IDWeek 2022 | Poster #P2176

In Vitro Evaluation of Delafloxacin Activity Against Contemporary US Isolates from Cystic Fibrosis Patients Hospitalized with Pneumonia: Results from the SENTRY Antimicrobial Surveillance Program (2019–2021)

Dee Shortridge[1], Jennifer M Streit[1], Michael D Huband[1], Mariana Castanheira[1]
[1]JMI Laboratories, North Liberty, Iowa, United States

Introduction

- Delafloxacin (DLX) is a broad-spectrum fluoroquinolone approved in the US for the treatment of community-acquired bacterial pneumonia and acute bacterial skin and skin structure infections.
- DLX is indicated to treat CABP caused by multiple pathogens, including methicillin-susceptible *Staphylococcus aureus* (MSSA) and *Pseudomonas aeruginosa* (PSA).
 - DLX has increased activity in acidic environments found at infection sites, and is able to penetrate *S. aureus* biofilms (Lemaire et al. 2011, Siala et al. 2014).
 - Fluoroquinolones can be used for oral treatment of PSA infections in cystic fibrosis (CF) patients (Langton Hewer et al. 2020, Millar et al. 2021).
- In this study, the *in vitro* susceptibilities of DLX and comparator quinolones were determined for clinical isolates from US CF patients collected in the SENTRY Antimicrobial Surveillance Program during 2019–2021.

Methods

- Isolates from CF patients hospitalized with pneumonia were consecutively collected from 19 US participating medical centers from all 9 US Census divisions.
 - Only 1 isolate per patient per infection episode was submitted.
- Isolates were identified at each site and confirmed with MALDI-TOF at JMI Laboratories.
- Susceptibility testing was performed according to CLSI broth microdilution methodology (M07, 2018).
 - FDA interpretive criteria for community acquired bacterial pneumonia were used for DLX.
 - CLSI (M100, 2022) criteria were applied to comparators.
 - As MOX does not have CLSI breakpoints for PSA, ciprofloxacin (CIP) was tested.

Results

- A total of 115 *S. aureus* (SA), including 72 MSSA, and 43 methicillin-resistant SA (MRSA), and 67 PSA (25 CIP-R), were tested.
- Susceptibility (%S) to DLX, levofloxacin (LEV), and moxifloxacin (MOX) for SA overall, or MSSA and %S to DLX, LEV, and ciprofloxacin (CIP) for PSA are shown in Table 1.
- Against all SA, DLX was 73.9%S, LEV was 67.0%S, and MOX was 67.0%S.
- DLX had the highest %S against MSSA (94.4%S).
 - Both LEV and MOX were 87.5%S.
- DLX had the highest %S against PSA (68.7%S).
 - LEV was 43.3%S and CIP was 50.7%S.
- The cumulative percent inhibition of SA, MSSA, MRSA, and PSA are shown in Figures 1 and 2.
 - DLX was the most active quinolone against SA, MSSA, and MRSA (Figure 1).
 - DLX %S against MRSA was 39.5%, while LEV %S was 32.6%.
 - DLX and CIP had similar activity against PSA; both were more active than LEV (Figure 2).
 - DLX %S against CIP-R PSA was 36.0% while LEV %S was 4.0%.

Table 1. Susceptibilities of delafloxacin and comparators tested against *S. aureus*, methicillin-susceptible *S. aureus* (MSSA), and *P. aeruginosa* isolates collected from US cystic fibrosis patients hospitalized with pneumonia

Organism/Antimicrobial agent	No. of isolates	Minimal inhibitory concentrations (MIC) mg/L			CLSI/FDA [a]		
		MIC_{50}	MIC_{90}	MIC range	%S	%I	%R
S. aureus							
Delafloxacin	115	0.008	0.5	0.002 to 4	73.9 [b]	9.6	16.5
Levofloxacin	115	0.25	>4	0.06 to >4	67.0	0.0	33.0
Moxifloxacin	115	≤0.06	>4	≤0.06 to >4	67.0	0.9	32.2
MSSA							
Delafloxacin	72	0.004	0.03	0.002 to 1	94.4 [b]	4.2	1.4
Levofloxacin	72	0.25	4	0.06 to >4	87.5	0.0	12.5
Moxifloxacin	72	≤0.06	2	≤0.06 to >4	87.5	0.0	12.5
P. aeruginosa							
Delafloxacin	67	0.5	4	0.015 to 16	68.7 [c]	11.9	19.4
Levofloxacin	67	2	8	0.06 to 32	43.3	17.9	38.8
Ciprofloxacin	67	0.5	4	0.015 to >4	50.7	11.9	37.3

[a] Criteria as published by FDA (2022) for delafloxacin or CLSI (2022) for comparator agents. S susceptible, I intermediate, and R resistant.
[b] Community-acquired bacterial pneumonia breakpoints for MSSA applied for delafloxacin (FDA).
[c] Community-acquired bacterial pneumonia breakpoints applied for delafloxacin (FDA).

Figure 1. Cumulative percent inhibition by delafloxacin (DLX) or levofloxacin (LEV) against all *S. aureus* and MSSA and MRSA phenotypes from cystic fibrosis patients

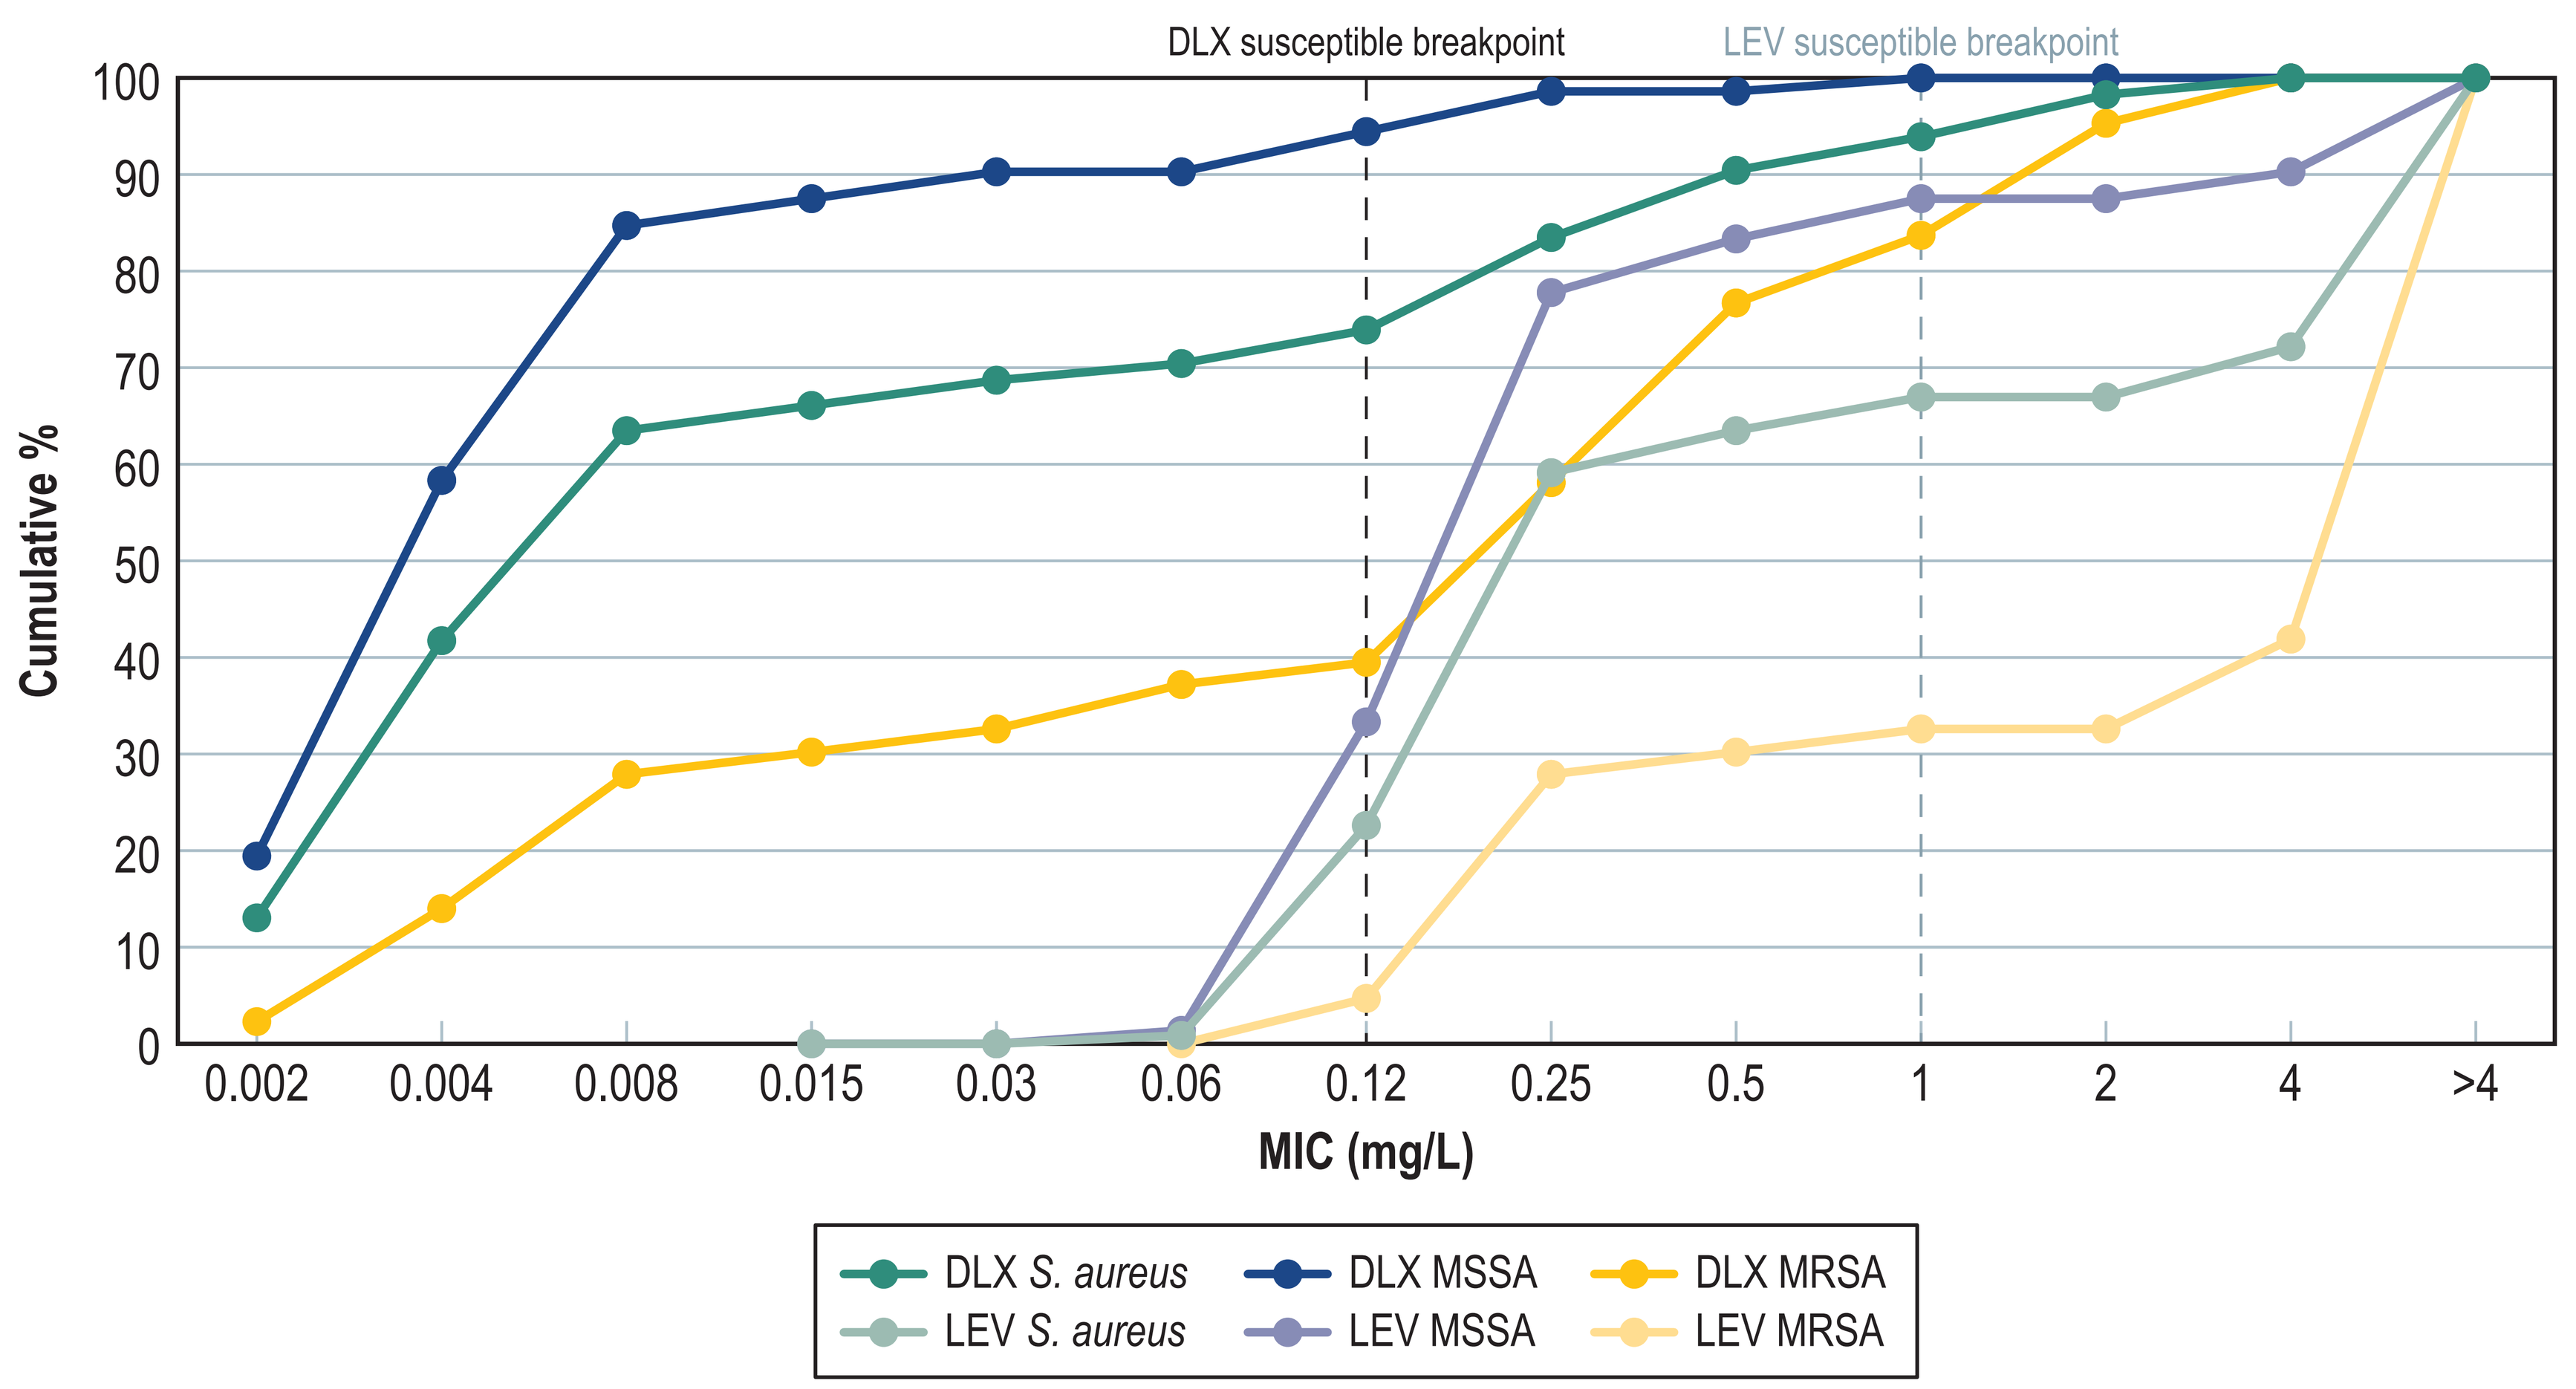
DLX susceptible breakpoint
LEV susceptible breakpoint
Cumulative %
100
90
80
70
60
50
40
30
20
10
0
0.002
0.004
0.008
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
>4
MIC (mg/L)
DLX S. aureus
LEV S. aureus
DLX MSSA
LEV MSSA
DLX MRSA
LEV MRSA

Figure 2. Cumulative percent inhibition by delafloxacin (DLX), levofloxacin (LEV), or ciprofloxacin (CIP) against *P. aeruginosa* and CIP-R *P. aeruginosa* from cystic fibrosis patients

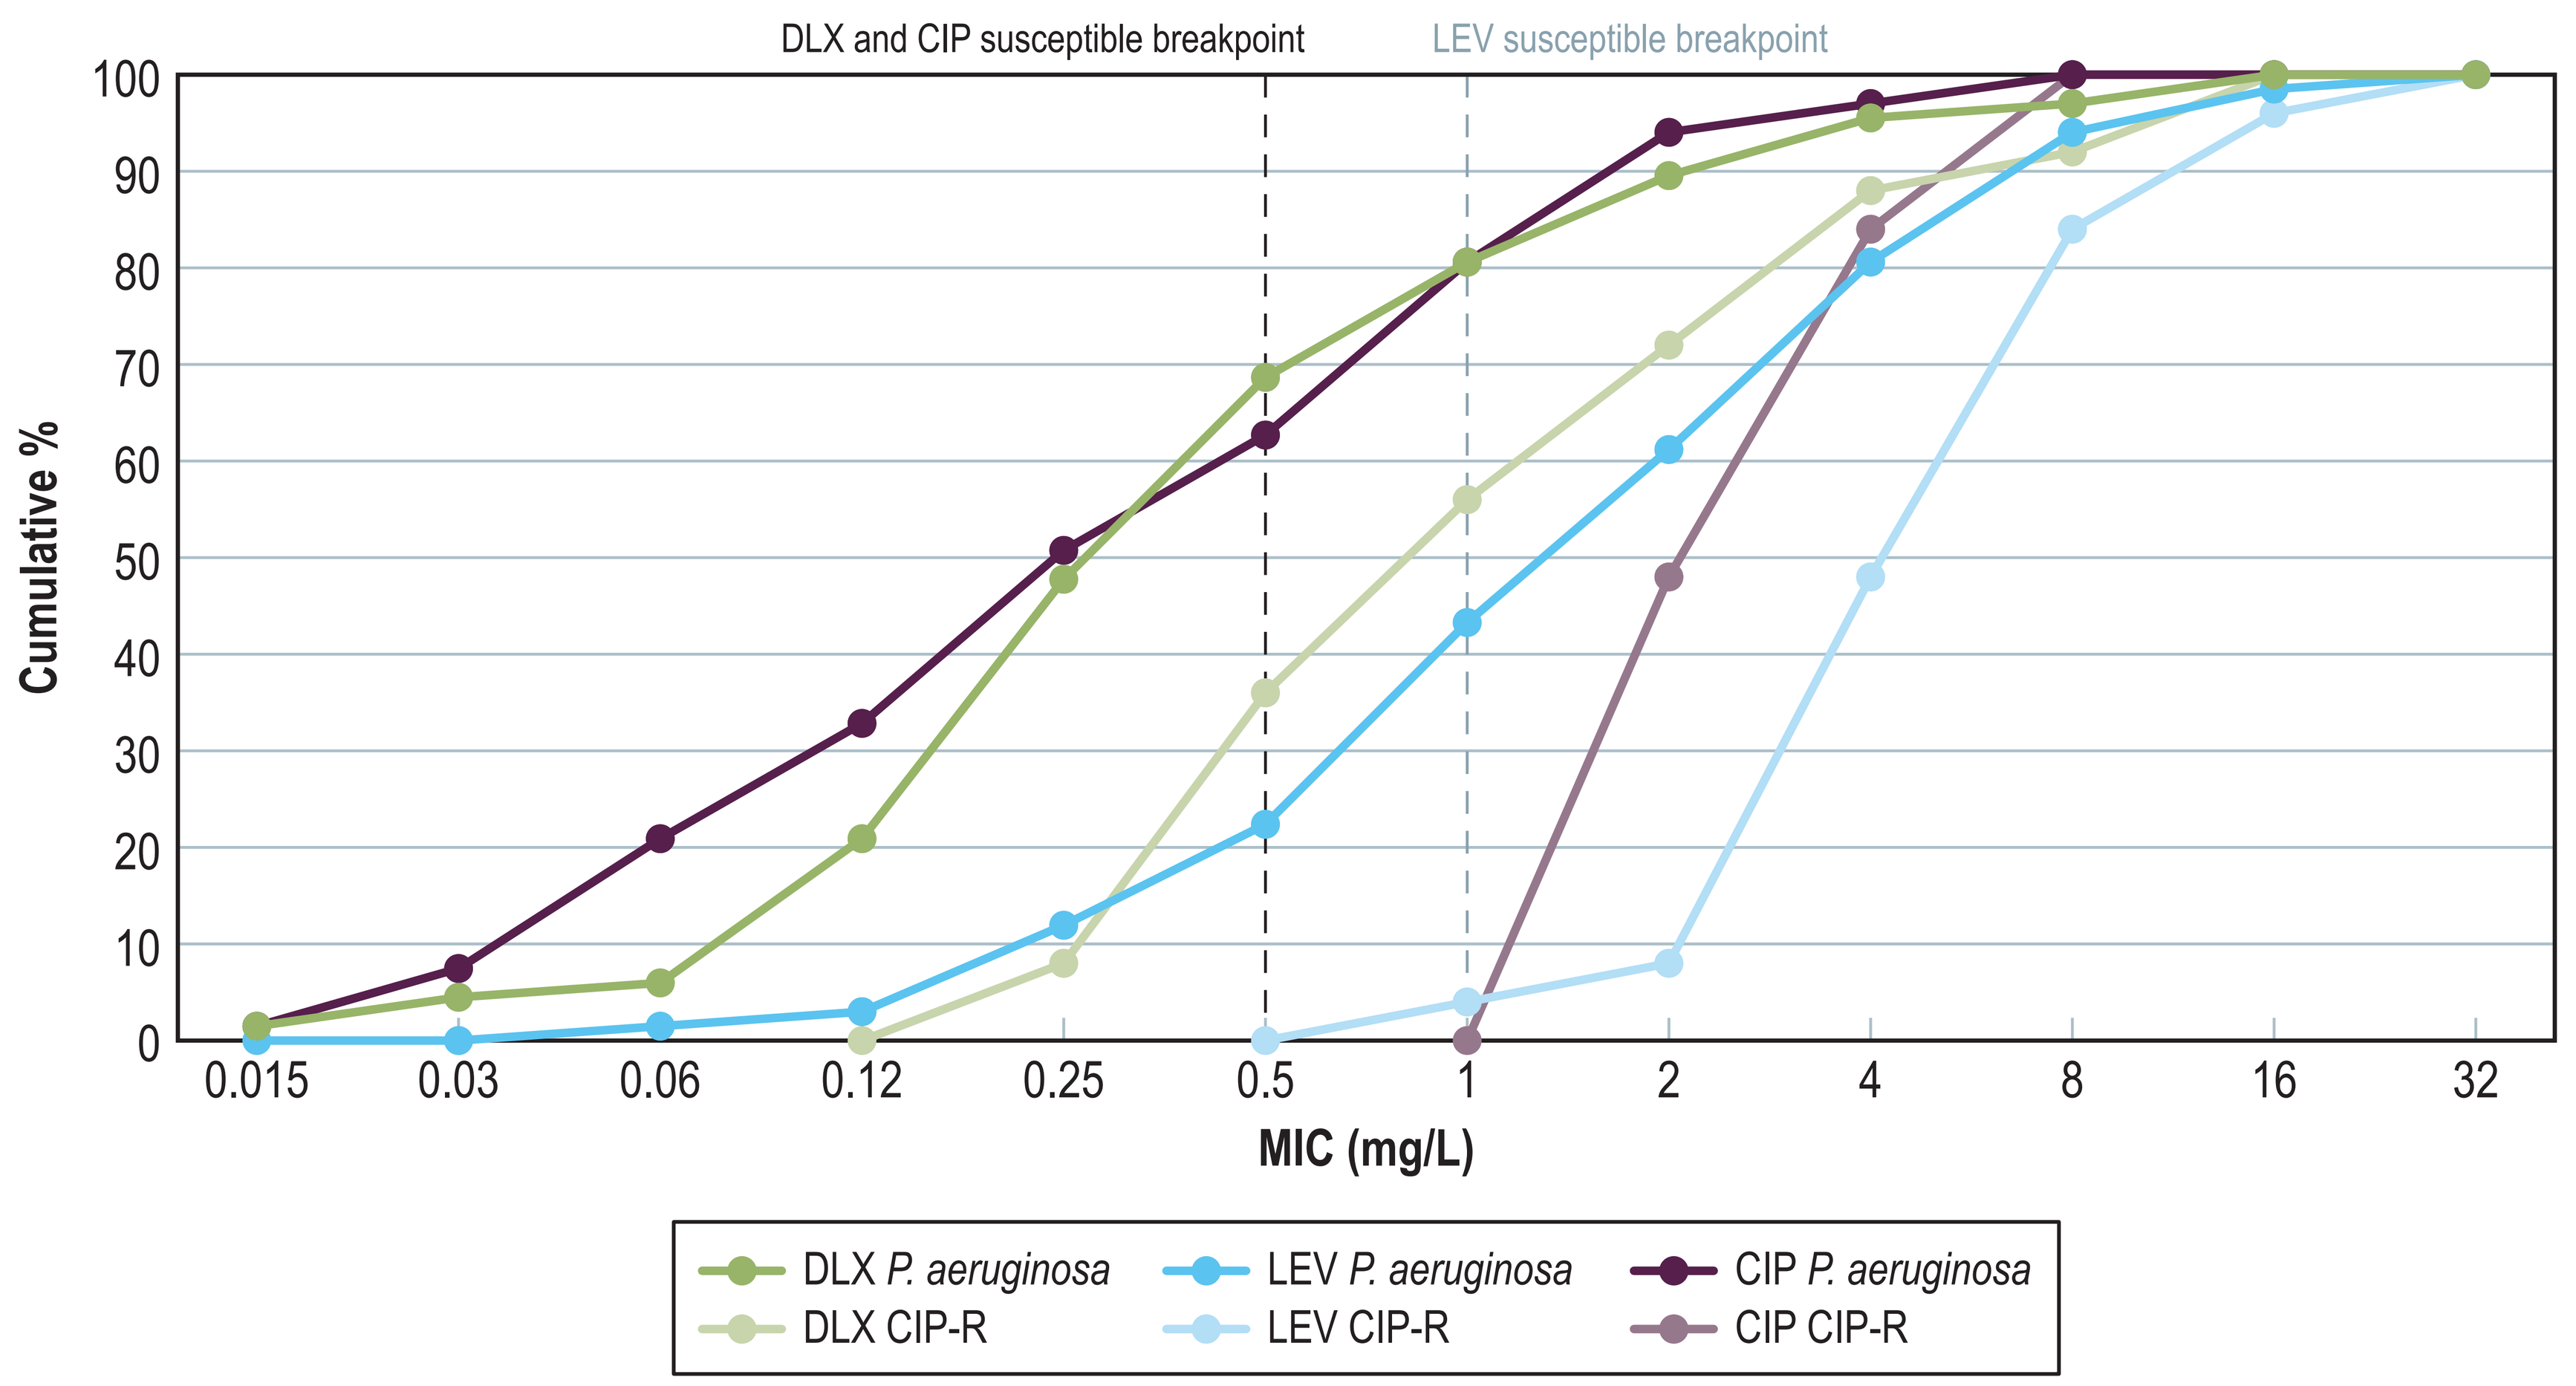
DLX and CIP susceptible breakpoint
LEV susceptible breakpoint
Cumulative %
100
90
80
70
60
50
40
30
20
10
0
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
MIC (mg/L)
DLX P. aeruginosa
DLX CIP-R
LEV P. aeruginosa
LEV CIP-R
CIP P. aeruginosa
CIP CIP-R

Conclusions

- DLX had good activity against recent MSSA and PSA isolates from US CF patients hospitalized with pneumonia.
- DLX had the highest %S of the fluoroquinolones tested for all organism groups.
 - Due to the increased activity of DLX compared to LEV or CIP, determination of the DLX MIC may be useful if considering a fluoroquinolone.
- These *in vitro* data suggest that DLX could be a useful therapy when both MSSA and PSA coverage is needed in CF patients.

Funding

This study was supported by Melinta Therapeutics.

Authors are employees of JMI Laboratories, which was paid consultant to Melinta Therapeutics in connection with the development of this poster.

Acknowledgments

The authors thank all of the participant centers for the work in providing isolates.

References

Langton Hewer, S.C., A.R. Smyth, M. Brown, A.P. Jones, H. Hickey, D. Kenna, D. Ashby, A. Thompson, P.R. Williamson and TORPEDO-CF study group. Intravenous vs. oral antibiotics for eradication of *Pseudomonas aeruginosa* in cystic fibrosis (TORPEDO-CF); a randomized controlled trial. Lancet Respir Med. 2020; 8 (10): 975–986.

Lemaire, S., P.M. Tulkens, F. Van Bambeke. Contrasting effects of acidic pH on the extracellular and intracellular activities of the anti-gram-positive fluoroquinolones moxifloxacin and delafloxacin against Staphylococcus aureus. Antimicrob Agents Chemother, 2011; 55(2): 649–658.

Millar, B.C., J. McCaughan, J.C. Rendall, J.E. Moore. Delafloxacin—A novel fluoroquinolone for the treatment of ciprofloxacin-resistant *Pseudomonas aeruginosa* in patients with cystic fibrosis. Clin Respir J, 2021; 15:116–120.

Siala W., M.-P. Mingeot-Leclercq, P.M. Tulkens, M. Hallin, O. Denis, F. Van Bambeke. Comparison of the antibiotic activities of daptomycin, vancomycin and the investigation fluoroquinolone delafloxacin against biofilms from *Staphylococcus aureus* clinical isolates. Antimicrob Agents Chemother, 2014; 58(11): 6385–6397.

Clinical and Laboratory Standards Institute (2018). *M07Ed11. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically: eleventh edition*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2022). *M100Ed32. Performance standards for antimicrobial susceptibility testing: 32nd informational supplement*. Wayne, PA: CLSI.

FDA. Antibacterial Susceptibility Test Interpretive Criteria: https://www.fda.gov /drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria.

Contact

Dee Shortridge, Senior Director
345 Beaver Kreek Center, Suite A
North Liberty, IA 52317
319-665-3370
dee-shortridge@jmilabs.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2022_DelaVsCFPneumonia.pdf

No personal information is stored.

IDWeek 2022, October 19–23, 2022, Washington, D.C.