imipenem-relebactam; MEV, meropenem-vaborbactam; CZA, ceftazidime-avibactam; MER, meropenem.
[a] MIC interpreted according to CLSI/EUCAST (2022) criteria.
[b] CRE, defined as isolates with imipenem (excluded for *P. mirabilis*, *P. penneri*, and indole-positive *Proteus*) and/or meropenem MIC values ≥4 mg/L.
[c] Includes 48 isolates carrying *bla*$_{\text{KPC-2}}$ and 68 isolates carrying *bla*$_{\text{KPC-3}}$; 8 isolates co-carrying *bla*$_{\text{NDM-1}}$ or *bla*$_{\text{VIM-1}}$ were excluded.
[d] Includes 22 isolates carrying *bla*$_{\text{OXA-48}}$, 25 carrying *bla*$_{\text{OXA-232}}$, and 3 carrying *bla*$_{\text{OXA-181}}$; 4 isolates co-carrying *bla*$_{\text{OXA-48}}$ and *bla*$_{\text{NDM-1}}$ or *bla*$_{\text{VIM-1}}$ were excluded.
[e] Includes 37 isolates carrying *bla*$_{\text{NDM-1}}$ (including 3 co-carrying *bla*$_{\text{OXA-48}}$ and 1 co-carrying *bla*$_{\text{KPC-2}}$), 2 isolates carrying *bla*$_{\text{NDM-5}}$, 16 isolates carrying *bla*$_{\text{VIM-1}}$ (including 7 isolates co-carrying *bla*$_{\text{KPC-2, or 3}}$ and 1 co-carrying *bla*$_{\text{OXA-48}}$).
[f] Includes CRE with no known carbapenemase genes detected.

Figure 1. Carbapenemases encountered amongst 7,739 Enterobacterales collected from hospitalised patients in 37 medical centres in 16 European countries, Israel, and Turkey during 2020–2021

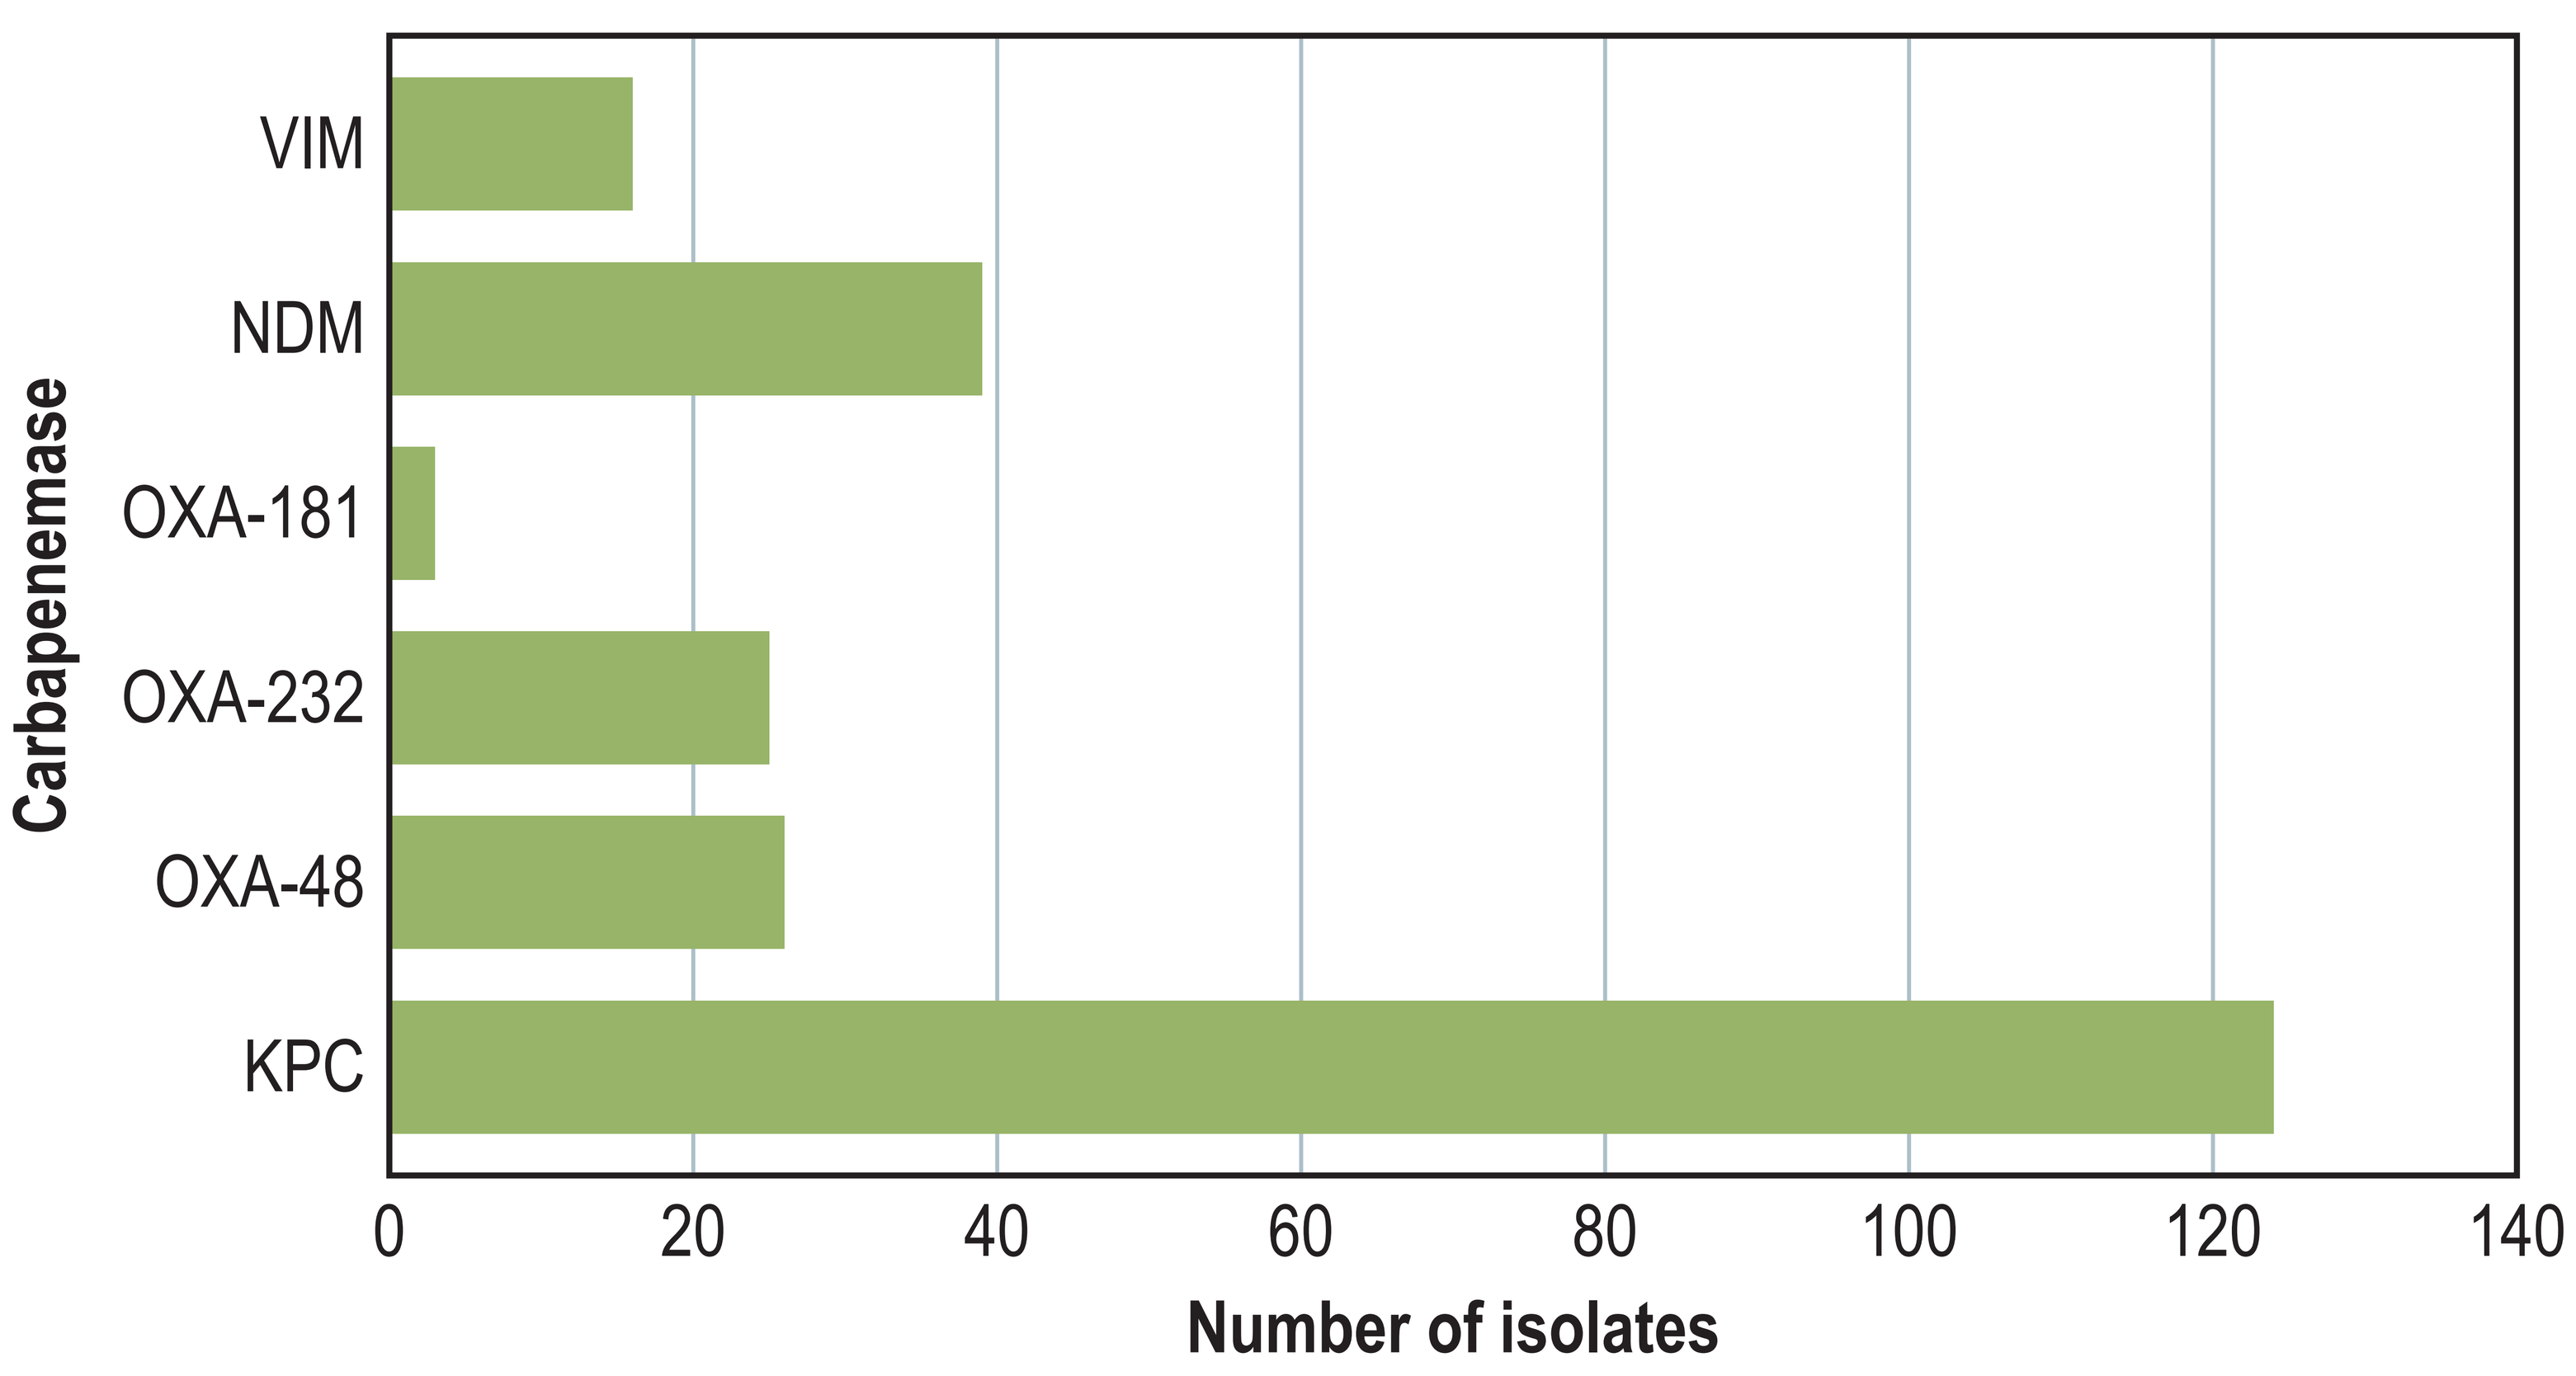

VIM
NDM
OXA-181
OXA-232
OXA-48
KPC
Carbapenemase
0
20
40
60
80
100
120
140
Number of isolates

Figure 2. CRE MIC distributions to cefiderocol and comparators (n=260)

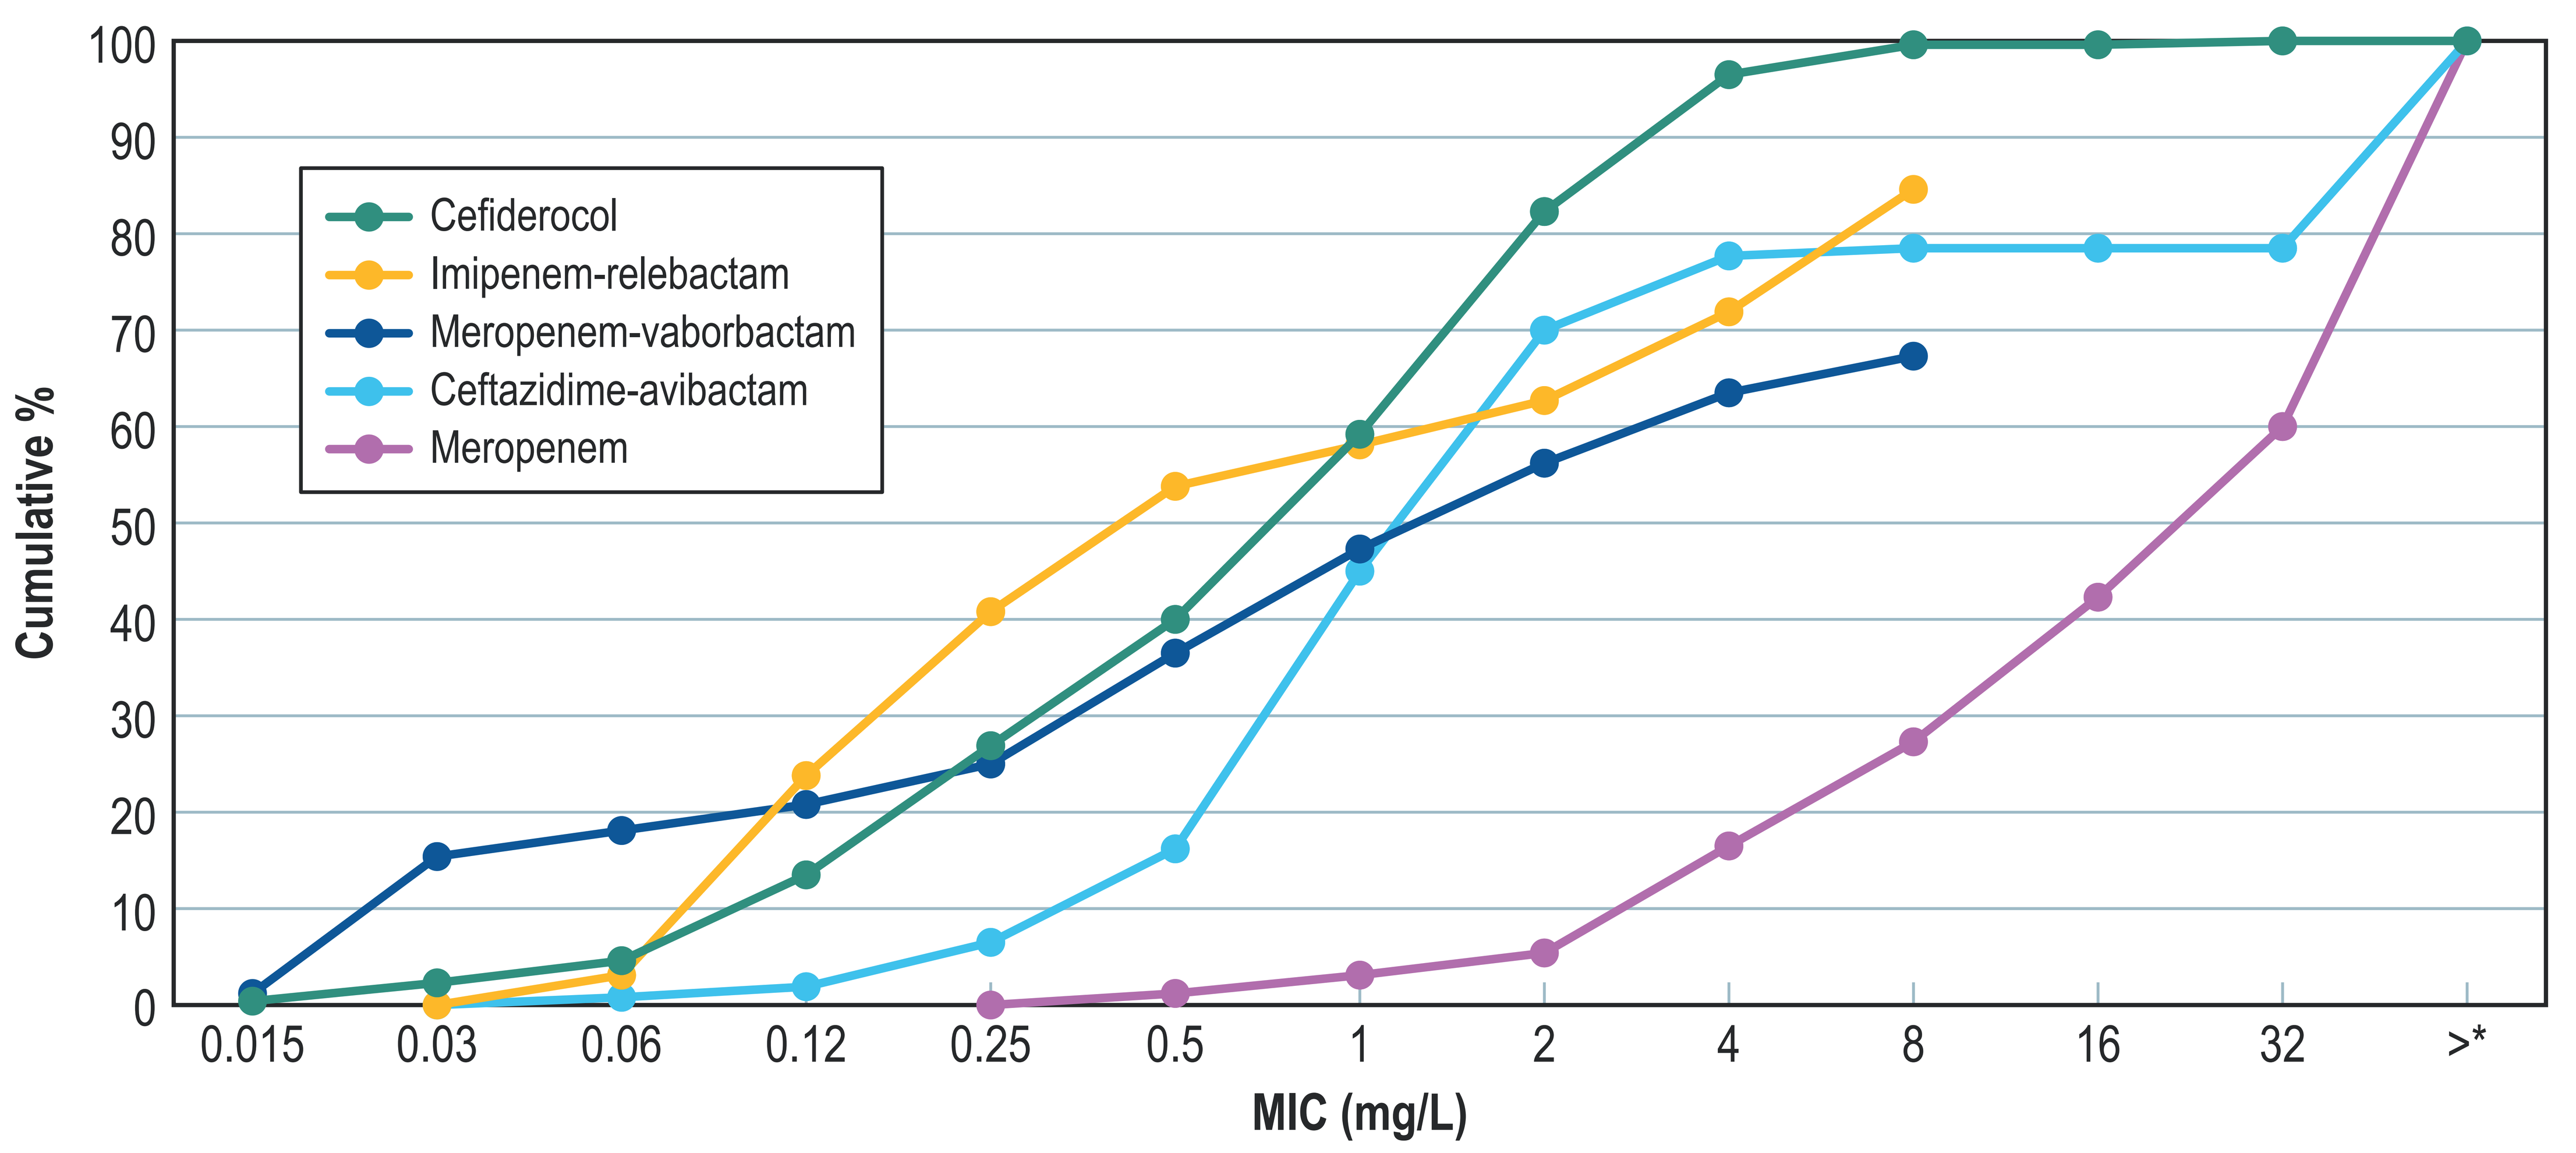

Cefiderocol
Imipenem-relebactam
Meropenem-vaborbactam
Ceftazidime-avibactam
Meropenem
Cumulative %
0
10
20
30
40
50
60
70
80
90
100
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
>*
MIC (mg/L)

* Greater than highest dilution tested

References

1. CLSI. M07Ed11. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition. Wayne, PA, Clinical and Laboratory Standards Institute, 2018.
2. CLSI. M100Ed32. Performance standards for antimicrobial susceptibility testing: 29th informational supplement. Wayne, PA, Clinical and Laboratory Standards Institute, 2022.
3. European Committee on Antimicrobial Susceptibility Testing. Breakpoint tables for interpretation of MICs and zone diameters Version 12.0, 2022.

Contact

Dee Shortridge
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ECCMID2023_EU_CRE.pdf

ECCMID 2023, April 15–18, 2023, Copenhagen, Denmark