IDWeek 2022 | Poster #P124

Activity of Novel β-Lactam/β-Lactamase Inhibitor Combinations Against AmpC-Producing Species Collected in United States Hospitals

Mariana Castanheira, Jennifer M. Streit, Cecilia G. Carvalhaes, Rodrigo E. Mendes, Dee Shortridge
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Inducible AmpC resistance is caused by the derepression of the chromosomal AmpC in the presence of a β-lactam and limits the use of these agents to treat infections caused by Enterobacterales species known to produce these enzymes (AmpC producers).
- Novel β-lactam/β-lactamase inhibitors (BL/BLIs), such as meropenem-vaborbactam, ceftazidime-avibactam, and imipenem-relebactam, display activity against isolates producing serine-carbapenemases, extended-spectrum β-lactamases, and AmpC enzymes.
- In this study, we evaluated the activity of novel BL/BLIs against a collection of AmpC producers collected in US hospitals during 2021.

Materials and Methods

- A total of 1,252 organisms of Enterobacterales species known to overexpress AmpC enzymes were consecutively collected in 31 US hospitals during 2021.
 - AmpC-producing species included in this study are displayed in Figure 1.
 - Isolate frequency by infection source is displayed in Figure 2.
 - Only 1 isolate per patient episode was included.
- Isolates were susceptibility tested against meropenem-vaborbactam, ceftazidime-avibactam, and comparator agents using the reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI) M07 (2018) document.
 - Vaborbactam was tested at a fixed concentration of 8 mg/L.
 - Avibactam and relebactam were tested at a fixed concentration of 4 mg/L.
- Quality control (QC) was performed according to the CLSI M100 (2022) criteria.
 - All QC MIC results were within acceptable ranges.
- Categorical interpretations for all comparator agents were those criteria found in the CLSI M100 (2022), or the US Food and Drug Administration (FDA) website.

Results

- Meropenem-vaborbactam ($MIC_{50/90}$, 0.03/0.06 mg/L; Figure 3) and amikacin were the most active agents tested against isolates belonging to AmpC producing species, inhibiting 99.8% (Figure 4).
- Ceftazidime-avibactam ($MIC_{50/90}$, 0.12/0.5 mg/L) inhibited 99.5% and imipenem-relebactam ($MIC_{50/90}$, 0.12/1 mg/L) inhibited 95.9% of all AmpC producers (Figures 3 and 4).
- Cefepime and meropenem, the recommended agents to treat infections caused by AmpC-producing species, were active against 92.0% and 97.6% of these isolates, respectively (Figure 4).
- Piperacillin-tazobactam and ceftolozane-tazobactam ($MIC_{50/90}$, 0.5/8 mg/L; Figure 3) displayed activity against 76.4% and 83.6% of the AmpC producers, respectively (Figure 4).
- Tigecycline was active against 96.8% of the isolates; only 53.8% of the isolates had a colistin MIC of ≤2 mg/L (Figure 4).
- A total of 39 (3.1%) AmpC producers were nonsusceptible to imipenem and/or meropenem (Figure 4) (carbapenem non-susceptible Enterobacterales [CNSE]).
 - Meropenem-vaborbactam ($MIC_{50/90}$, 0.25/2 mg/L; data not shown) was active against 92.3% of the CNSE AmpC producers and displayed higher potency than other agents (Figure 3).
 - Imipenem-relebactam ($MIC_{50/90}$, 0.25/2 mg/L) and ceftazidime-avibactam ($MIC_{50/90}$, 1/>32 mg/L) were active against 89.7% of the CNSE AmpC producers.
- Against cefepime-resistant AmpC producers (*n*=45; 3.6%), meropenem-vaborbactam, imipenem-relebactam, and ceftazidime-avibactam exhibited 93.3%, 88.9%, and 86.7% activity when current CLSI breakpoints were applied.

Figure 1. AmpC-producing bacterial species collected from US hospitals in 2021

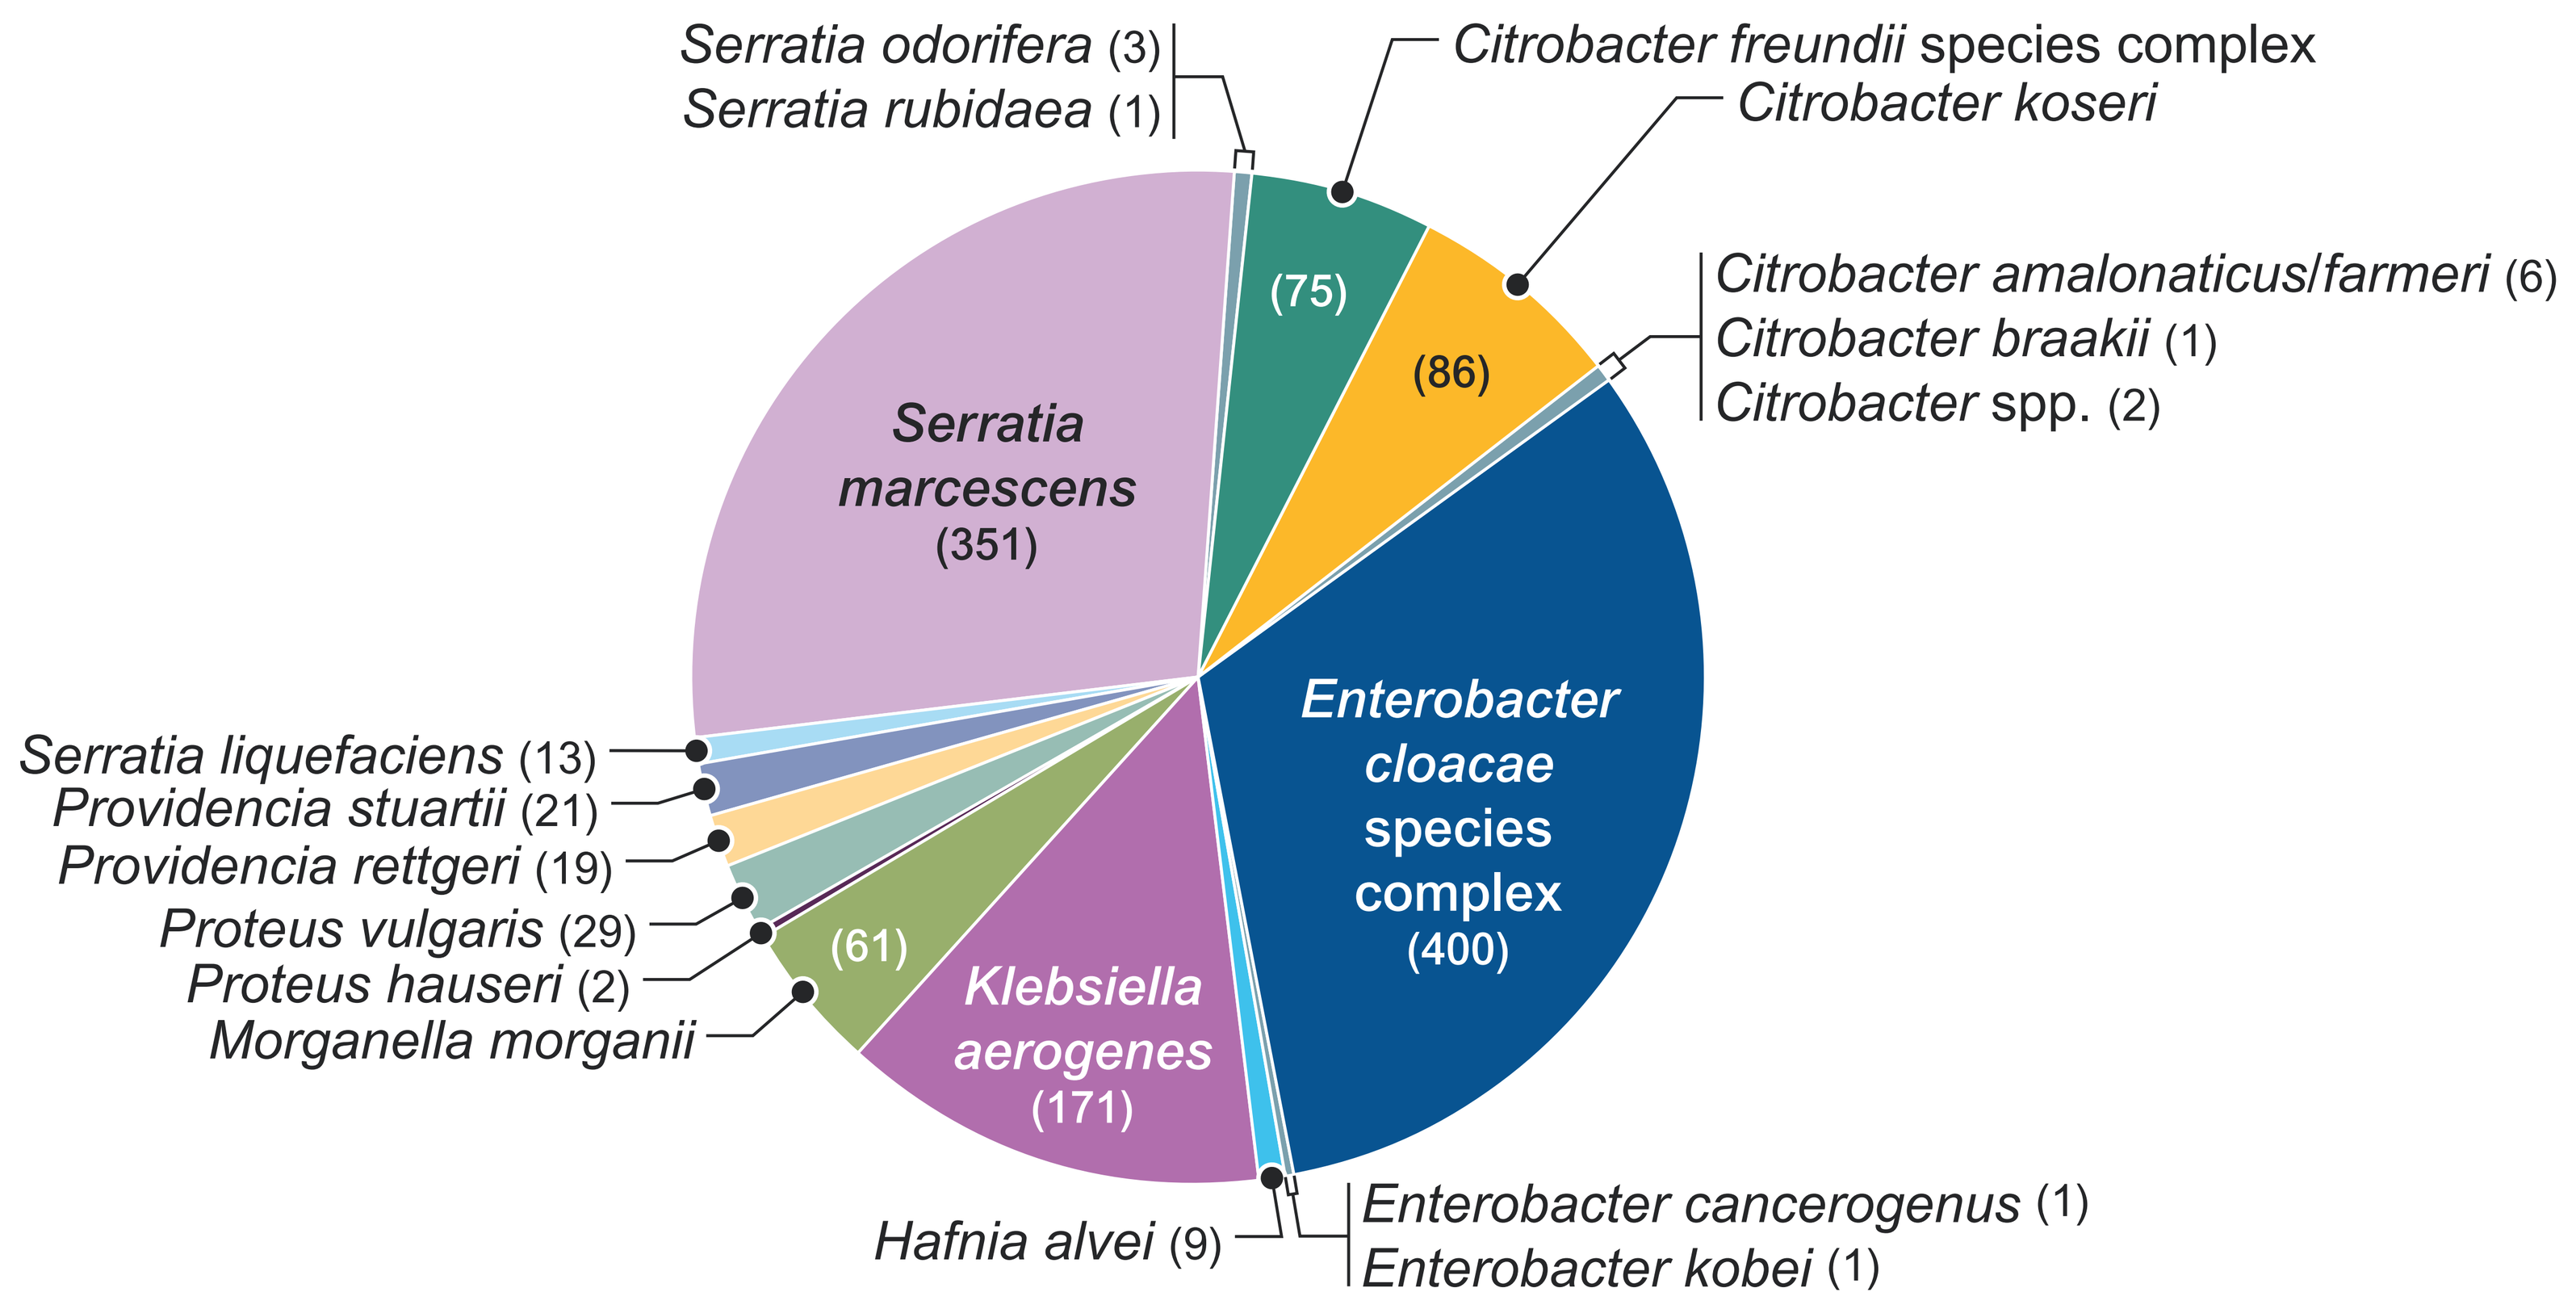
Serratia odorifera (3)
Serratia rubidaea (1)
Citrobacter freundii species complex
Citrobacter koseri
Citrobacter amalonaticus/farmeri (6)
Citrobacter braakii (1)
Citrobacter spp. (2)
Serratia marcescens (351)
(75)
(86)
Enterobacter cloacae species complex (400)
Serratia liquefaciens (13)
Providencia stuartii (21)
Providencia rettgeri (19)
Proteus vulgaris (29)
Proteus hauseri (2)
Morganella morganii
(61)
Klebsiella aerogenes (171)
Hafnia alvei (9)
Enterobacter cancerogenus (1)
Enterobacter kobei (1)

Figure 2. AmpC-producing isolates by infection site

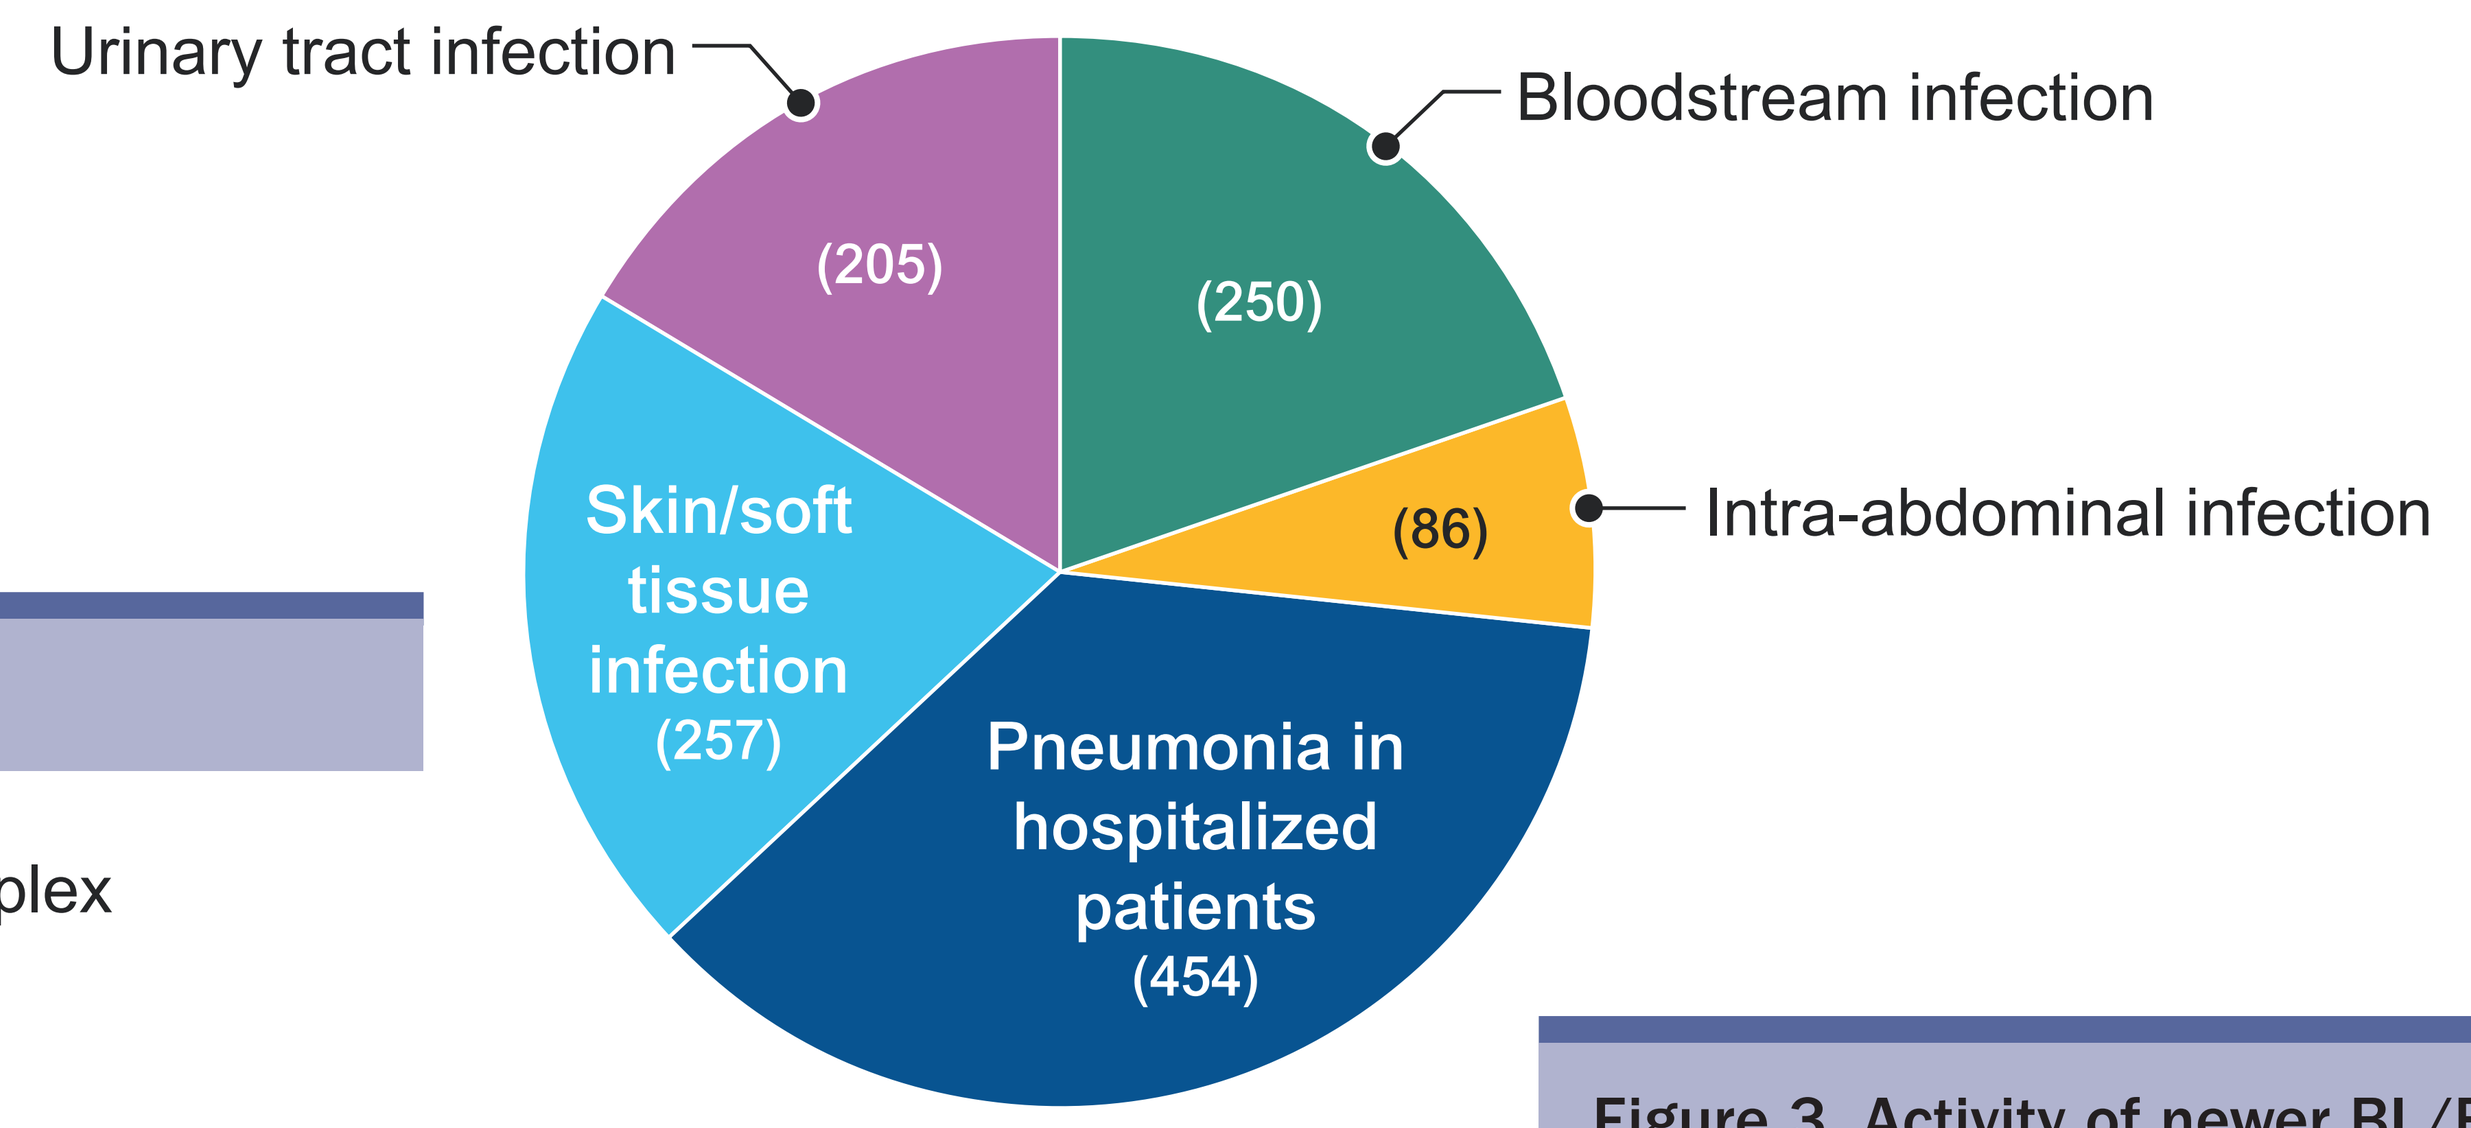
Urinary tract infection
(205)
Bloodstream infection
(250)
Intra-abdominal infection
(86)
Skin/soft tissue infection (257)
Pneumonia in hospitalized patients (454)

Figure 3. Activity of newer BL/BLI combinations against AmpC producers

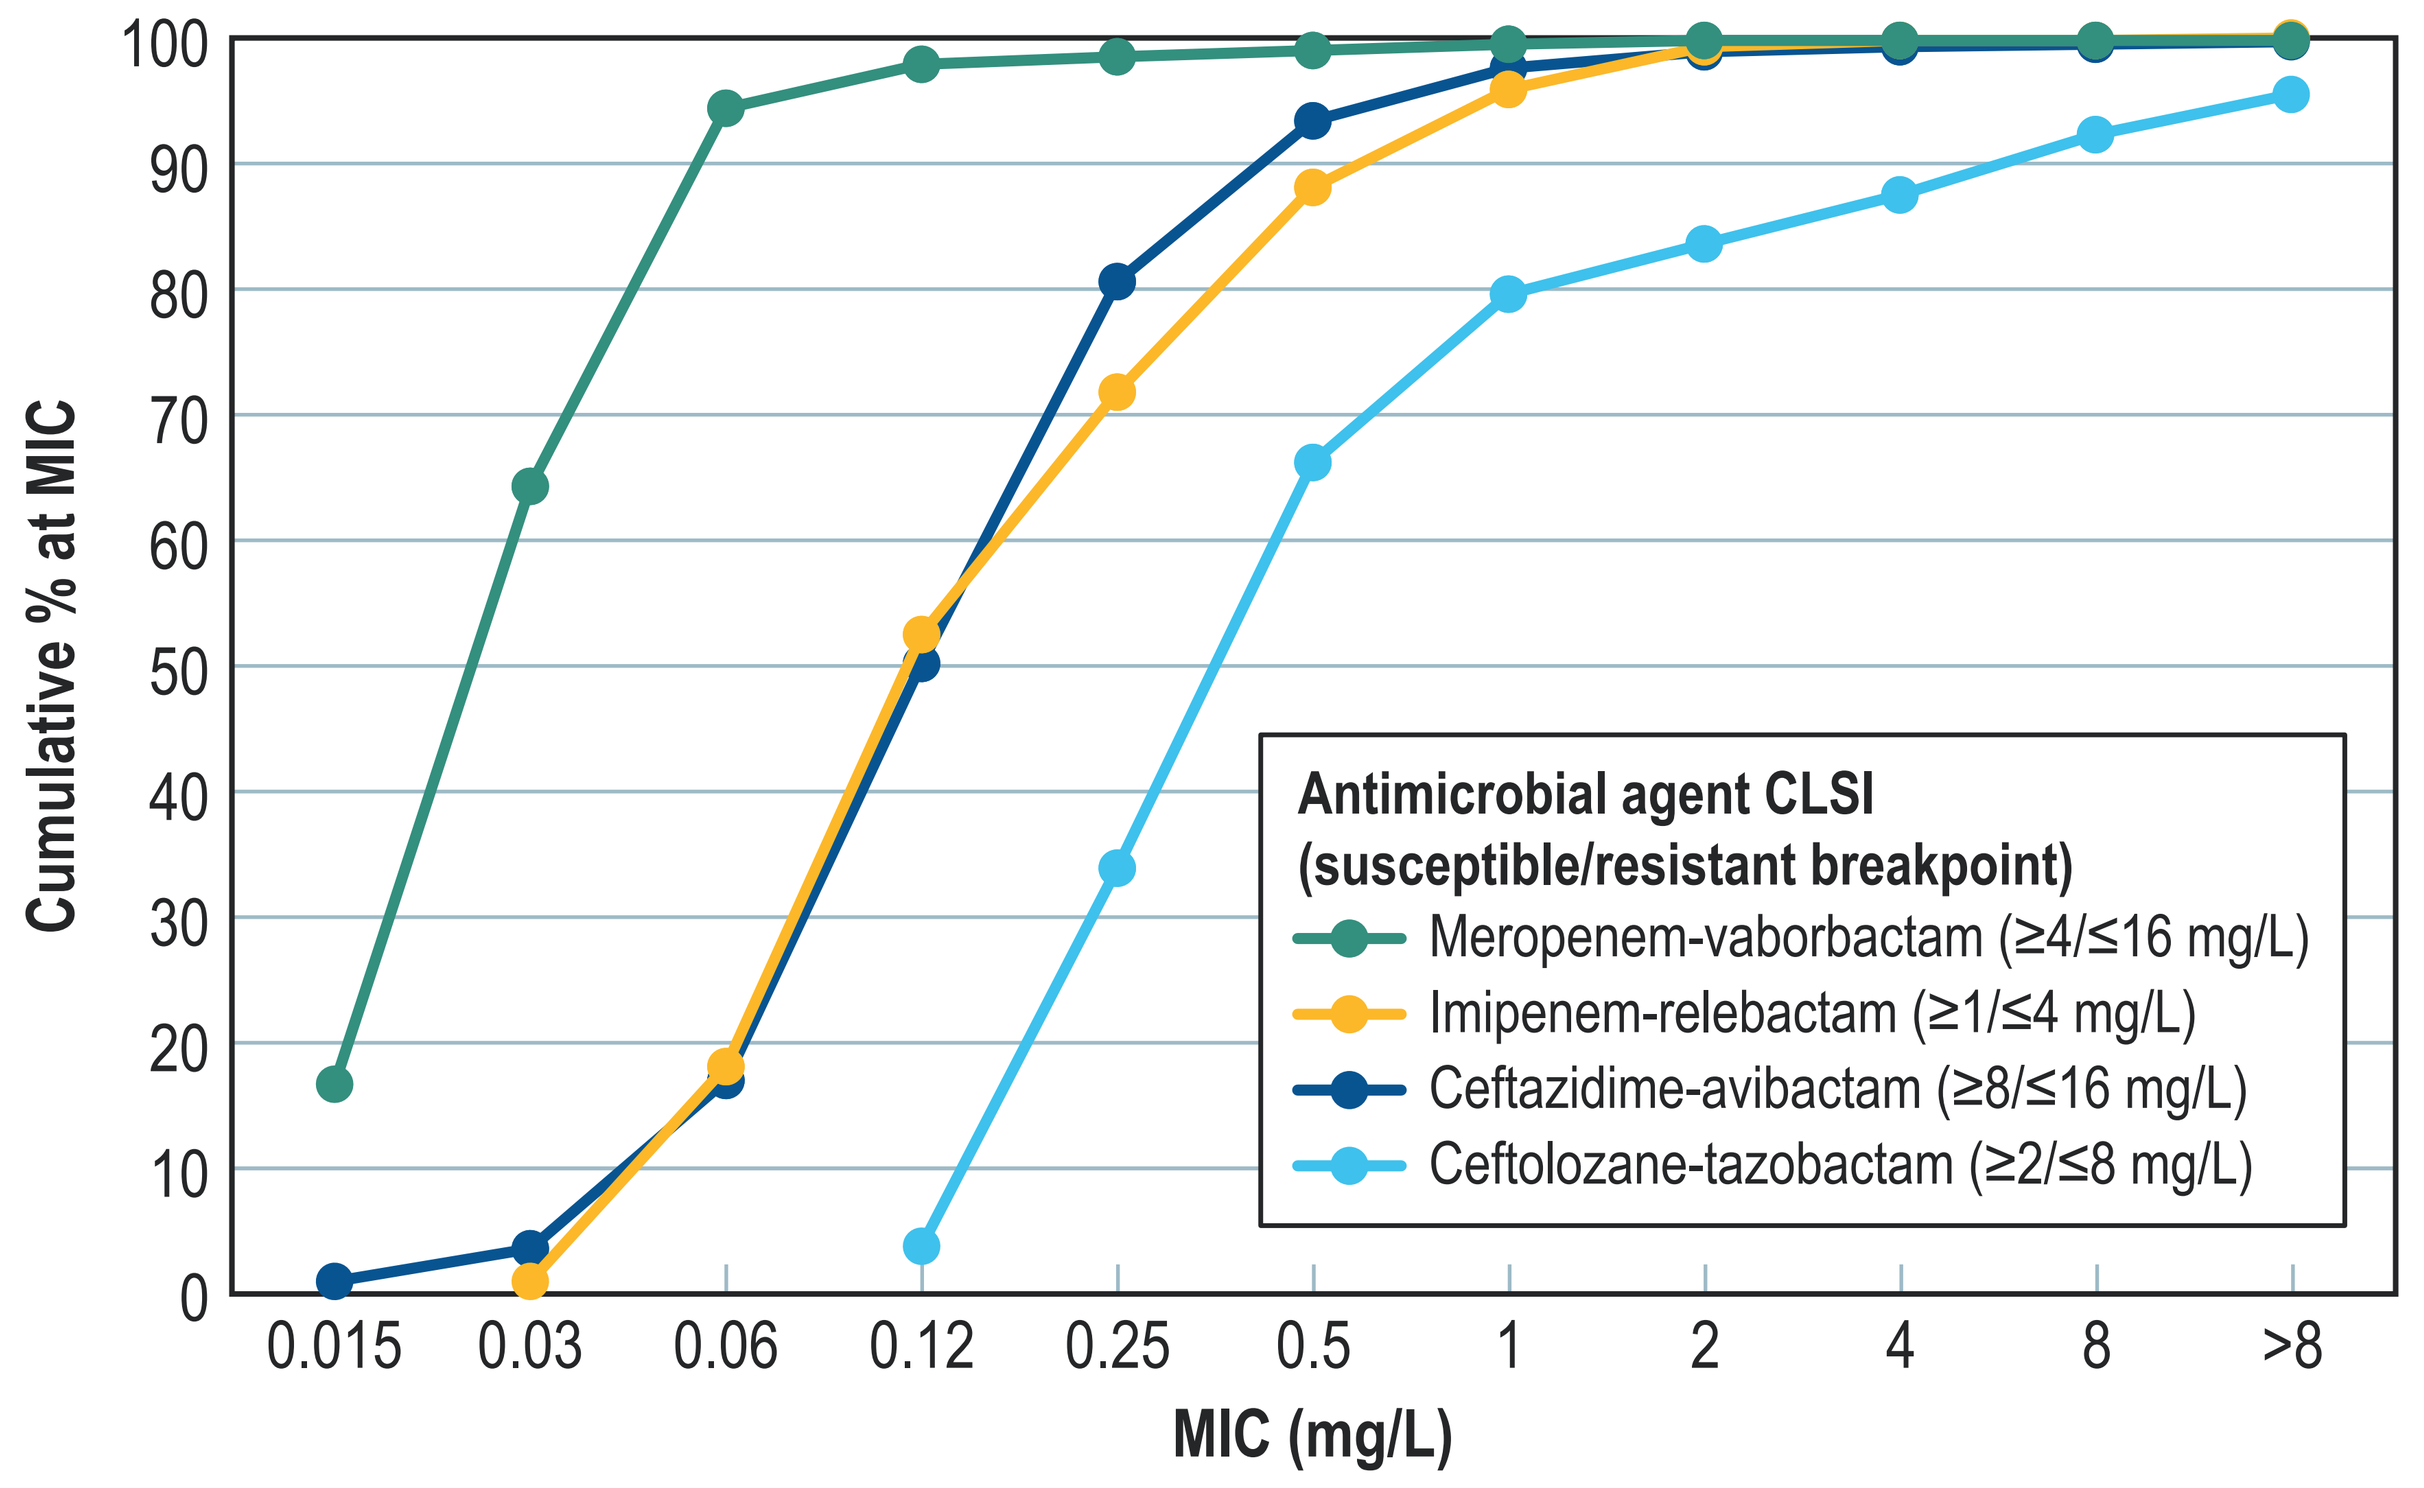
Cumulative % at MIC
0
10
20
30
40
50
60
70
80
90
100
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
>8
MIC (mg/L)
Antimicrobial agent CLSI (susceptible/resistant breakpoint)
Meropenem-vaborbactam (≥4/≤16 mg/L)
Imipenem-relebactam (≥1/≤4 mg/L)
Ceftazidime-avibactam (≥8/≤16 mg/L)
Ceftolozane-tazobactam (≥2/≤8 mg/L)

Figure 4. Activity of BL/BLI and other agents against AmpC-producing isolates

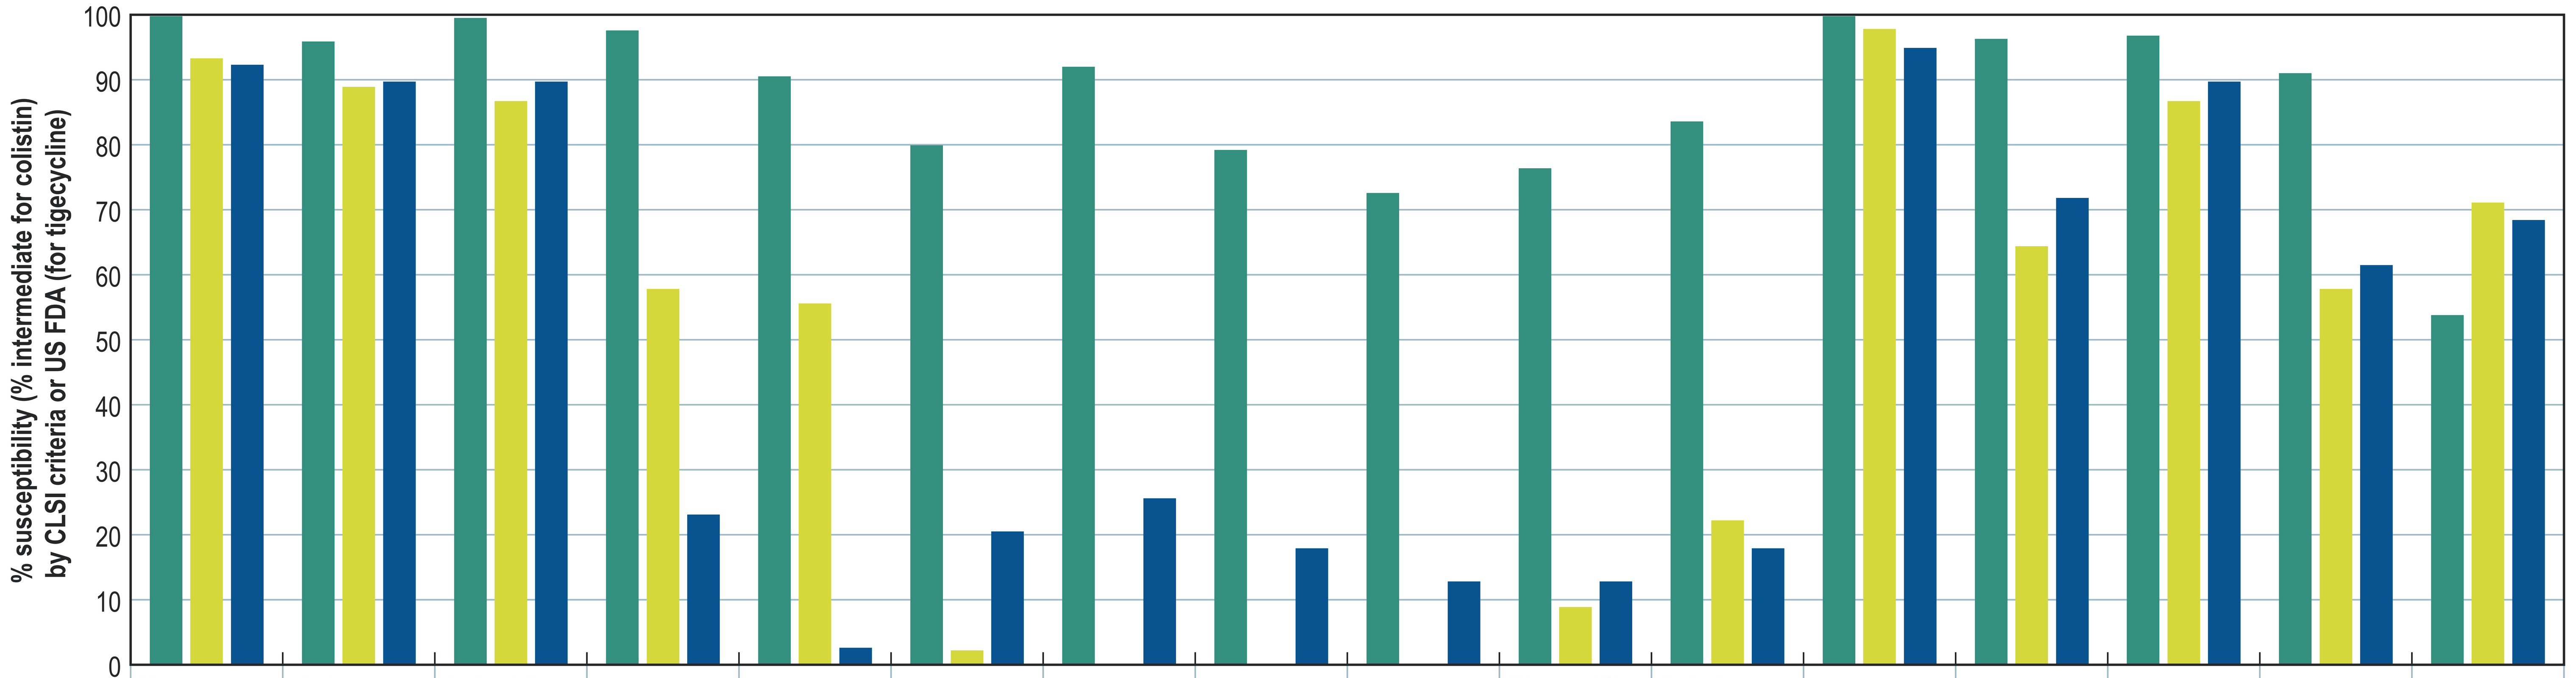
% susceptibility (% intermediate for colistin) by CLSI criteria or US FDA (for tigecycline)
0
10
20
30
40
50
60
70
80
90
100

	Meropenem-vaborbactam	Imipenem-relebactam	Ceftazidime-avibactam	Meropenem	Imipenem	Aztreonam	Cefepime	Ceftazidime	Ceftriaxone	Piperacillin-tazobactam	Ceftolozane-tazobactam	Amikacin	Gentamicin	Tigecycline	Levofloxacin	Colistin
All AmpC-producers (*n*=1,252)	99.8	95.9	99.5	97.6	90.5	79.9	92.0	79.2	72.6	76.4	83.6	99.8	96.3	96.8	91.0	53.8
Cefepime-resistant (*n*=45)	93.3	88.9	86.7	57.8	55.6	2.2	0.0	0.0	0.0	8.9	22.2	97.8	64.4	86.7	57.8	71.1
CNSE (*n*=39)	92.3	89.7	89.7	23.1	2.6	20.5	25.6	17.9	12.8	12.8	17.9	94.9	71.8	89.7	61.5	68.4

Conclusions

- Infections caused by AmpC-producing species often are challenging to treat.
- Understanding the activity of new BL/BLIs is critical, as the use of cefepime and meropenem can lead to resistance.
- Meropenem-vaborbactam, imipenem-relebactam, and ceftazidime-avibactam displayed good activity against AmpC producers.
 - When analyzing carbapenem-nonsusceptible or cefepime-resistant isolates, meropenem-vaborbactam was slightly more active and also more potent than other BL/BLI combinations.

Funding

This study was supported by Melinta Therapeutics, Inc. Authors are employees of JMI Laboratories, which was a paid consultant to Melinta in connection with the development of this poster.

Acknowledgements

The authors thank all of the SENTRY Antimicrobial Surveillance Program participants for providing isolates.

References

1. Bassetti M, Giacobbe D, Castaldo N, Russo A and Vena A. (2021) Role of new antibiotics in extended-spectrum β-lactamase-, AmpC- infections. *Current opinion in infectious diseases* 34: 748-755.
2. CLSI. M07Ed11. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically: eleventh edition. Wayne, PA, Clinical and Laboratory Standards Institute, 2018.
3. CLSI. M100Ed32. Performance standards for antimicrobial susceptibility testing: 32nd informational supplement. Wayne, PA, Clinical and Laboratory Standards Institute, 2022.
4. Jacoby GA. (2009) AmpC beta-lactamases. *Clin. Microbiol. Rev.* 22: 161-182.
5. Tamma PD, Aitken S, Bonoma R, Mathers A, van Duin D and Clancy C. (2022) Infectious Diseases Society of America Guidance on the Treatment of AmpC β-Lactamase-Producing Enterobacterales, Carbapenem-Resistant Acinetobacter baumannii, and Stenotrophomonas maltophilia Infections. *Clin. Infect. Dis.* 74: 2089-2114.
6. Tamma PD, Doi Y, Bonomo RA, Johnson JK, Simner PJ and Antibacterial Resistance Leadership G. (2019) A primer on AmpC beta-lactamases: Necessary knowledge for an increasingly multidrug-resistant world. *Clin. Infect. Dis.* 69: 1446-1455.

Contact

Mariana Castanheira, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
mariana-castanheira@jmilabs.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2022_BLCombosVsAmpCs.pdf

No personal information is stored.

IDWeek 2022, October 19–23, 2022, Washington, D.C.