ECCMID 2023 | Poster #P0278

Activity of Cefiderocol and Comparator Agents against US Isolates of *Pseudomonas aeruginosa*, *Acinetobacter baumannii-calcoaceticus* species complex, and *Stenotrophomonas maltophilia*, Including Carbapenem-Resistant Isolates from the SENTRY Antimicrobial Surveillance Program (2020–2022)

Dee Shortridge, Rodrigo Mendes, Mariana Castanheira
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Cefiderocol is a siderophore-conjugated cephalosporin with broad activity against Gram-negative bacteria, including multidrug-resistant organisms.
- Cefiderocol was approved by the EMA for the treatment of infections caused by Gram-negative bacteria in adult patients with limited treatment options and the US FDA for complicated urinary tract infection, hospital-acquired bacterial pneumonia, and ventilator-associated bacterial pneumonia.
- Non–glucose-fermenting species including *Pseudomonas aeruginosa*, *Acinetobacter baumannii-calcoaceticus* complex, and *Stenotrophomonas maltophilia* are often extensively drug-resistant (XDR), presenting serious treatment challenges.
- The susceptibility of cefiderocol and comparator agents was investigated against non–glucose-fermenting US isolates collected in 2020–2022 as part of the SENTRY Antimicrobial Surveillance Program.

Materials and Methods

- A total of 2,982 *P. aeruginosa*, 799 *A. baumannii-calcoaceticus* species complex, and 585 *S. maltophilia* were isolated from hospitalised patients in 63 US medical centres.
- Susceptibility testing was performed using the broth microdilution method with cation-adjusted Mueller-Hinton broth (CAMHB) for comparator agents and iron-depleted CAMHB for cefiderocol.
- CLSI, FDA, and EUCAST breakpoints were applied.
- For the EUCAST assessment of cefiderocol activity against *A. baumannii-calcoaceticus* species complex and *S. maltophilia*, PK/PD (non–species-related) breakpoints were used.
- XDR was defined as non-susceptible to at least 1 agent in all but 2 or fewer drug classes using CLSI criteria.
- Other agents tested included meropenem, levofloxacin, trimethoprim-sulfamethoxazole, and the β-lactam/β-lactamase inhibitor (BL/BLI) combinations ampicillin-sulbactam, ceftazidime-avibactam, ceftolozane-tazobactam, imipenem-relebactam, and meropenem-vaborbactam.

Results

- The most common infection type from which isolates were collected was pneumonia (*n*=2,340), followed by skin and skin structure (*n*=827), bloodstream infection (*n*=543), urinary tract infection (*n*=391), intra-abdominal infection (*n*=142), and other sites (*n*=123).
- *P. aeruginosa* susceptibilities to cefiderocol and BL/BLI combinations were >96.0%, except for meropenem-vaborbactam (90.8%; Table 1).
- Cefiderocol was the most active agent against XDR *P. aeruginosa* isolates (susceptibility 98.2/97.1/93.0% CLSI/EUCAST/FDA, respectively). The susceptibilities of the BL/BLI combinations against these XDR isolates ranged from 42.1% to 78.8%.
- Cefiderocol had higher susceptibilities than comparator agents against BL/BLI-resistant *P. aeruginosa* isolates (Table 1, Figure 1).
- *A. baumannii-calcoaceticus* complex susceptibility to cefiderocol was 98.5/97.2/93.6% (CLSI/EUCAST/FDA; Table 2, Figure 1).
- Cefiderocol retained good activity against XDR, meropenem-resistant, or imipenem-relebactam–resistant *A. baumannii-calcoaceticus* complex isolates, with ≥85.0% susceptibility.
- Cefiderocol was active against *S. maltophilia* (98.5/99.3% susceptible, CLSI/EUCAST; Table 2, Figure 1).

Table 1. Susceptibilities of *Pseudomonas aeruginosa* isolates and resistant subgroups, collected in the SENTRY (2020–2022) study, for cefiderocol and comparator agents

Organism/organism group Antimicrobial agent	MIC_{50} (mg/L)	MIC_{90} (mg/L)	%S CLSI[a]	%S EUCAST[a]	%S US FDA[a]
All (*n*=2,982)					
Cefiderocol	0.12	0.25	99.8	99.6	98.5
Meropenem	0.5	8	80.4	80.4	80.4
Meropenem-vaborbactam	0.5	8		90.8	
Imipenem-relebactam	0.25	1	97.3	97.3	97.3
Ceftolozane-tazobactam	0.5	2	97.4	97.4	97.4
Ceftazidime-avibactam	2	8	96.4	96.4	96.4
XDR[b] (*n*=273)					
Cefiderocol	0.12	1	98.2	97.1	93.0
Meropenem	16	32	7.3	7.3	7.3
Meropenem-vaborbactam	>8	>8		42.1	
Imipenem-relebactam	2	4	77.7	77.7	77.7
Ceftolozane-tazobactam	2	16	78.8	78.8	78.8
Ceftazidime-avibactam	8	32	69.6	69.6	69.6
Meropenem MIC >4 mg/L (*n*=418)					
Cefiderocol	0.12	0.5	99.3	98.6	95.7
Meropenem	16	32	0.0	0.0	0.0
Meropenem-vaborbactam	>8	>8		35.2	
Imipenem-relebactam	2	4	81.1	81.1	81.1
Ceftolozane-tazobactam	2	8	85.6	85.6	85.6
Ceftazidime-avibactam	4	16	79.9	79.9	79.9
Meropenem-vaborbactam MIC >8 mg/L (*n*=273)					
Cefiderocol	0.12	0.5	98.9	98.2	94.5
Meropenem	16	32	0.0	0.0	0.0
Meropenem-vaborbactam	>8	>8		0.0	
Imipenem-relebactam	2	8	74.0	74.0	74.0
Ceftolozane-tazobactam	2	16	82.1	82.1	82.1
Ceftazidime-avibactam	8	32	72.2	72.2	72.2
Imipenem-relebactam MIC >4 mg/L (*n*=30)					
Cefiderocol	0.25	1	100.0	100.0	90.0
Meropenem	32	>32	0.0	0.0	0.0
Meropenem-vaborbactam	>8	>8		3.3	
Imipenem-relebactam	8	>8	0.0	0.0	0.0
Ceftolozane-tazobactam	8	>16	46.7	46.7	46.7
Ceftazidime-avibactam	16	>32	33.3	33.3	33.3
Ceftolozane-tazobactam MIC >8 mg/L (*n*=43)					
Cefiderocol	0.5	8	88.4	83.7	69.8
Meropenem	16	>32	7.0	7.0	7.0
Meropenem-vaborbactam	>8	>8		30.2	
Imipenem-relebactam	2	>8	55.8	55.8	55.8
Ceftolozane-tazobactam	>16	>16	0.0	0.0	0.0
Ceftazidime-avibactam	>32	>32	16.3	16.3	16.3
Ceftazidime-avibactam MIC >8 mg/L (*n*=106)					
Cefiderocol	0.25	2	95.3	92.5	87.7
Meropenem	16	>32	10.4	10.4	10.1
Meropenem-vaborbactam	>8	>8		28.3	
Imipenem-relebactam	2	8	66.0	66.0	66.0
Ceftolozane-tazobactam	4	>16	54.7	54.7	54.7
Ceftazidime-avibactam	16	>32	0.0	0.0	0.0

[a] Breakpoints as published by CLSI, EUCAST, and US FDA (2022).
[b] XDR, extensively drug-resistant, defined as non-susceptible to all but 2 or fewer drug classes using CLSI breakpoints (2022).

Table 2. Susceptibilities of *Acinetobacter baumannii-calcoaceticus* species isolates, including resistant subgroups, and *Stenotrophomonas maltophilia* collected in the SENTRY (2020–2022) study for cefiderocol and comparator agents

Organism/organism group Antimicrobial agent	MIC_{50} (mg/L)	MIC_{90} (mg/L)	%S CLSI[a]	%S EUCAST[a]	%S US FDA[a]
All *A. baumannii-calcoaceticus* complex (*n*=799)					
Cefiderocol	0.12	1	98.5	97.2	93.6
Meropenem	0.5	>32	68.3	68.3	68.3
Imipenem-relebactam	0.25	>8		69.6	69.6
Ampicillin-sulbactam	4	64	67.1		67.1
Minocycline	0.12	8	84.9		84.9
Colistin	0.5	1		96.0	
XDR[b] (*n*=192)					
Cefiderocol	0.5	2	96.4	92.7	85.4
Meropenem	>32	>32	3.1	3.1	3.1
Imipenem-relebactam	>8	>8		4.7	4.7
Ampicillin-sulbactam	32	>64	4.2		4.2
Minocycline	8	16	42.7		42.7
Colistin	0.5	2		92.7	
Meropenem MIC >4 mg/L (*n*=247)					
Cefiderocol	0.5	2	96.0	92.7	85.0
Meropenem	>32	>32	0.0	0.0	0.0
Imipenem-relebactam	>8	>8		1.6	1.6
Ampicillin-sulbactam	32	>64	10.1		10.1
Minocycline	4	16	57.9		57.9
Colistin	0.5	1		94.3	
Imipenem-relebactam MIC >2 mg/L (*n*=243)					
Cefiderocol	0.5	2	95.9	93.0	85.2
Meropenem	>32	>32	0.0	0.0	0.0
Imipenem-relebactam	>8	>8		0.0	0.0
Ampicillin-sulbactam	32	>64	9.5		9.5
Minocycline	4	16	58.4		58.4
Colistin	0.5	1		94.2	
S. maltophilia (*n*=585)					
Cefiderocol	0.06	0.25	98.5	99.3	
Levofloxacin	1	8	82.4		
Trimethoprim-sulfamethoxazole	≤0.12	0.5	97.8	98.5[c]	
Minocycline	0.5	1	99.8		

[a] Breakpoints as published by CLSI, EUCAST, and US FDA (2022).
[b] XDR, extensively drug-resistant, defined as non-susceptible to all but 2 or fewer drug classes using CLSI breakpoints (2022).
[c] Susceptible-increased exposure shown (EUCAST, 2022).

Figure 1. Cefiderocol MIC distributions of *P. aeruginosa*, all isolates and those resistant to meropenem or ceftazidime-avibactam; *A. baumannii-calcoaceticus* complex, all isolates and those resistant to meropenem; *S. maltophilia*, all isolates

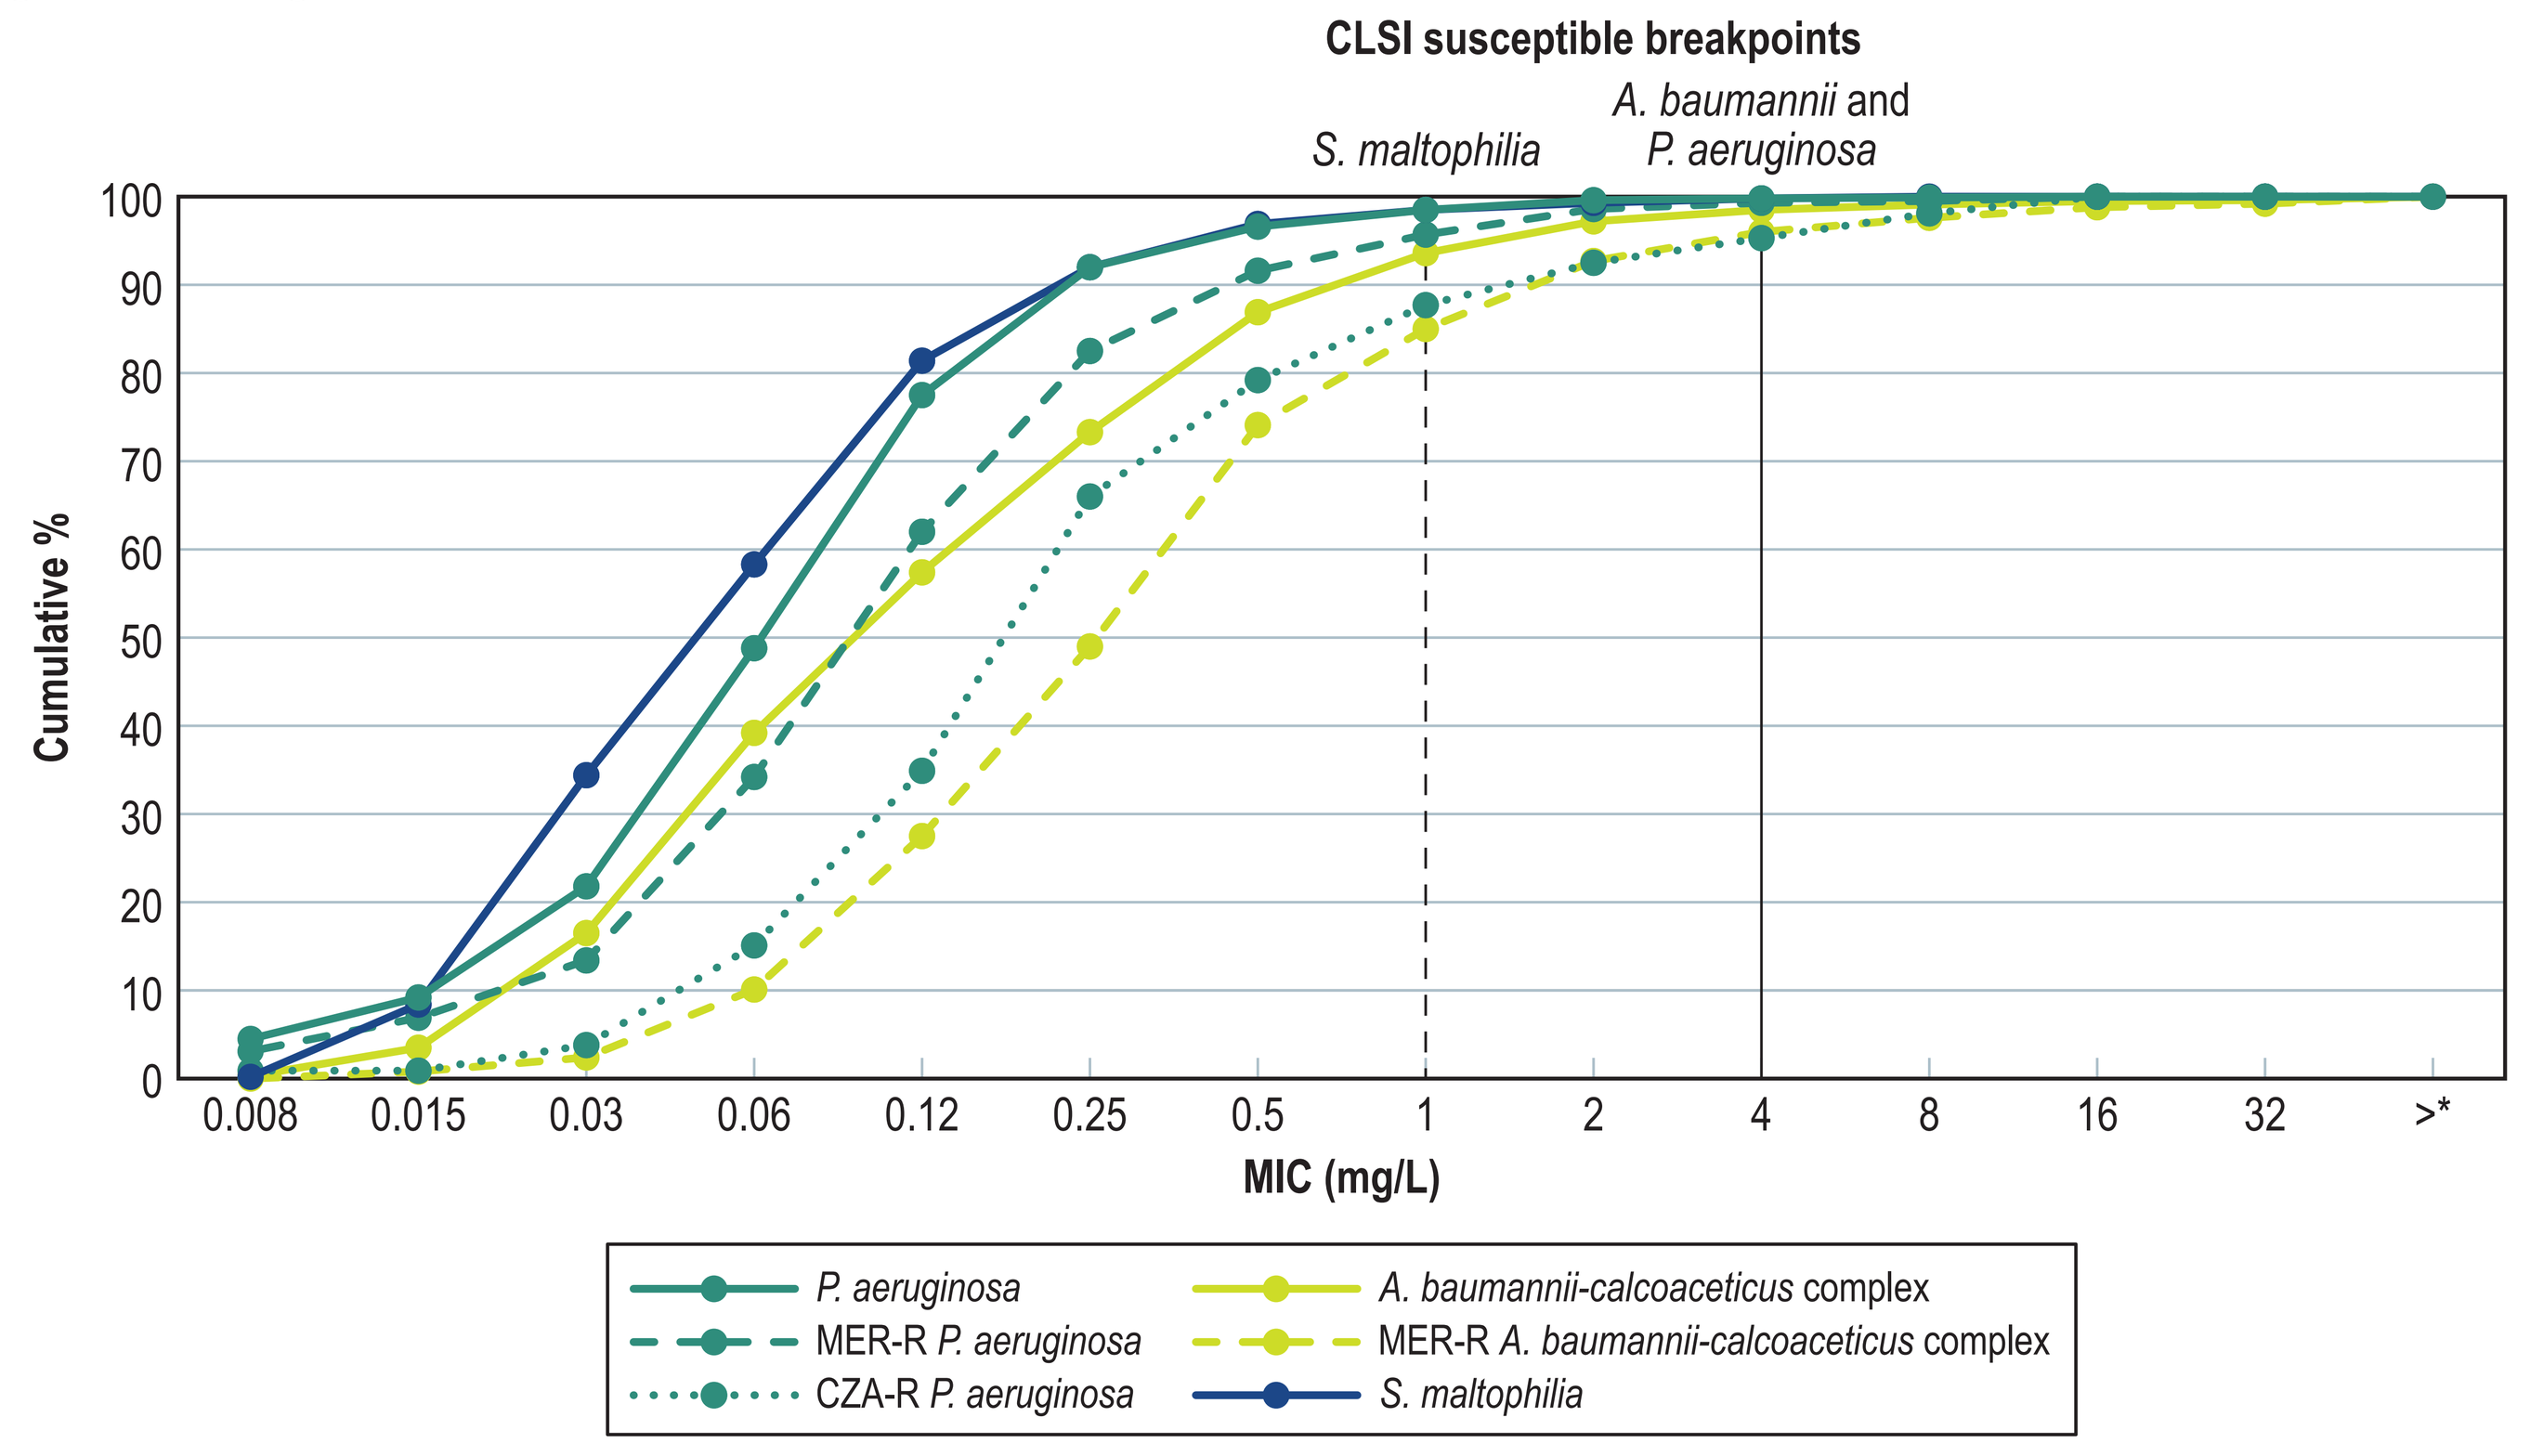
CLSI susceptible breakpoints
S. maltophilia
A. baumannii and P. aeruginosa
Cumulative %
0
10
20
30
40
50
60
70
80
90
100
0.008
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
>*
MIC (mg/L)
P. aeruginosa
MER-R P. aeruginosa
CZA-R P. aeruginosa
A. baumannii-calcoaceticus complex
MER-R A. baumannii-calcoaceticus complex
S. maltophilia

*Greater than the highest concentration tested.
Abbreviations: MER-R, meropenem-resistant; CZA-R, ceftazidime-avibactam–resistant.

Conclusions

- Cefiderocol was the most active β-lactam with broad activity against contemporary US isolates of drug-resistant subsets of *P. aeruginosa* and *A. baumannii-calcoaceticus* complex as well as *S. maltophilia*, where treatment options are limited.
- These *in vitro* data suggest that cefiderocol is an important treatment option for infections caused by non–glucose-fermenting pathogens, including meropenem-, BL/BLI-resistant, and XDR isolates.

Acknowledgments

This research and poster presentation were sponsored by Shionogi & Co., LTD.

References

1. CLSI. M07Ed11. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard. Wayne, PA, Clinical and Laboratory Standards Institute, 2018.
2. CLSI. M100Ed32. Performance standards for antimicrobial susceptibility testing. Wayne, PA, Clinical and Laboratory Standards Institute, 2022.
3. US FDA. Antibacterial Susceptibility Test Interpretive Criteria, 2022. https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria.
4. EUCAST. Breakpoint tables for interpretation of MICs and zone diameters. Version 12.0. Växjö, Sweden, European Committee on Antimicrobial Susceptibility Testing, 2022.

Contact

Dee Shortridge
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ECCMID2023_US_Cefiderocol.pdf

ECCMID 2023, April 15–18, 2023, Copenhagen, Denmark